Investigating the role of Extracellular Vesicles in CMS4 colorectal tumour immunomodulation

Contact: a.walsh131@universityofgalway.ie
@AnastasiiijaW
linkedin.com/in/anastasija-walsh

SCAN ME

Anastasija Walsh[1], Lei Lei[1], Norashikin Zakaria[1], Karolina Salciute[3], Koyena Pal[1], Amy O'Connor[1], Roisin Dwyer[2], Aideen Ryan[1]

[1]Discipline of Pharmacology & Therapeutics, School of Medicine, University of Galway, Ireland
[2]Discipline of Surgery, School of Medicine, University of Galway, Ireland
[3]Discipline of Anatomy, School of Medicine, University of Galway, Ireland

Introduction

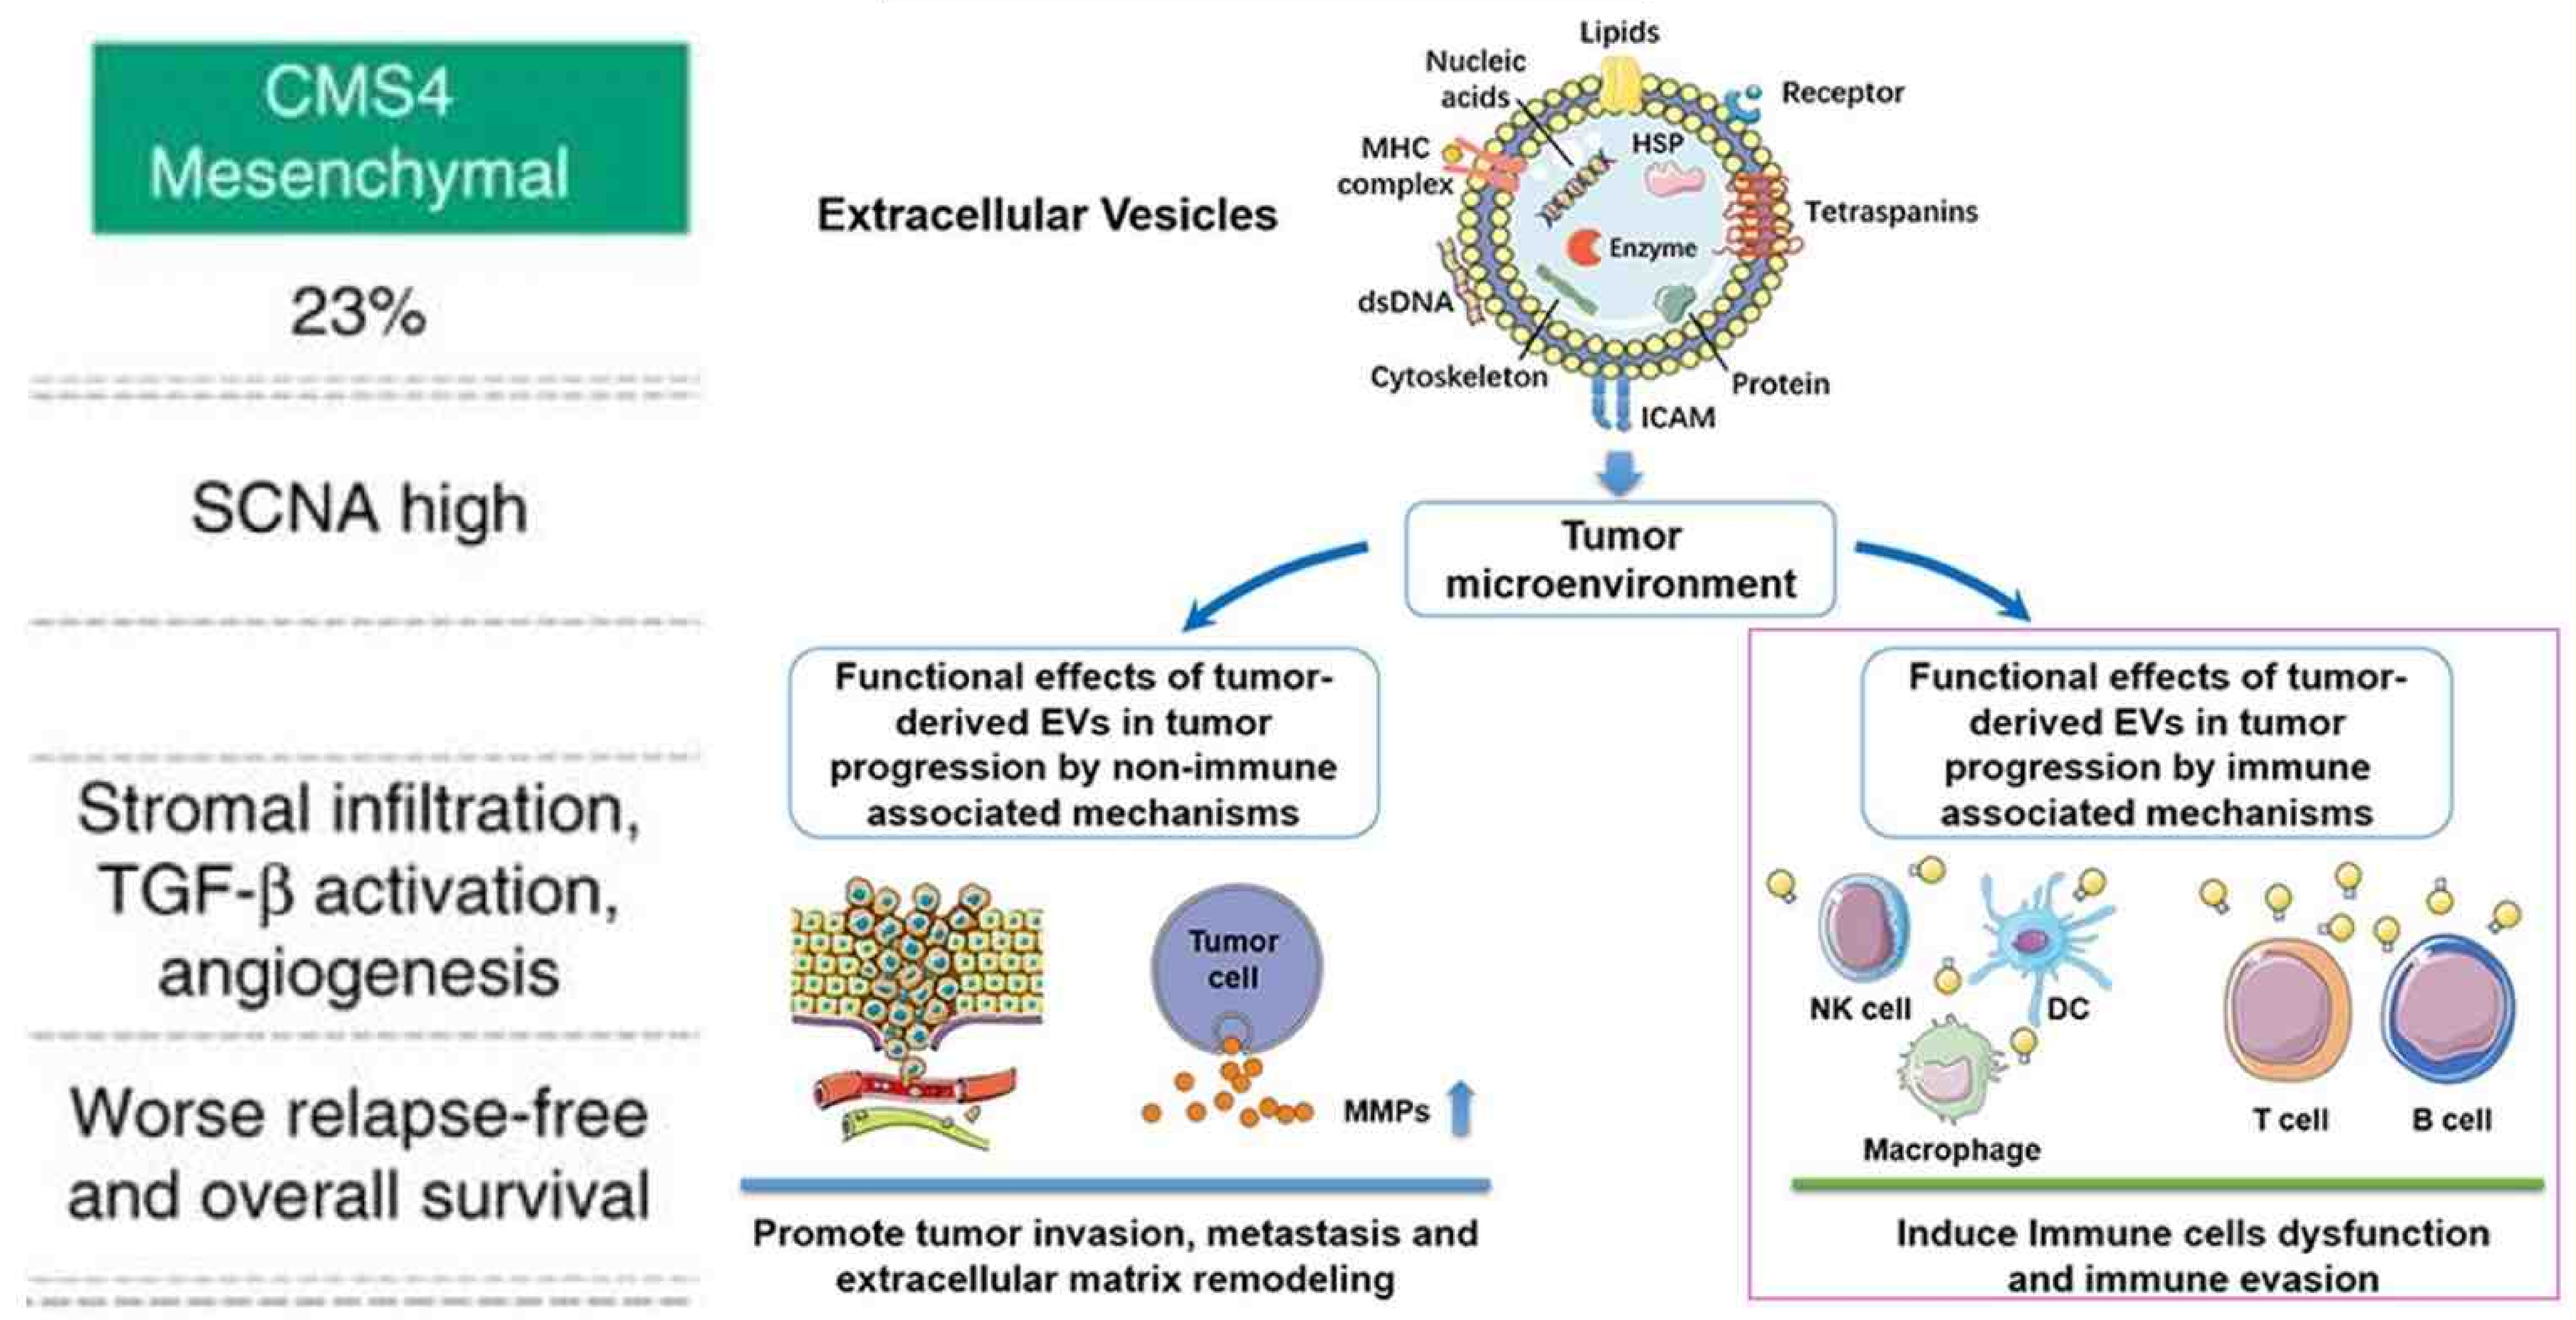

CMS4 Mesenchymal
23%
SCNA high
Stromal infiltration, TGF-β activation, angiogenesis
Worse relapse-free and overall survival
Extracellular Vesicles
Lipids
Nucleic acids
Receptor
MHC complex
HSP
Tetraspanins
Enzyme
dsDNA
Cytoskeleton
Protein
ICAM
Tumor microenvironment
Functional effects of tumor-derived EVs in tumor progression by non-immune associated mechanisms
Tumor cell
MMPs
Promote tumor invasion, metastasis and extracellular matrix remodeling
Functional effects of tumor-derived EVs in tumor progression by immune associated mechanisms
NK cell
DC
Macrophage
T cell
B cell
Induce Immune cells dysfunction and immune evasion

CMS4 colorectal cancer (CRC) is highly immunosuppressive and characterized by high levels of TGF-β signalling and stromal invasion, leading to a poor prognosis.

Extracellular Vesicles (EVs) promote immunosuppression in the Tumour Microenvironment by influencing surrounding immune cells [1] [2]. I aim to elucidate their immunosuppressive mechanisms in CMS4 CRC.

Methods

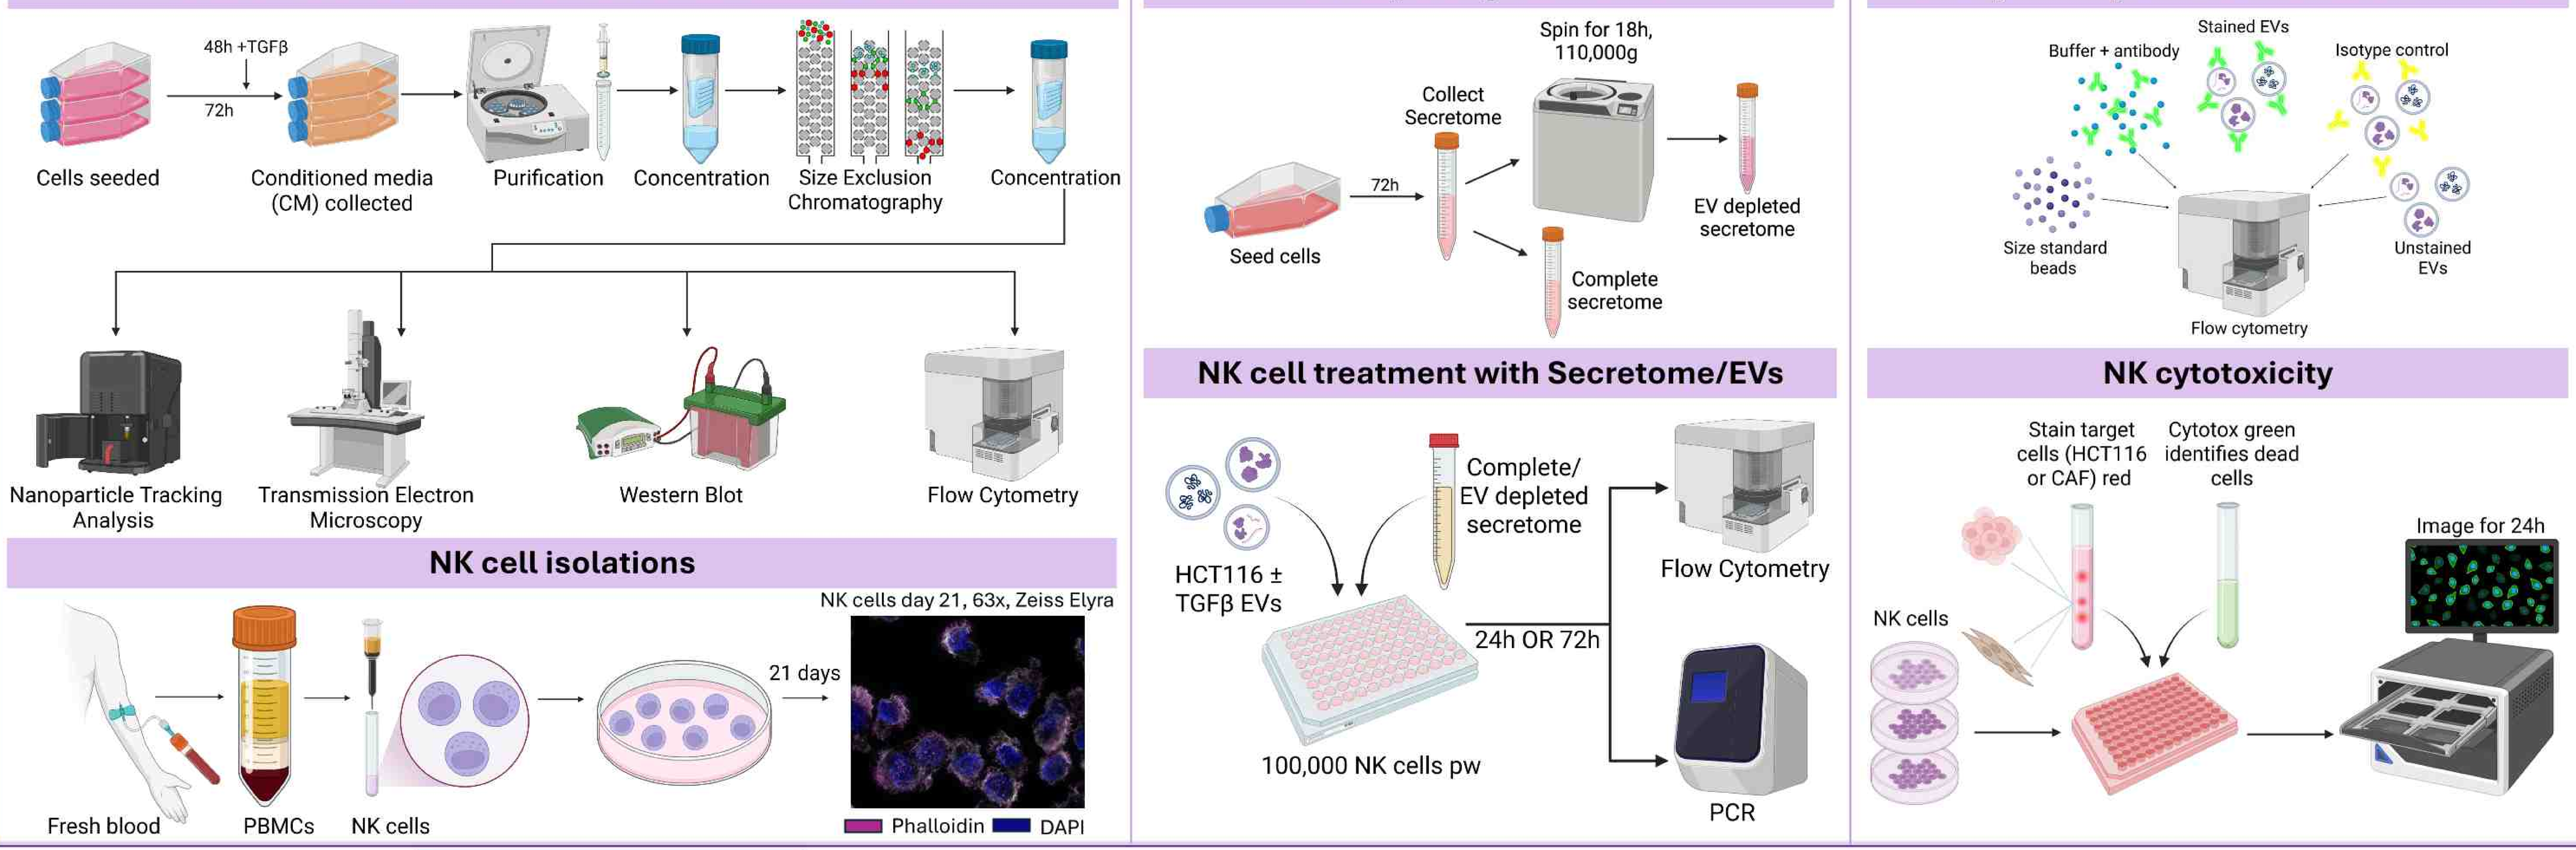

Isolation & characterisation of Extracellular Vesicles
48h +TGFβ
72h
Cells seeded
Conditioned media (CM) collected
Purification
Concentration
Size Exclusion Chromatography
Concentration
Nanoparticle Tracking Analysis
Transmission Electron Microscopy
Western Blot
Flow Cytometry
NK cell isolations
NK cells day 21, 63x, Zeiss Elyra
21 days
Fresh blood
PBMCs
NK cells
Phalloidin
DAPI
EV depleting secretome
Spin for 18h, 110,000g
Collect Secretome
72h
Seed cells
EV depleted secretome
Complete secretome
NK cell treatment with Secretome/EVs
HCT116 ± TGFβ EVs
Complete/ EV depleted secretome
24h OR 72h
100,000 NK cells pw
Flow Cytometry
PCR
Flow cytometry with Extracellular Vesicles
Stained EVs
Buffer + antibody
Isotype control
Size standard beads
Unstained EVs
Flow cytometry
NK cytotoxicity
Stain target cells (HCT116 or CAF) red
Cytotox green identifies dead cells
Image for 24h
NK cells

Results

Cancer Extracellular Vesicles express immunosuppressive ligands

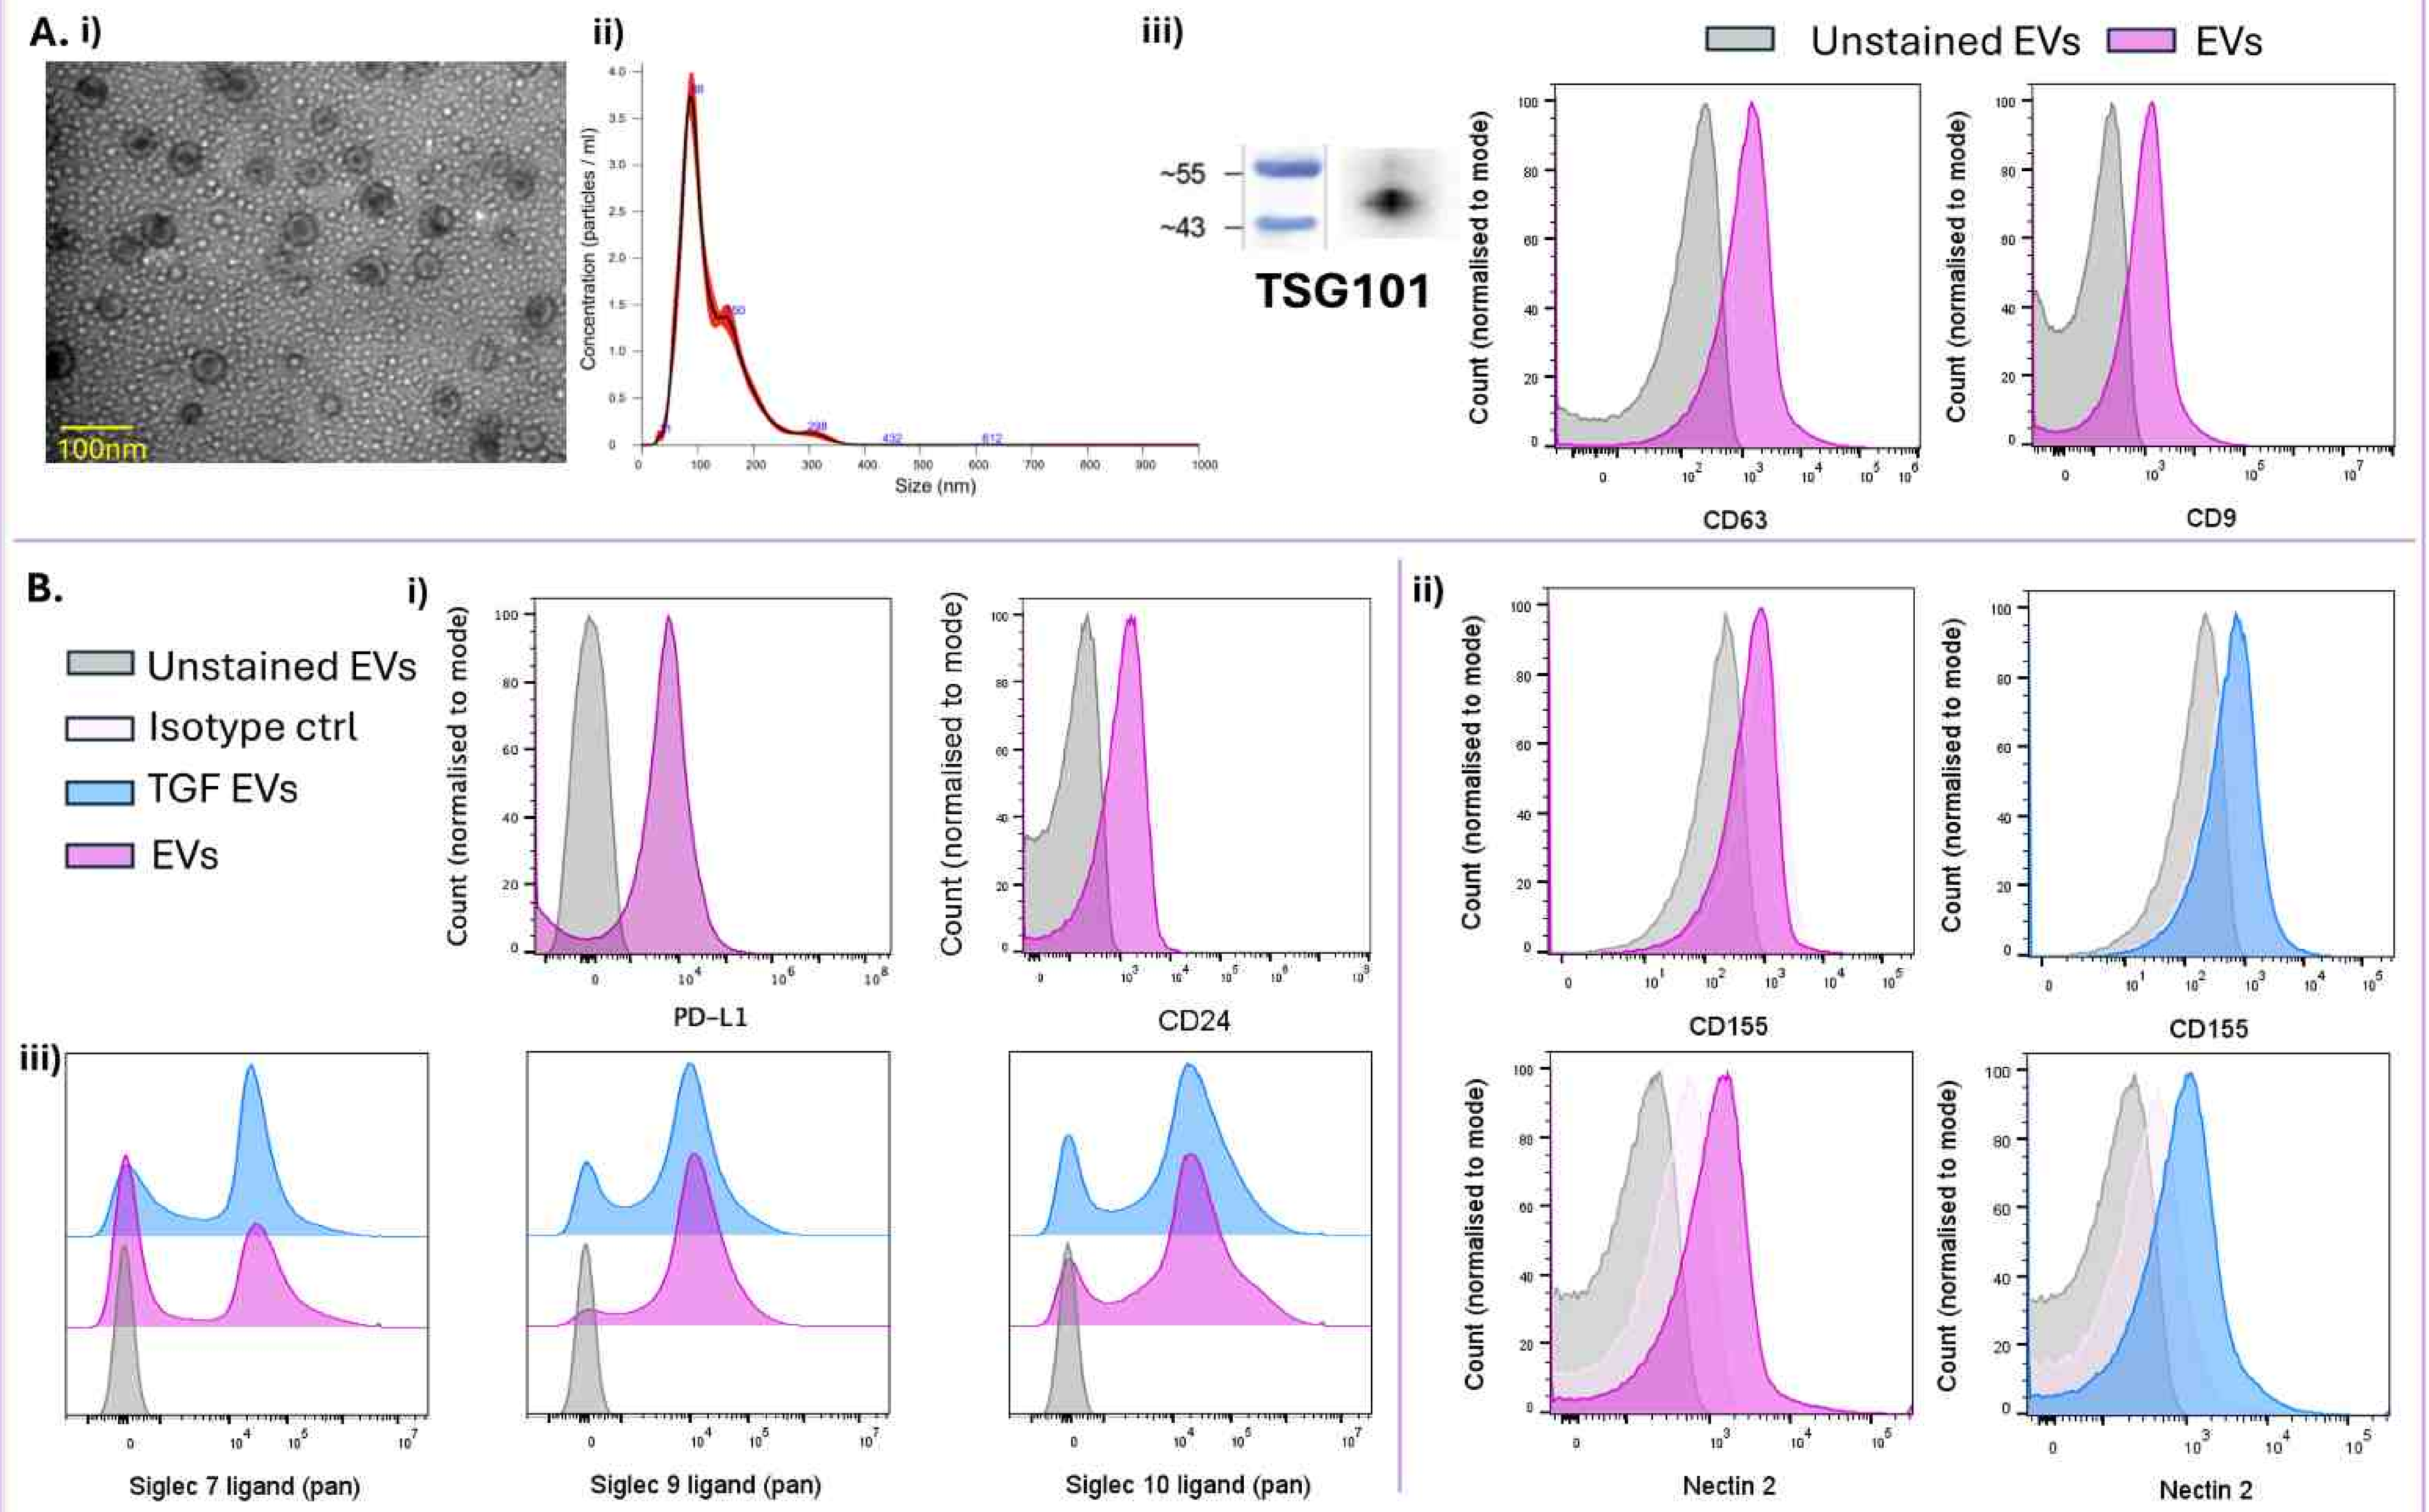

A. i)
100nm
ii)
Concentration (particles / ml)
Size (nm)
iii)
~55
~43
TSG101
Unstained EVs
EVs
Count (normalised to mode)
CD63
CD9
B.
Unstained EVs
Isotype ctrl
TGF EVs
EVs
i)
PD-L1
CD24
ii)
CD155
CD155
Nectin 2
Nectin 2
iii)
Siglec 7 ligand (pan)
Siglec 9 ligand (pan)
Siglec 10 ligand (pan)

Figure 1. A. HCT116 EV characterisation. i) Transmission Electron Microscopy, image 120000X. ii) EV modal size, 88 nm. iii) Intraluminal marker TSG101 & tetraspanin CD9 & CD63 expression stained vs relevant unstained EVs (grey). **B. HCT116 EVs are positive for immunomodulatory markers.** i) CD24 (Siglec 10 ligand) & PD-L1 expression, ii) CD155 (NK ligand for CD96/DNAM-1/TIGIT) & Nectin 2 (DNAM-1/TIGIT ligand) expression, iii) Pan Siglec 7, 9 & 10 ligand expression, count normalised to mode.

Cancer cell EVs modulate NK cell immune responses in the CMS4 microenvironment

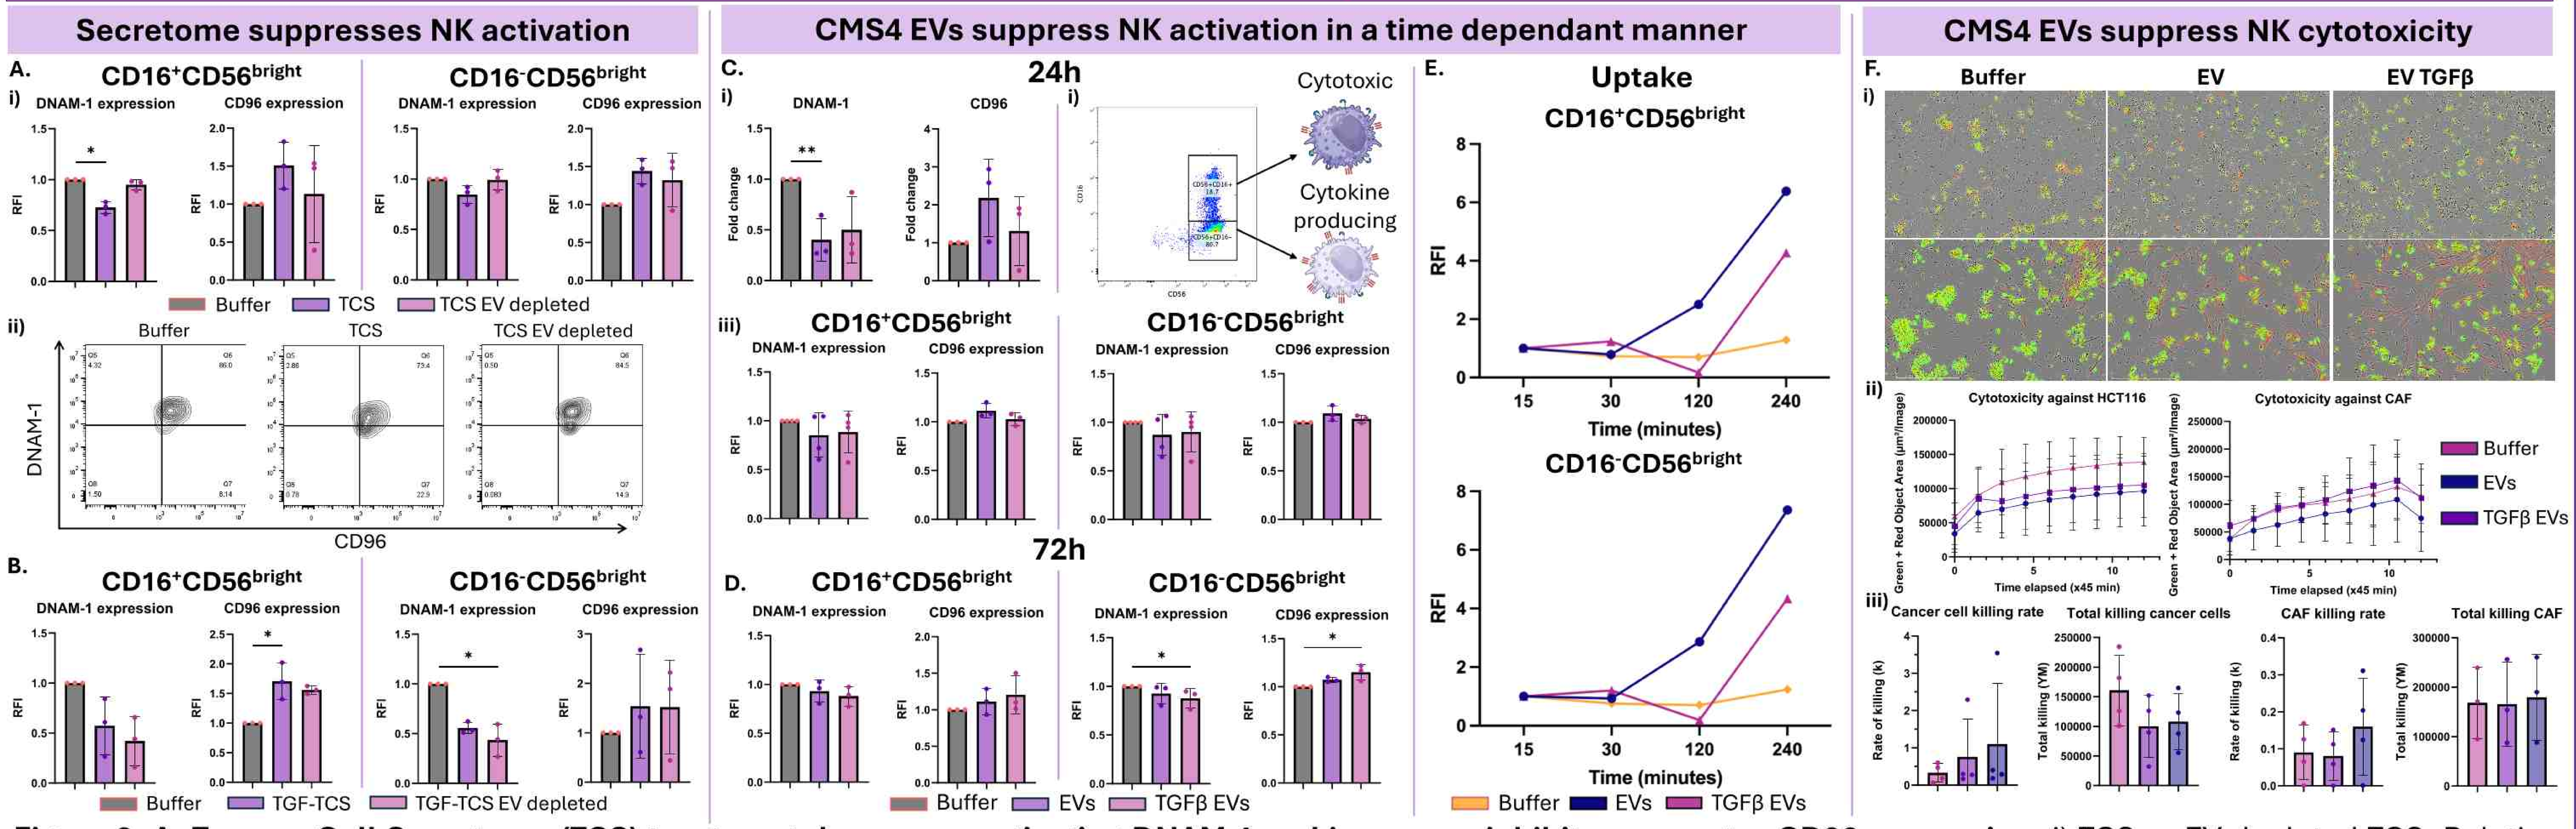

Secretome suppresses NK activation
A.
CD16+CD56bright
CD16-CD56bright
i)
DNAM-1 expression
CD96 expression
RFI
Buffer
TCS
TCS EV depleted
ii)
Buffer
TCS
TCS EV depleted
DNAM-1
CD96
B.
TGF-TCS
TGF-TCS EV depleted
CMS4 EVs suppress NK activation in a time dependant manner
C.
24h
i)
DNAM-1
CD96
Fold change
Cytotoxic
Cytokine producing
iii)
72h
D.
Buffer
EVs
TGFβ EVs
E.
Uptake
Time (minutes)
CMS4 EVs suppress NK cytotoxicity
F.
Buffer
EV
EV TGFβ
Cytotoxicity against HCT116
Cytotoxicity against CAF
Time elapsed (x45 min)
Cancer cell killing rate
Total killing cancer cells
CAF killing rate
Total killing CAF
Rate of killing (k)
Total killing (YM)

Figure 2. A. Tumour Cell Secretome (TCS) treatment decreases activating DNAM-1 and increases inhibitory receptor CD96 expression. i) TCS vs EV-depleted TCS; Relative Fluorescent Intensity (RFI), n=3; ii) Co-expression of DNAM-1/CD96. **B. EV depletion does not restore TGFβ-TCS–induced reduction of DNAM-1.** TGF-TCS vs EV-dep. TGF-TCS, RFI, n=3. **C. HCT116 EVs downregulate DNAM-1 and upregulate CD96 after 24 h** 1) mRNA fold change (2^-ΔΔCT), n=3, 2) NK subpopulation gating; iii) protein expression RFI, n=4. **D. TGFβ EVs are more suppressive at 72 h. E. TGFβ EVs are internalized slower than EVs.** CFSE-labelled EV vs TGFβ EV uptake, n=1. **F. EV treatment increases the NK-cell killing rate but reduces total killing capacity.** i) HCT116 (top, 16 h) and CAF (bottom, 20 h) targets; ii) dead targets over time; iii) killing rate (k) and total killing (Y_m), nonlinear regression, n=3/4. *Stats:* Kruskal–Wallis test, $p < 0.05$ (*), $p < 0.001$ (**).

Conclusion

- CMS4 cancer cell (HCT116) EVs express immunosuppressive ligands.
- Immunomodulatory effect of CMS4 TCS partially depends on the presence of EVs. TGFβ changes Secretome dynamics.
- CMS4 tumour cell EV treatment suppresses NK cell activation & cytotoxicity in a time sensitive manner

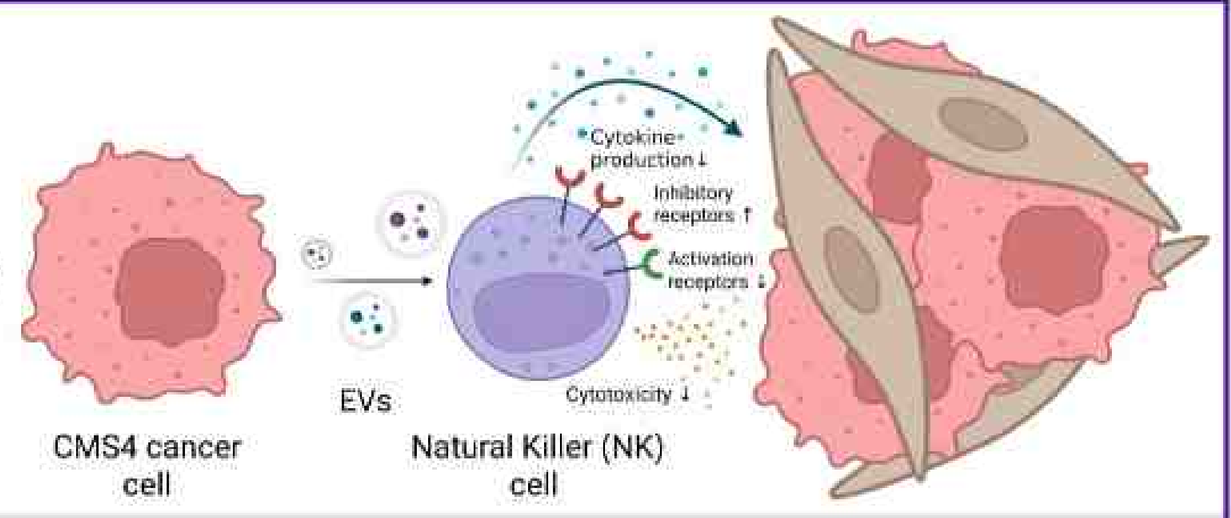

CMS4 cancer cell
EVs
Natural Killer (NK) cell

References

[1] Yang, S., Wei, S. & Wei, F. Extracellular vesicles mediated gastric cancer immune response: tumor cell death or immune escape?. *Cell Death Dis* **15**, 377 (2024). https://doi.org/10.1038/s41419-024-06758-8

[2] Ghoshal, B., Jhunjhunwala, S. A game of hide-and-seek: how extracellular vesicles evade the immune system. *Drug Deliv. and Transl. Res.* (2025). https://doi.org/10.1007/s13346-025-01789-w

Society for Immunotherapy of Cancer