Stromal hypersialylation within colorectal tumors contributes to immunosuppression of T cell adaptive immunity in the tumor microenvironment

Oliver Treacy[1,2], Jenny Che[3], Wayne Gatlin[3], Lizhi Cao[3], Hannah Egan[1,2], Aoise O'Neill[1], Michael O'Dwyer[4], Pushpa Jayaraman[3], Li Peng[3], **Aideen E Ryan[1,2]**.

[1]Discipline of Pharmacology and Therapeutics, College of Medicine, Nursing and Health Sciences, University of Galway, Ireland, [2]Regenerative Medicine Institute (REMEDI), College of Medicine, Nursing and Health Sciences, University of Galway, Ireland, [3]Palleon Pharmaceuticals, Waltham, MA 02451, USA, [4]Department of Hematology, Galway University Hospital, Galway, Ireland.

Background

- The tumor microenvironment (TME) is abundant in cancer-associated fibroblasts (CAFs) that can radically influence the cancer disease trajectory, especially in colorectal cancers (CRC).
- Directly targeting cancer-associated fibroblasts may hold great promise in augmenting CRC treatment, however, the limited understanding of the mechanisms mediating stromal immunosuppression remains an obstacle.
- The glyco-immune checkpoint (Siglec/Sialoglycan) axis has recently emerged as a new mechanism of cancer immune evasion.

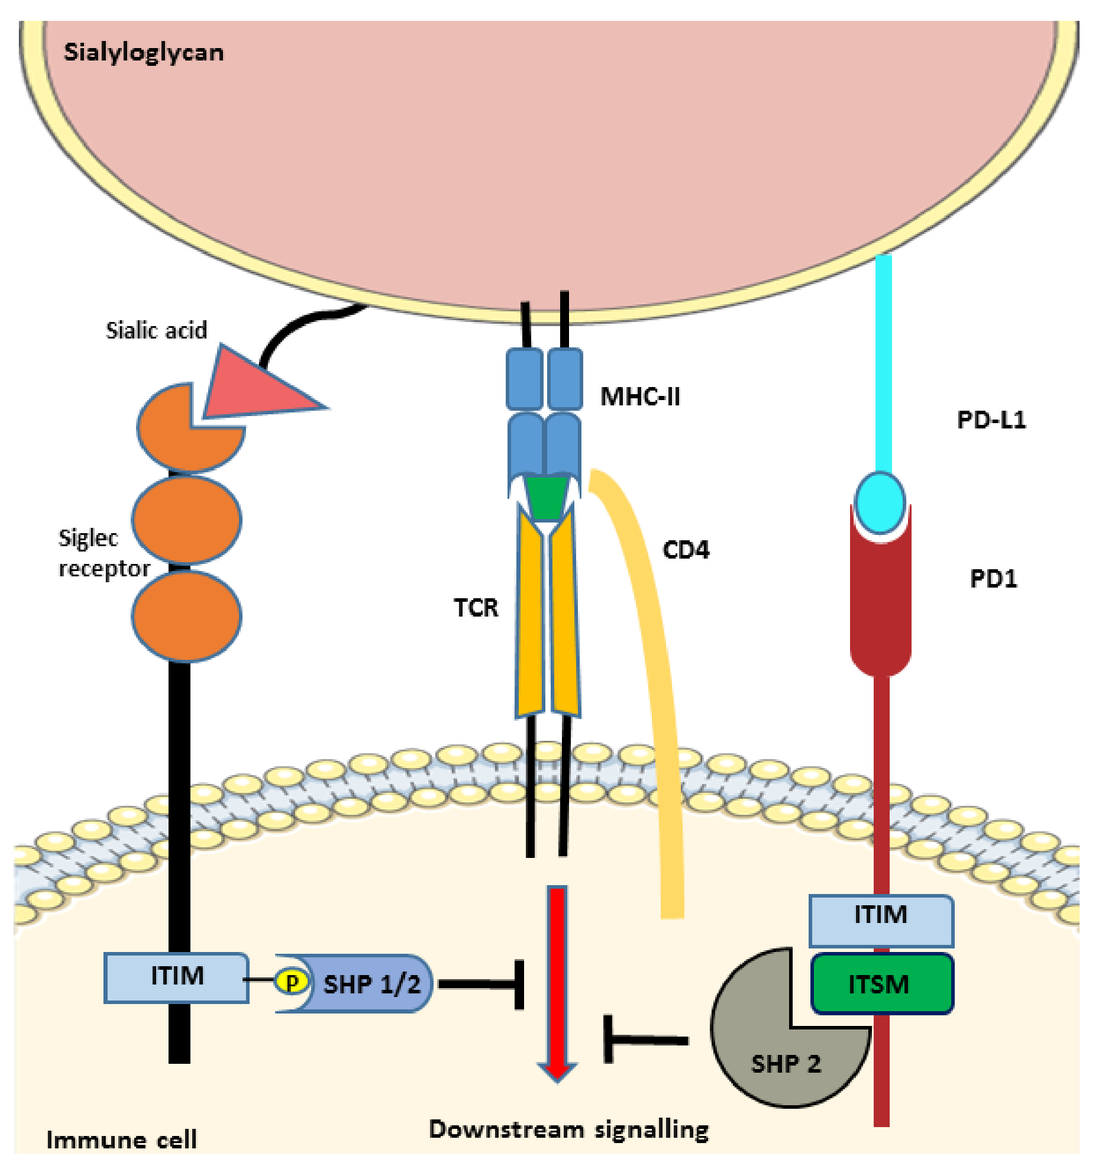
Sialyloglycan
Sialic acid
Siglec receptor
MHC-II
TCR
CD4
PD-L1
PD1
ITIM
SHP 1/2
ITIM
ITSM
SHP 2
Immune cell
Downstream signalling

The aim of this study was to evaluate the role of stromal hypersialylation on the CRC tumor microenvironment.

Methods

Tissue microarrays of human CRC tumors were profiled for sialoglycan reactivity by immunohistochemistry (IHC), using the HYDRA-3, -7 and -9.

The HYDRA platform developed by Palleon Pharmaceuticals, is a set of proprietary reagents, consisting of a hexametric fusion of the extracellular domain (containing the carbohydrate recognition domain) of Siglec-3, -7 or -9, a trimerization motif, and a mouse Fc domain.

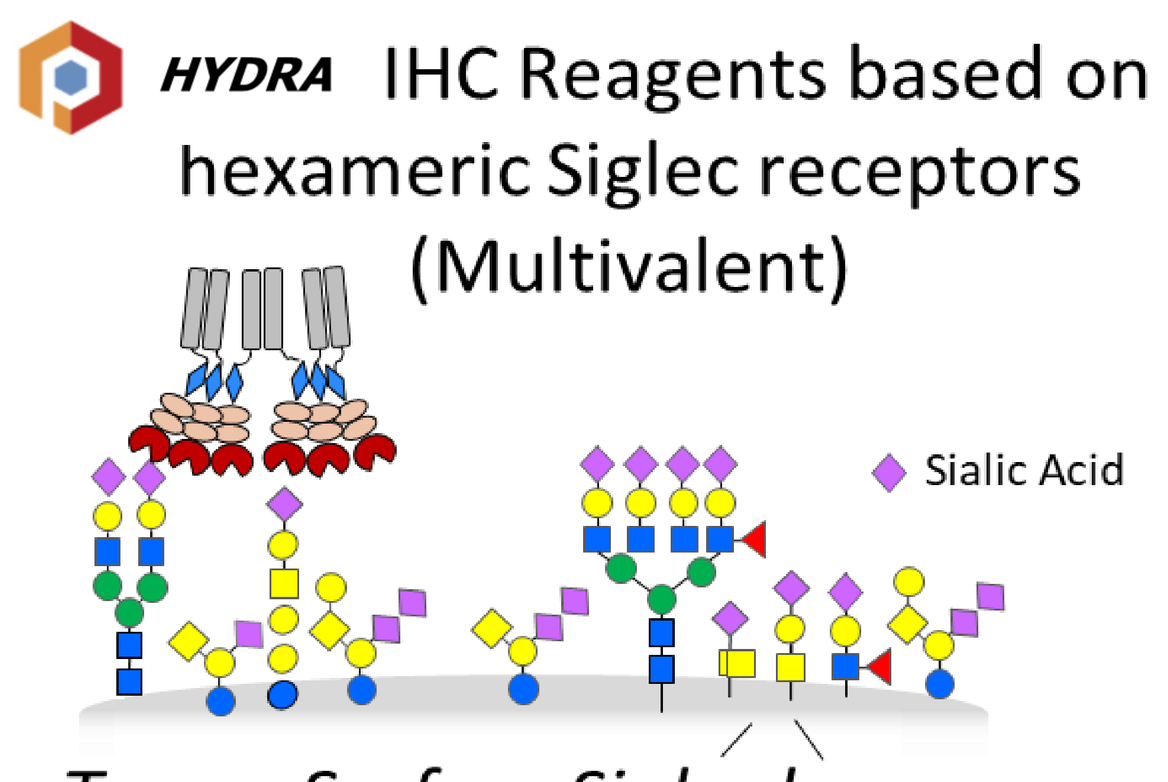
HYDRA IHC Reagents based on hexameric Siglec receptors (Multivalent)
Sialic Acid
Tumor Surface Sialoglycans

Human bone marrow derived mesenchymal cells (BM-MSCs) were isolated and conditioned (Figure 3A). Siglec-7 and Siglec-9 ligand expression was evaluated by flow cytometry.

Human normal-associated fibroblasts (NAFs) or CAFs from dissociated CRC tumors were characterized for sialic acid expression using lectins (MAL-II and SNA-1) and using HYDRA reagents (Siglec-7,9 HYDRA) by flow cytometry.

NAFs and CAFs were pre-treated with human Neu-2 engineered sialidase (SIA) or untreated before co-culture with CD3-sorted T cells from healthy donors. The effect of sialidase pre-treatment on NAF/CAF immunomodulation was evaluated by measuring NAF/CAF sialyation levels, as well as T cell proliferation, function, and checkpoint receptor expression.

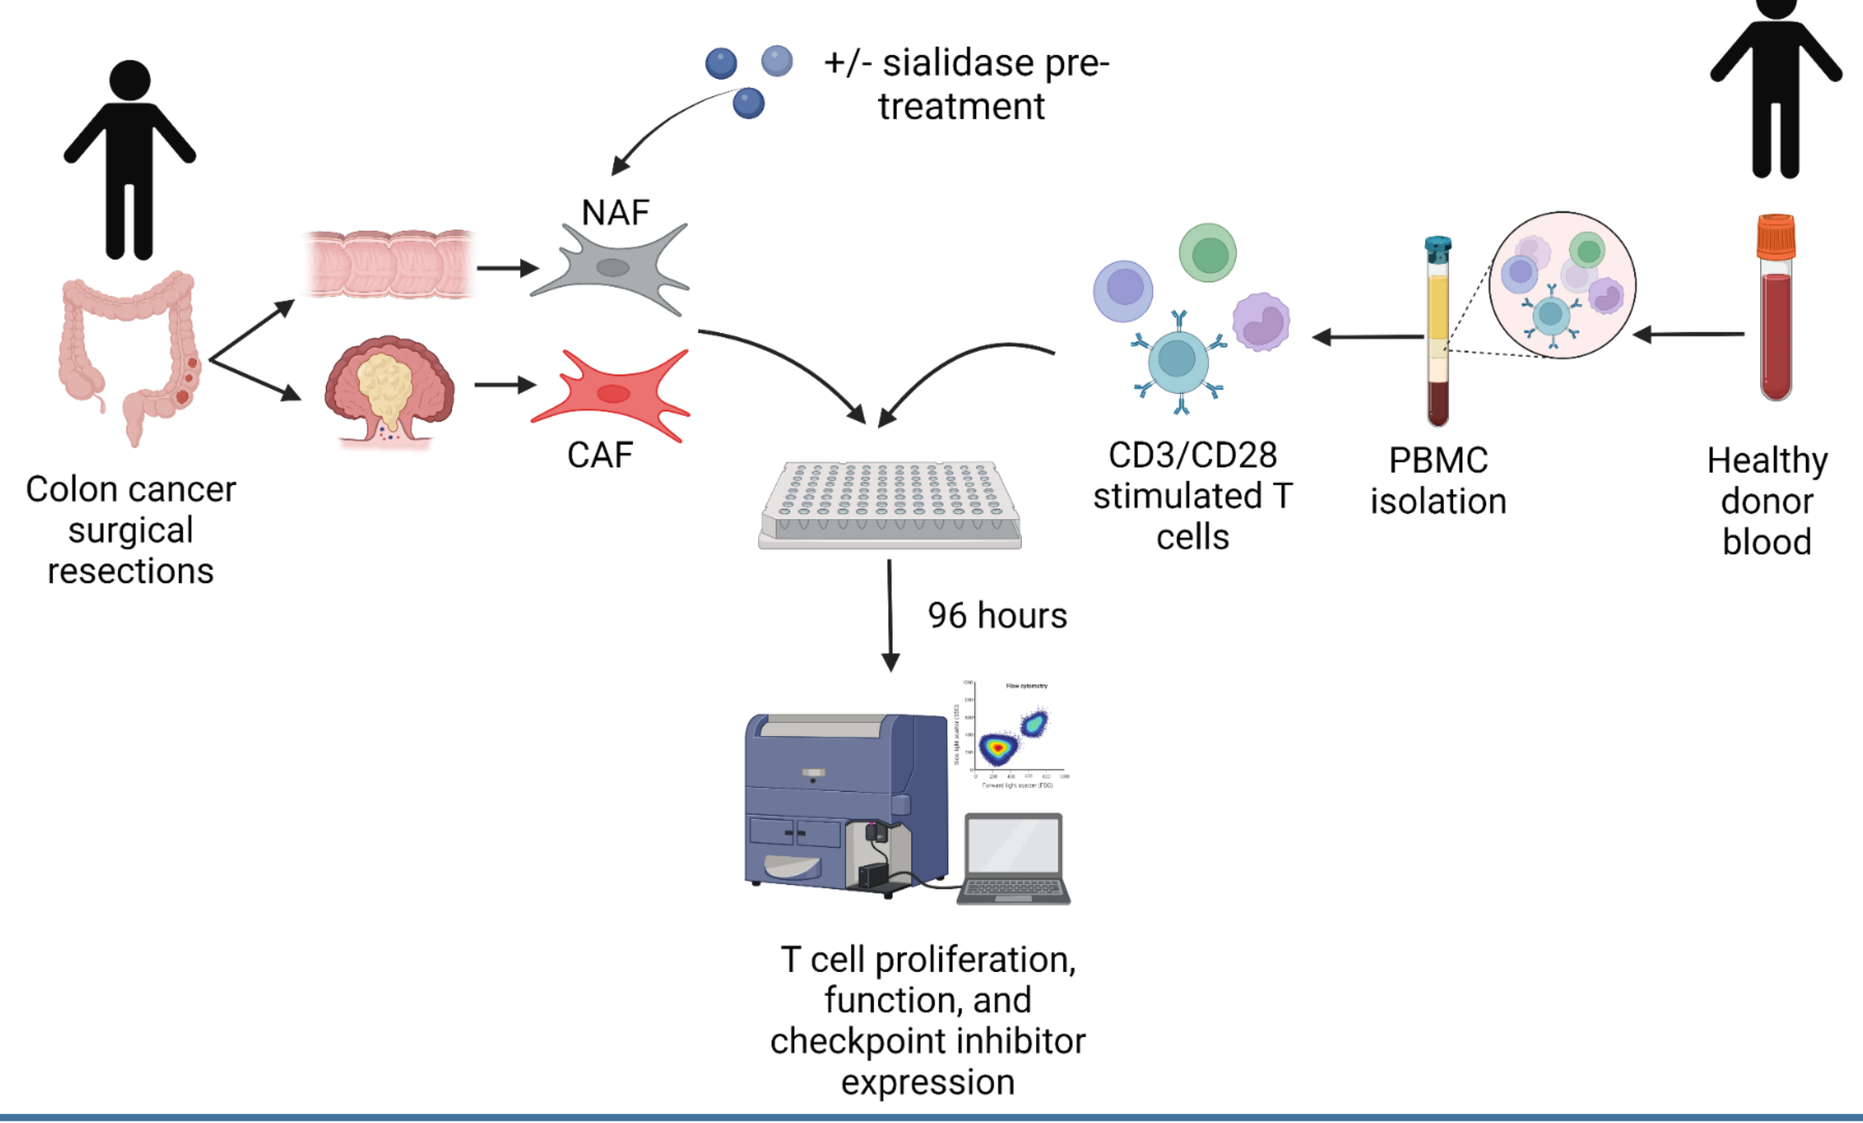
+/- sialidase pre-treatment
NAF
CAF
Colon cancer surgical resections
CD3/CD28 stimulated T cells
PBMC isolation
Healthy donor blood
96 hours
T cell proliferation, function, and checkpoint inhibitor expression

Human CRC tumours are hypersialylated at the tumor-stromal interface

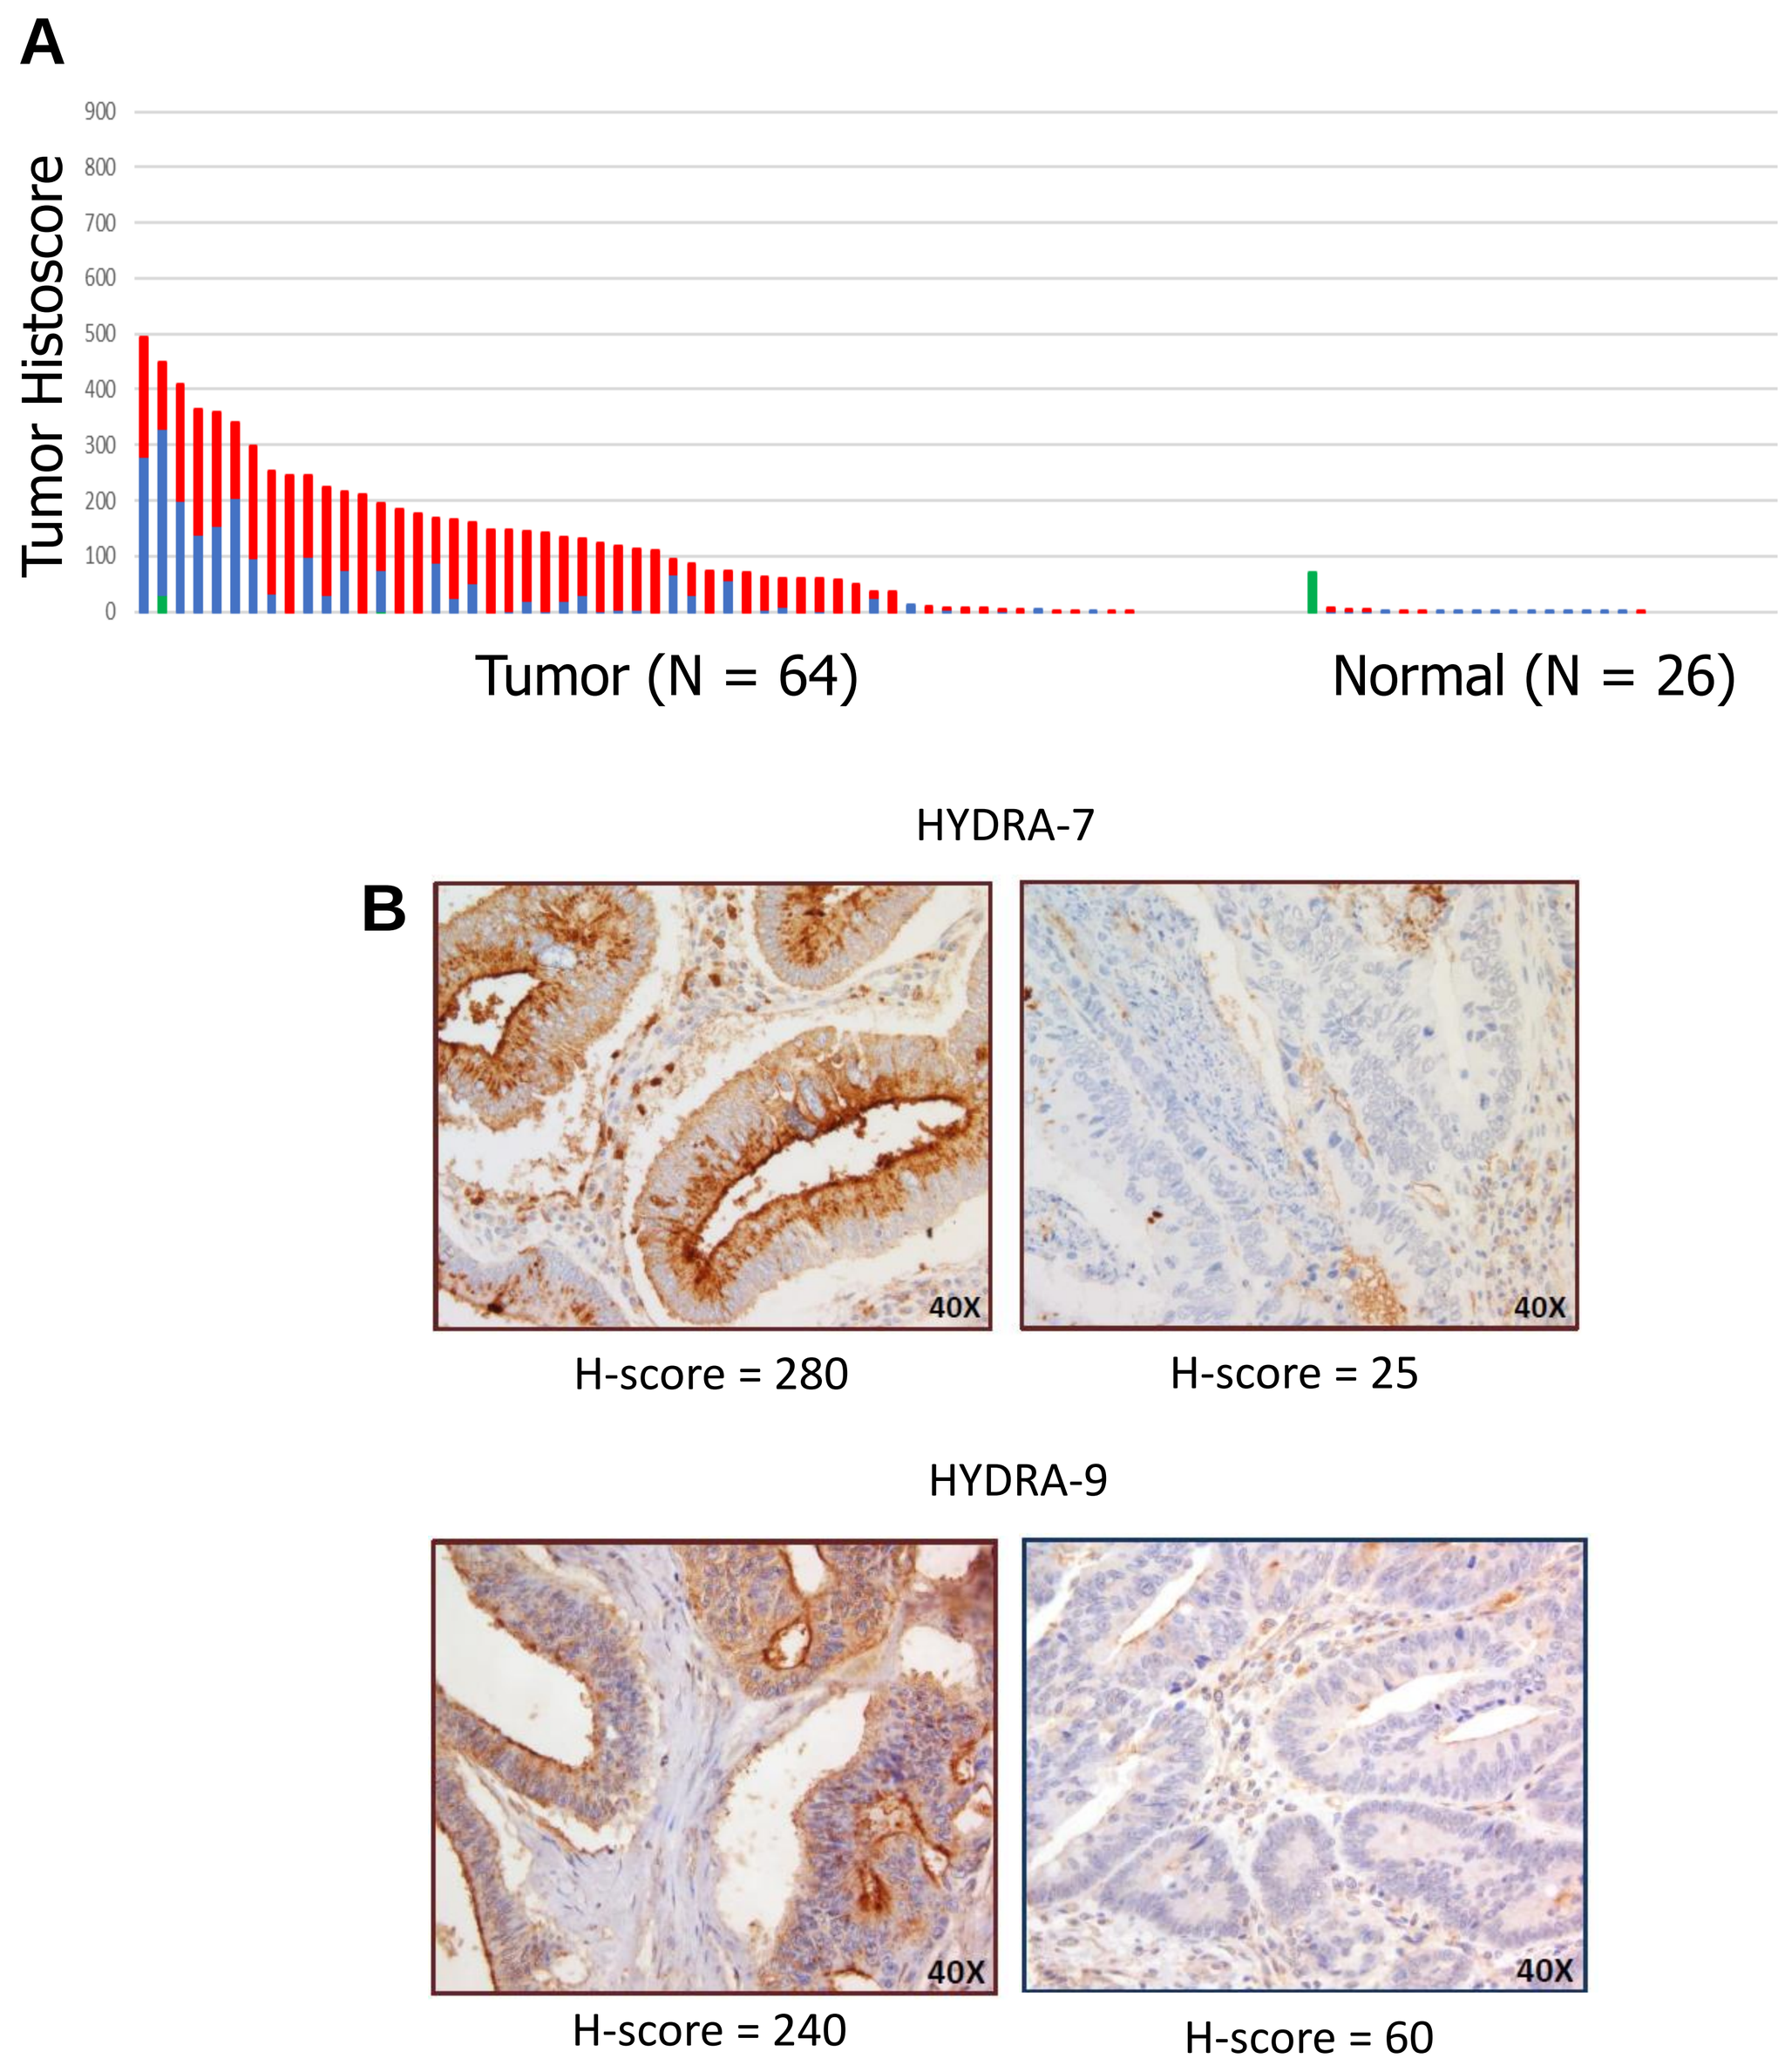
A
Tumor Histoscore
HYDRA-9
HYDRA-7
HYDRA-3
Tumor (N = 64)
Normal (N = 26)
B
HYDRA-7
H-score = 280
H-score = 25
HYDRA-9
H-score = 240
H-score = 60

Figure 1. Colorectal tumors are hypersialylated and show predominantly HYDRA-7 (Siglec 7L) and -9 (Siglec 9L) reactivity. (A) Tumor tissue microarray of commercial source containing colon cancer and normal colon epithelial tissue samples were stained by IHC with HYDRA-3, -7, -9 separately and scored by histoscore (H-score, range 0-300). The sum of all three HYDRA H-scores were plotted to compare tumor vs normal tissue samples. HYDRA H-score was found ≥50 (any HYDRA) in 63% (44/64) of tumor samples tested. HYDRA H-score of normal tissue are typically <10. (B) Representative IHC images of colorectal tumors stained with HYDRA-7 or -9. Images were taken at 40x magnification.

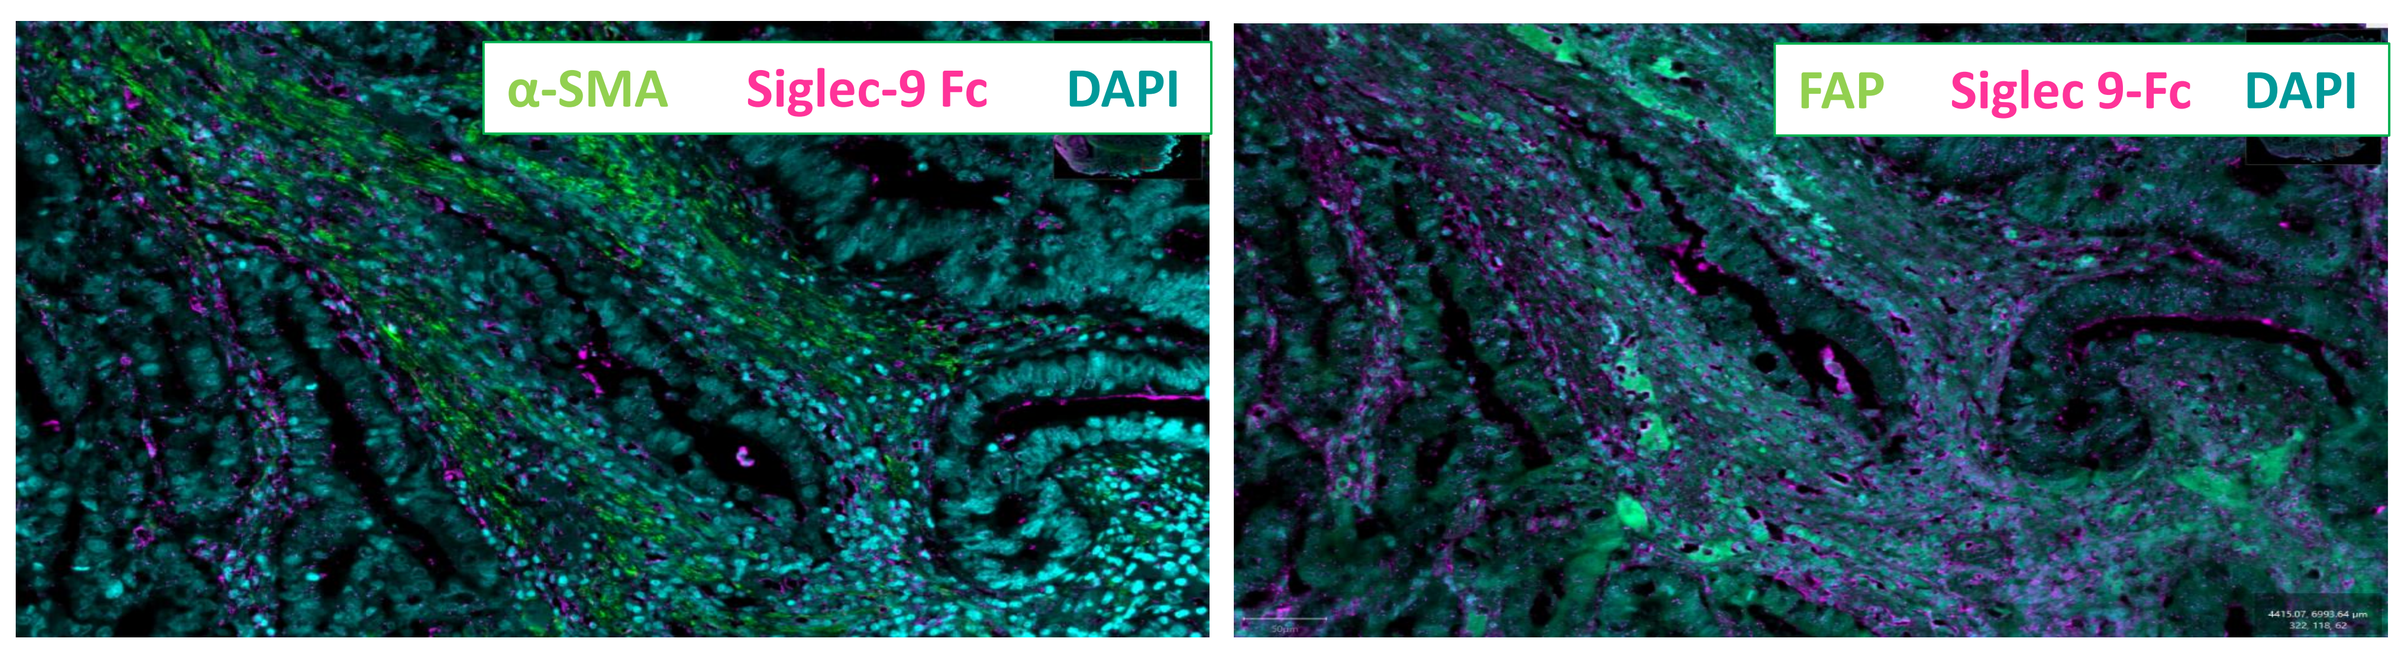
α-SMA Siglec-9 Fc DAPI
FAP Siglec 9-Fc DAPI

Figure 2. Siglec-9 ligand expression (stained by Siglec-9 Fc) co-localizes with stromal cells in the colorectal tumor microenvironment. Immunofluorescence-stained FFPE tissue sections from human CRC patient tumors using recombinant human Siglec-9 Fc and two separate fibroblast characterization markers, α-smooth muscle actin (α-SMA) and fibroblast activation protein (FAP).

Tumor-conditioned stromal cells are more highly sialylated than cancer cells

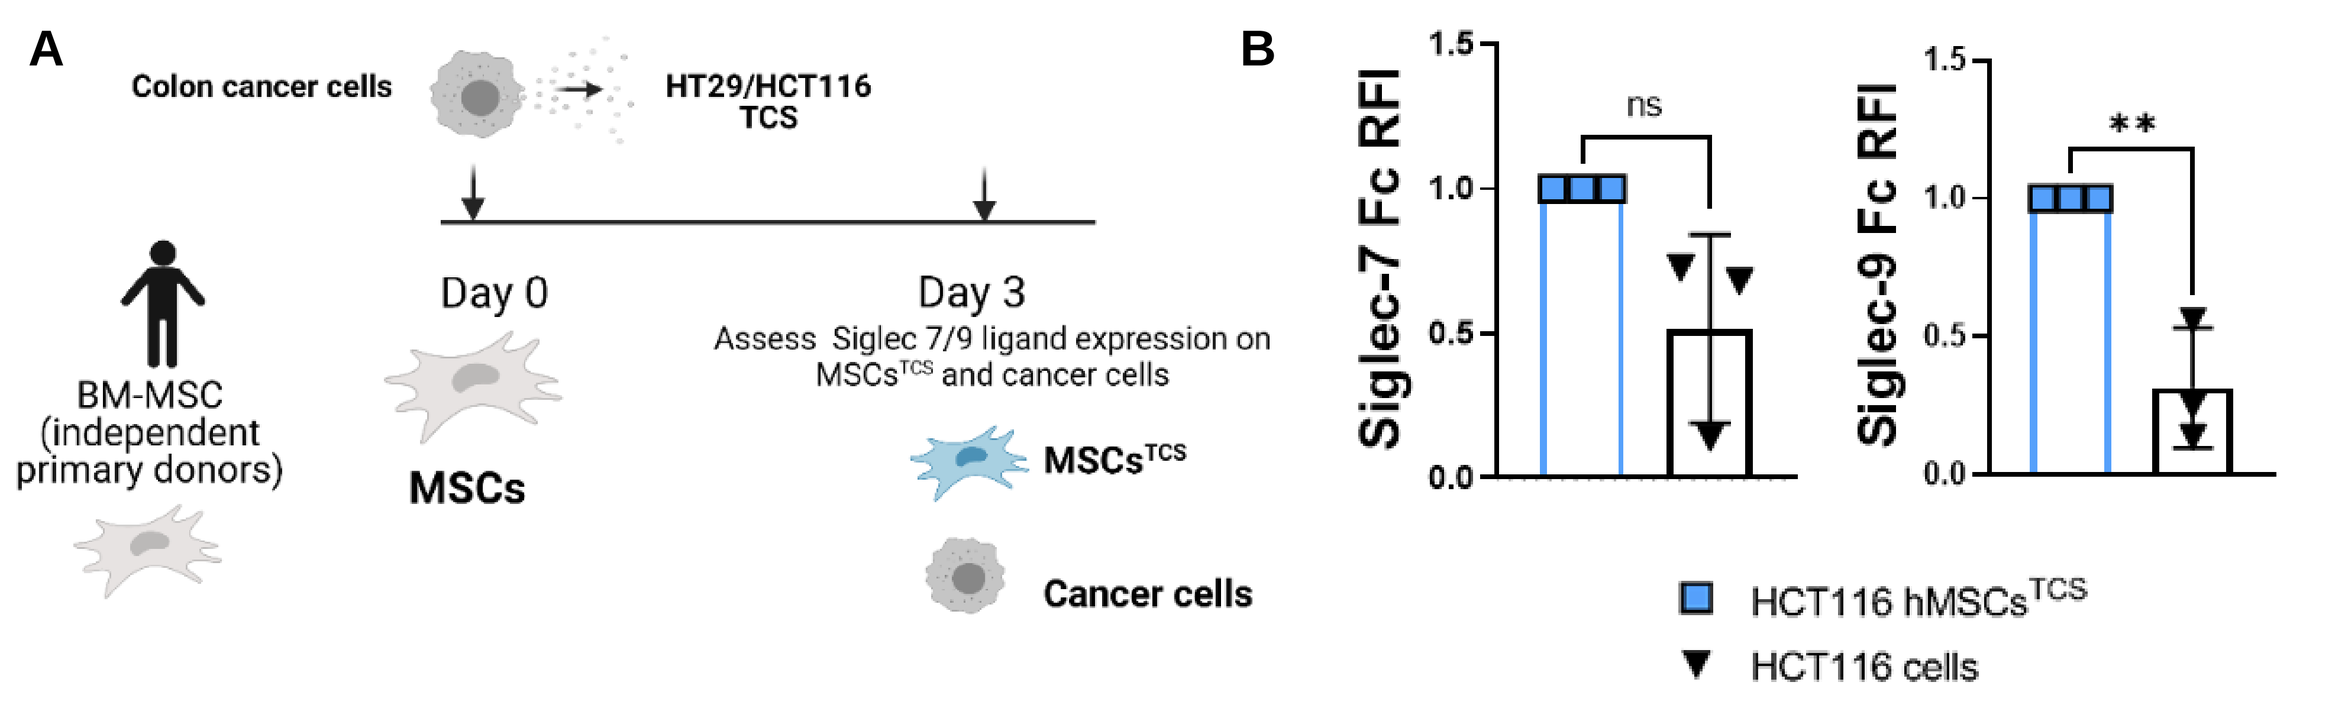
A
Colon cancer cells
HT29/HCT116 TCS
Day 0
Day 3
Assess Siglec 7/9 ligand expression on MSCsTCS and cancer cells
BM-MSC (independent primary donors)
MSCs
MSCsTCS
Cancer cells
B
Siglec-7 Fc RFI
Siglec-9 Fc RFI
ns
**
HCT116 hMSCsTCS
HCT116 cells

Figure 3. Tumor-conditioned human MSCs express higher levels of Siglec ligands compared to cancer cells. (A) Schematic overview of human bone marrow-derived MSC-conditioning regime using tumor cell secretome (TCS) from the CRC cell line HCT116. (B) RFI (relative to HCT116 $MSCs^{TCS}$) of Siglec-7 and Siglec-9 ligand expression on HCT116 cancer cells. Data are mean ± SD; **p < 0.01 using unpaired t test. n = 3 biological replicates.

Human CRC patient-derived CAFs are hypersialylated

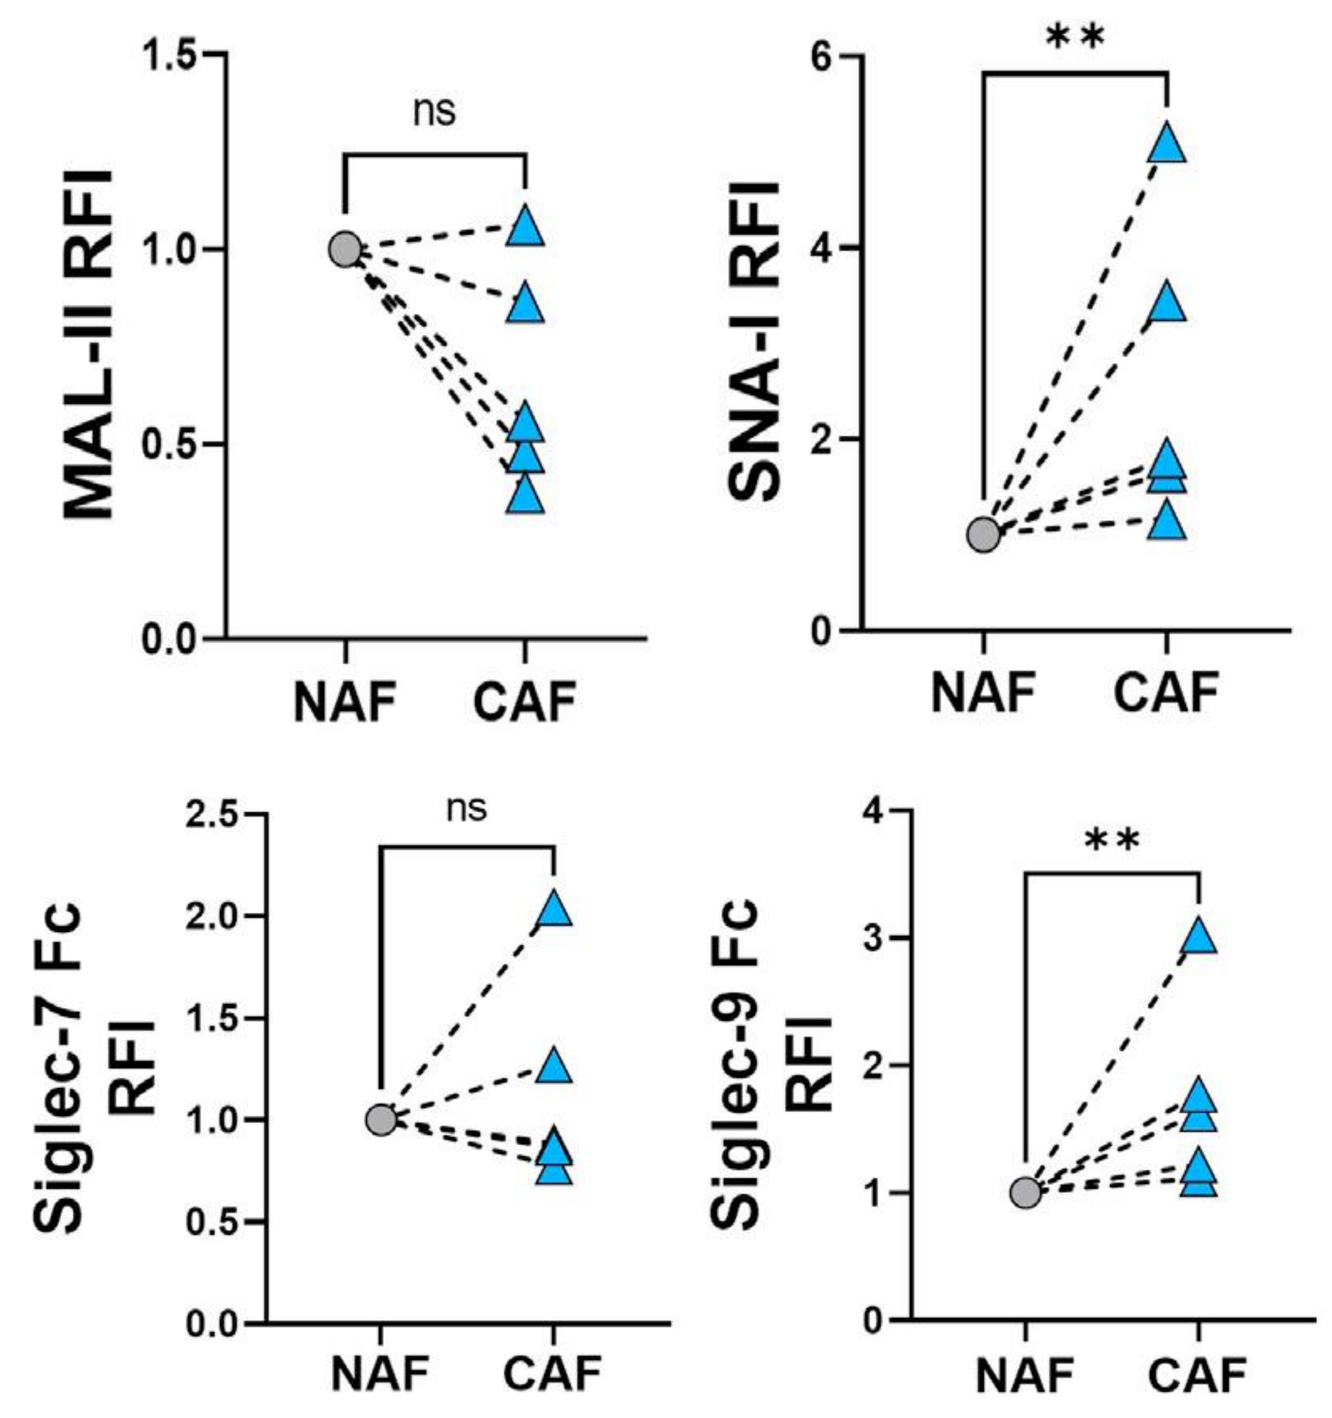
MAL-II RFI
SNA-I RFI
Siglec-7 Fc RFI
Siglec-9 Fc RFI
ns
**
NAF
CAF

Figure 3. CRC tumor-derived CAFs have elevated levels of sialic acid expression. For cell surface characterization, $5x10^4$ NAFs or CAFs were incubated with specific lectins (biotinylated Maackia Amurensis Lectin II (MAL-II) and biotinylated Sambucus Nigra Lectin (SNA-I)) or Siglec-Fc chimeras (Siglec-7 and Siglec-9) where indicated. **p < 0.01 using nonparametric Mann-Whitney test. n = 5 biological replicates.

CD3+ T cells reside in the stroma of colorectal tumors

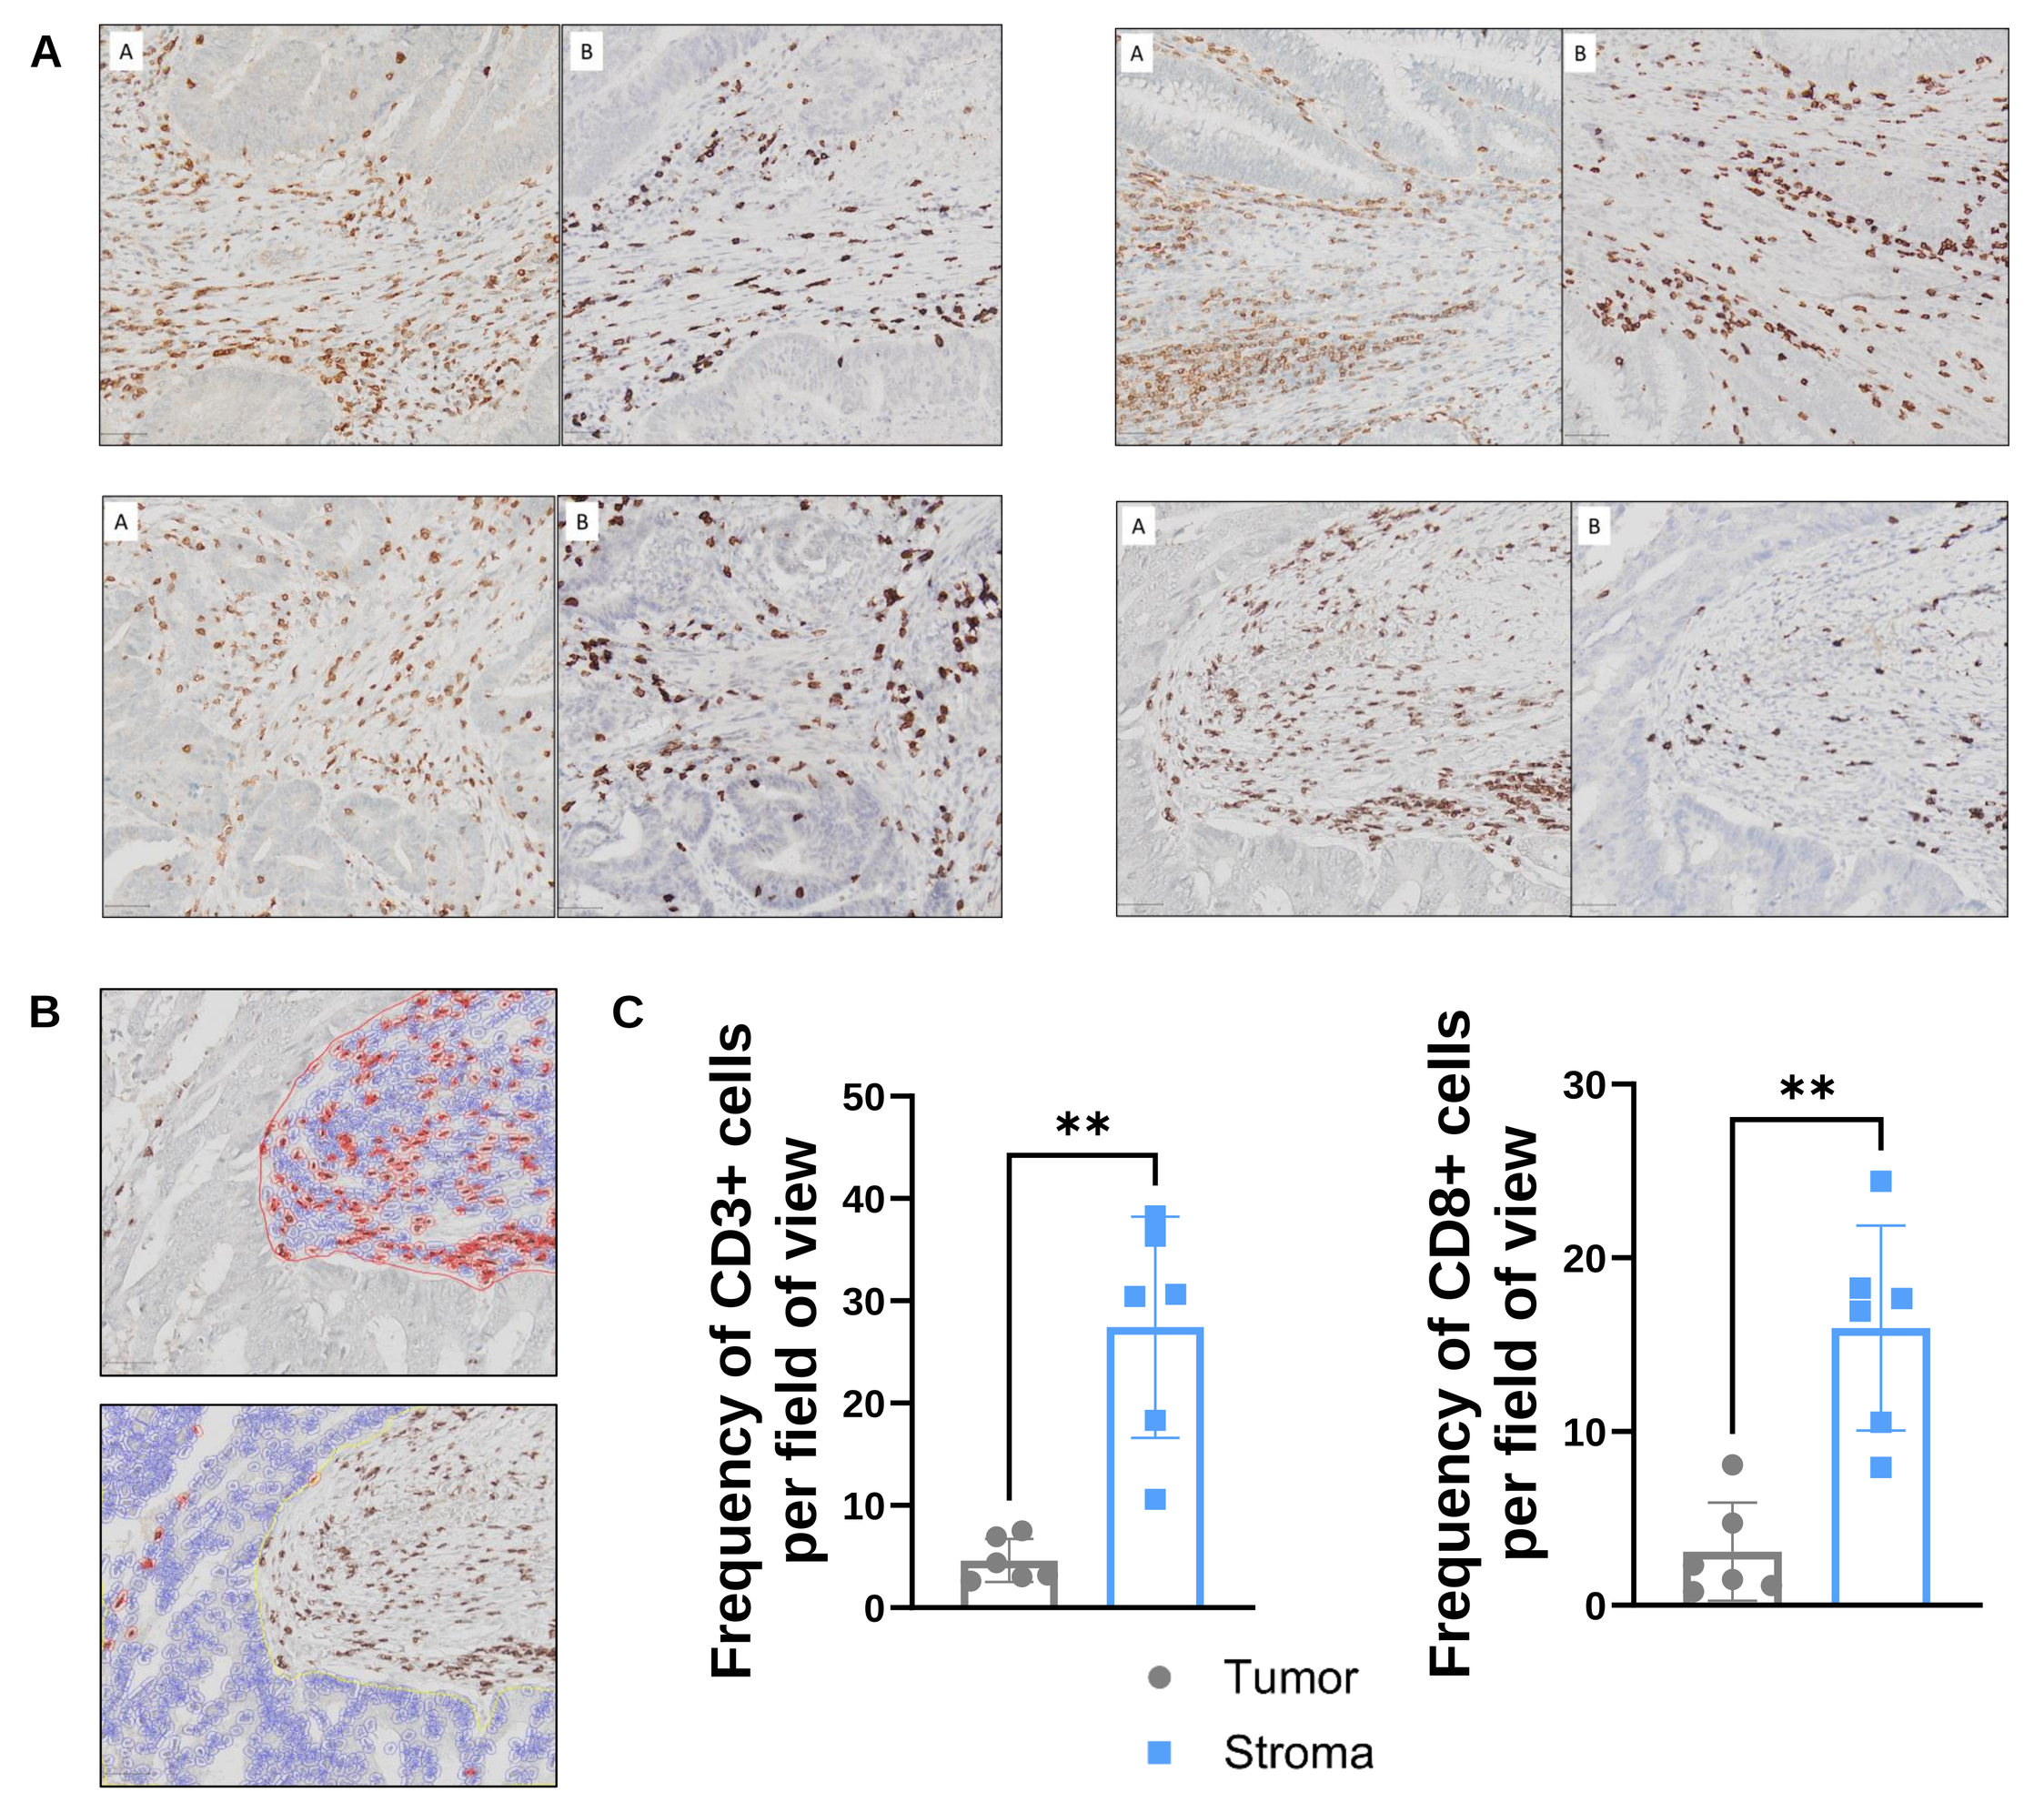
A
B
C
Frequency of CD3+ cells per field of view
Frequency of CD8+ cells per field of view
**
Tumor
Stroma

Figure 4. CRC tumor-associated stroma has higher T cell infiltration. (A) Representative IHC stained sections of six individual CRC patient tumors. Sections in A are stained with anti-CD3 (DAB-brown); sections in B are stained with anti-CD8 (DAB-brown) (n = 6). Nuclei were counterstained with hematoxylin. (B) Representative section quantifying CD3+ cells using QuPath software for both stroma (top panel) and tumor (bottom panel). Positive cells are detected with a red outline and negative cells are blue. Magnification for A and B = 200X; scale bar, 50 µm. (C) Scatter bar graphs showing frequency (%) of CD3+ cells (left) and CD8+ T cells (right) per field of view. Data are mean ± SD; **p < 0.01 using nonparametric Mann-Whitney test.

Sialidase pre-treatment of CAFs reduced the frequency of PD-1/Siglec-9-expressing CD8 T cells

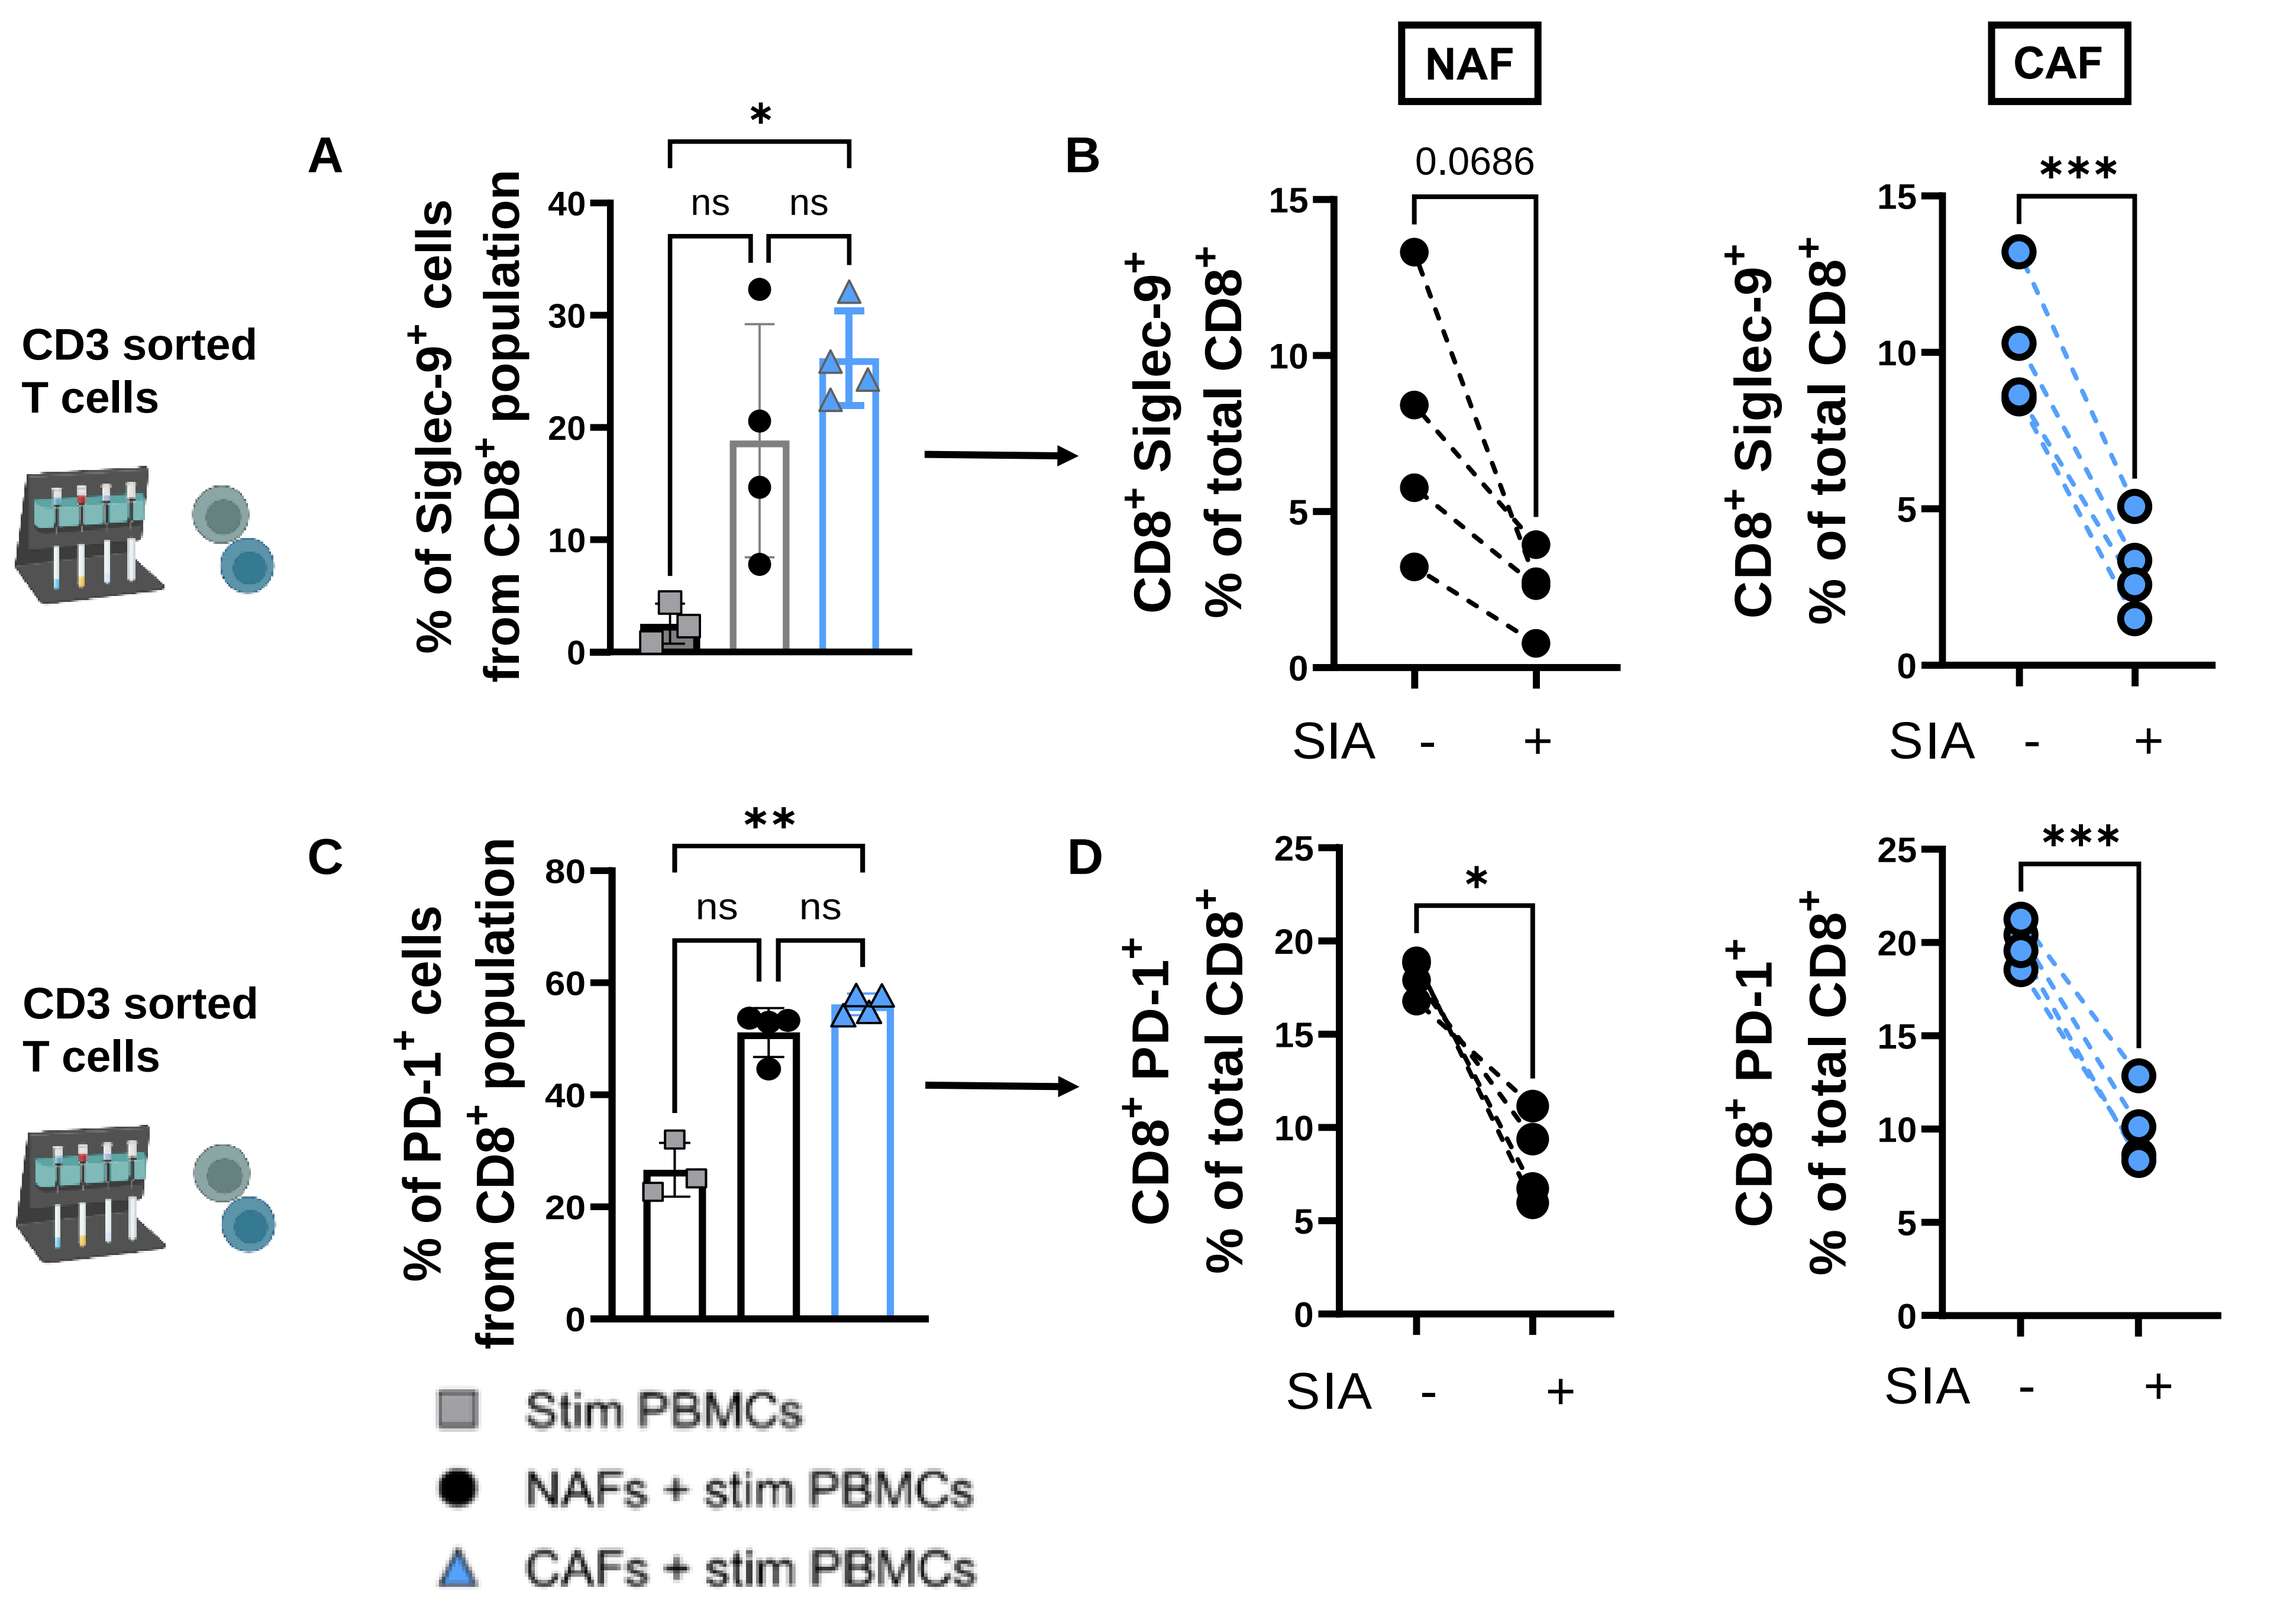
NAF
CAF
A
B
C
D
CD3 sorted T cells
% of Siglec-9+ cells from CD8+ population
CD8+ Siglec-9+ % of total CD8+
% of PD-1+ cells from CD8+ population
CD8+ PD-1+ % of total CD8+
0.0686

SIA - +
Stim PBMCs
NAFs + stim PBMCs
CAFs + stim PBMCs

Figure 5. CAFs induce a sialylation-dependent exhausted phenotype in CD8+ T cells. (A) Frequency (%) of CD3-sorted CD8+Siglec-9+ T cells after co-culture with NAFs or CAFs. (B) Frequency (%) of CD3-sorted CD8+Siglec-9+ T cells expressed as a percentage of the parent (CD8+) population after co-culture with NAFs (left) or CAFs (right) pre-treated and cultured directly or not with SIA. (C) Frequency (%) of CD3-sorted CD8+PD-1+ T cells after co-culture with NAFs or CAFs. (D) Frequency (%) of CD3-sorted CD8+PD-1+ T cells expressed as a percentage of the parent (CD8+) population after co-culture with NAFs (left) or CAFs (right) pre-treated and cultured directly or not with SIA.

Conclusion

- Using a proprietary HYDRA platform to measure cell-associated sialoglycans, we showed that both tumor cells and associated stromal cells are hypersialylated in colorectal cancer.
- Targeting stromal sialylation with an engineered human sialidase reversed stromal cell-mediated immunosuppression in CAF/T cell co-cultures and may contribute to antitumor immunity by increasing activated and functional CD8+ T cells.
- We propose that targeting stromal cell sialylation and/or Siglec-Siglec ligand interactions reactivates T cell activation and may represent an innovative strategy to enhance anti-tumor immunity in immunosuppressive TMEs.
- An engineered human sialidase, being evaluated in a Phase 1/2 trial (NCT005259696) for patients of advanced solid tumors, could be potentially applied to target stromal cell desialylation.

Fondúireacht Eolaíochta Éireann
Science Foundation Ireland
SFI Research Centre for Medical Devices
Society for Immunotherapy of Cancer