E-688/HLX316: A First-in-Class B7-H3-Targeted Sialidase For Boosting Innate and Adaptive Anti-Tumor Immunity

PALLEON PHARMACEUTICALS
Henlius

Wayne Gatlin[1]*, Jen-Kuan Chang[2]*, Lizhi Cao[1], Hui Xu[1], Rishi Mehta[1], Maryann Timins[1], Mark Yang[1], Yanling Wang[2], Wilbert Tam[2], Grace Chung[2], Chunlei Ge[3], Lixin Feng[2], James Broderick[1], Li Peng[1]

*[1]Palleon Pharmaceuticals, Waltham, MA, USA, [2]Henlius USA, Milpitas, CA, USA, [3]Shanghai Henlius Biotech, China, *authors contributed equally to this work*

Abstract 7158

Hypersialylation is a Potent Mechanism of Cancer Immune Evasion and the Target for Therapeutic Sialidase

Abstract

Tumor hypersialylation is a driver of immune evasion that operates independently of the PD-1/PD-L1 axis. Tumors upregulate sialic acid-containing glycans (sialoglycans) on their surface, engaging inhibitory Siglec receptors on immune cells to actively suppress innate and adaptive anti-tumor immunity. E-688/HLX316 is a first-in-class targeted sialidase: an engineered human sialidase fused to an anti-B7-H3 VHH domain via an IgG1 Fc, designed to enzymatically remove immunosuppressive sialoglycans from the tumor while simultaneously directing antibody-mediated tumor killing. Targeting sialidase activity to B7-H3-expressing tumors yielded 5,000–10,000-fold improvement in desialylation potency compared to untargeted sialidase (E-612), with markedly extended durability of desialylation in vivo. E-688/HLX316 potentiated both ADCC and ADCP, enhancing NK cell- and macrophage-mediated tumor killing. In a humanized A375 xenograft mouse model, E-688/HLX316 achieved 87.5% tumor growth inhibition - outperforming untargeted sialidase (15%) and anti-PD-1 (38%) as a single agent. E-688/HLX316 was well tolerated in a GLP one-month repeat-dose NHP toxicology study, with a NOAEL of 150 mg/kg and no clinically significant findings. Many common tumor types have high B7-H3 expression and hypersialylation, including lung (NSCLC and SCLC), ovarian, and prostate. A Phase 1 dose escalation trial with ovarian cancer expansion is planned for 2026, supported by strong in vitro, in vivo, and GLP toxicology data.

KEY POINTS

- **B7-H3 targeting increases desialylation potency 5,000–10,000-fold and extends durability from ~2 days to 5–7 days compared to untargeted sialidase, addressing the key limitations identified in first-generation clinical experience**
- **E-688/HLX316 achieved 87.5% single-agent tumor growth inhibition in a humanized mouse model, outperforming anti-PD-1; wide safety margin confirmed in GLP NHP toxicology studies**
- **E-688/HLX316 enters Phase 1 dose escalation in 2026 in platinum-resistant ovarian cancer, an indication with high B7-H3 and hypersialylation co-expression, significant unmet need, and no approved IO therapy**

Tumor Hypersialylation: A Distinct and Addressable Axis of Immune Evasion

- Tumors upregulate sialic acid-containing glycans (sialoglycans) on their surface, engaging inhibitory Siglec receptors on immune cells to actively suppress innate and adaptive anti-tumor immunity
- Tumor hypersialylation correlates with poor outcomes across >50 independent studies and drives resistance to IO, chemotherapy, and radiotherapy
- Siglec-mediated immune suppression is sialic acid-dependent; the 14 Siglec receptors each bind multiple sialoglycan ligands, creating a redundant suppressive network that renders conventional receptor/ligand blocking insufficient
- Enzymatic cleavage of terminal sialic acid residues by a tumor-targeted sialidase disrupts all Siglec-ligand interactions simultaneously — the most rational approach to overcoming this redundancy

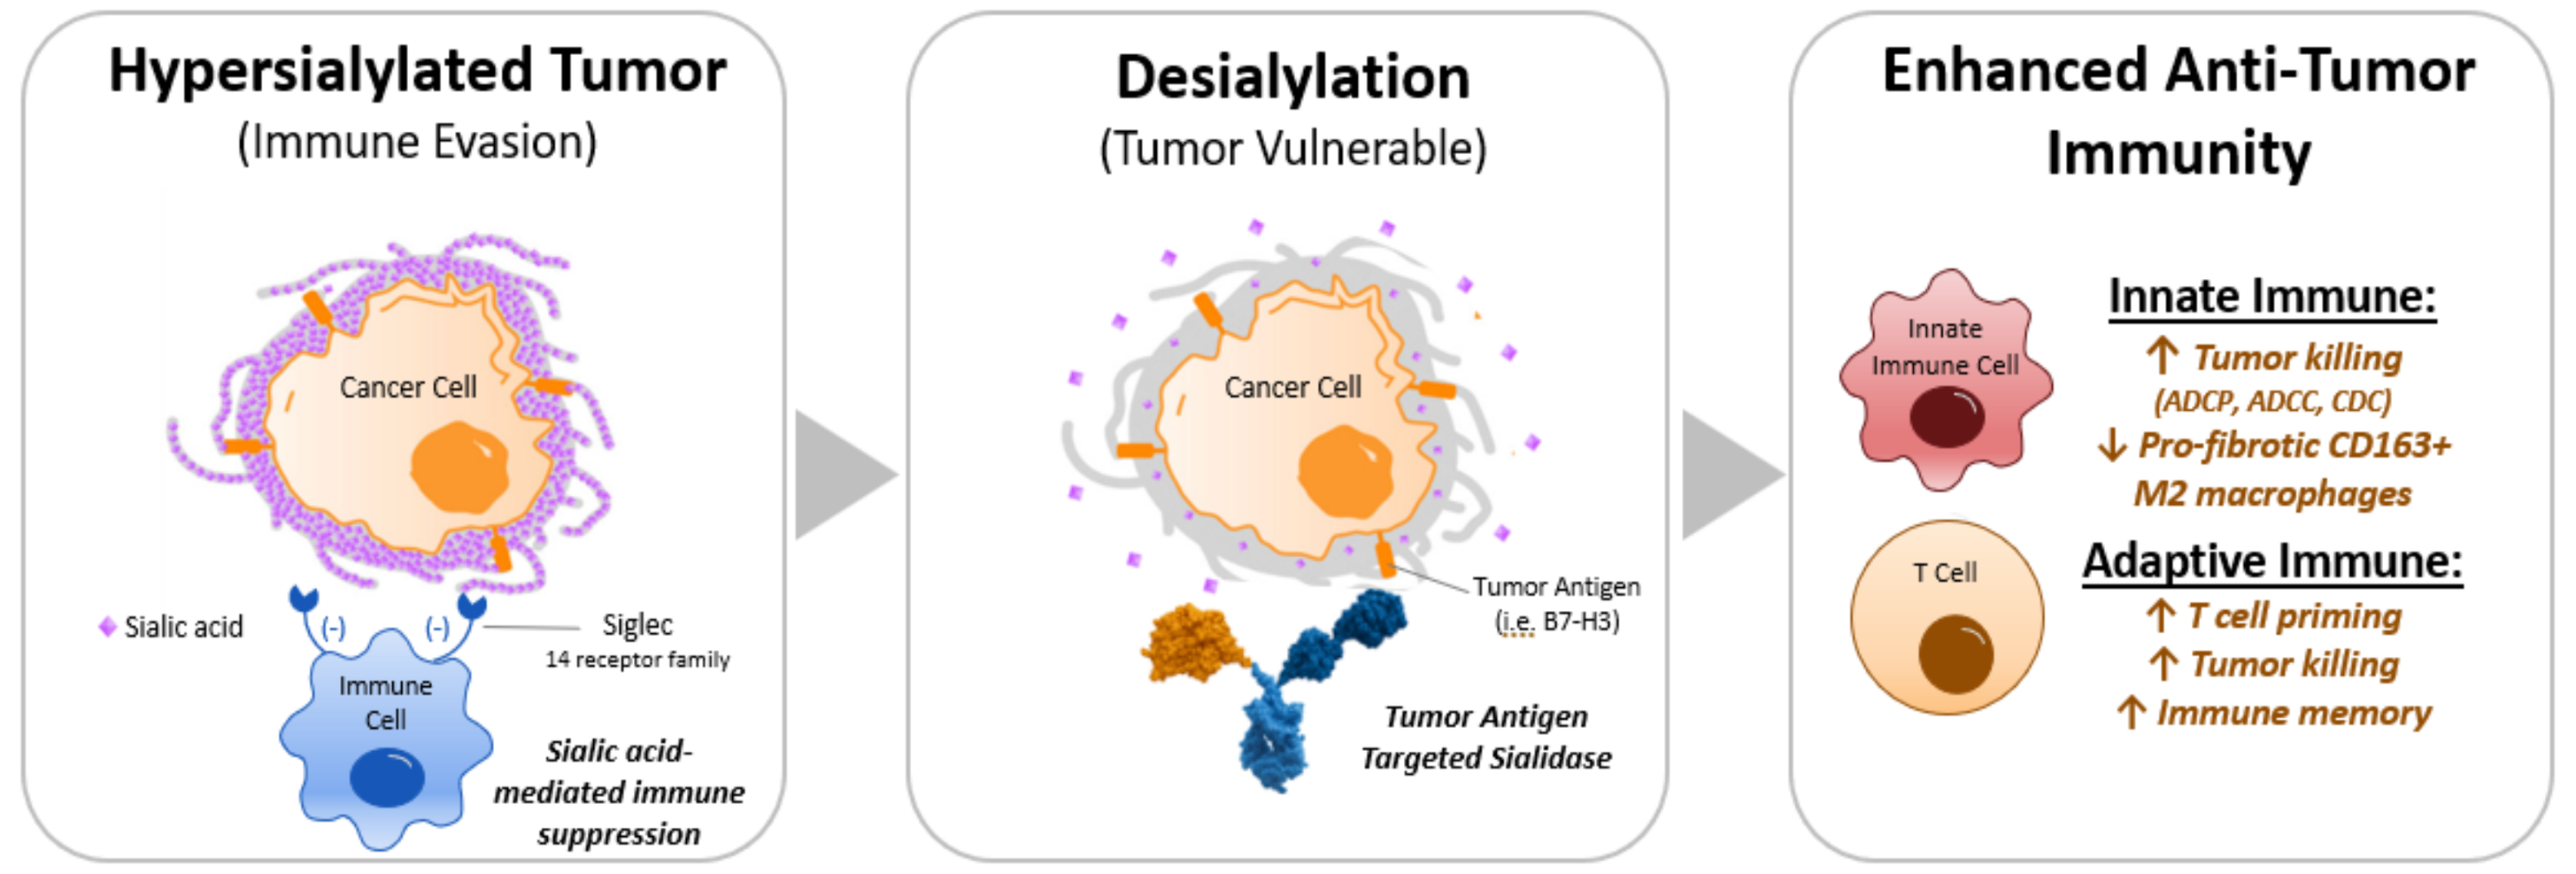
Hypersialylated Tumor
(Immune Evasion)
Cancer Cell
Sialic acid
Siglec
14 receptor family
Immune Cell
Sialic acid-mediated immune suppression
Desialylation
(Tumor Vulnerable)
Cancer Cell
Tumor Antigen (i.e. B7-H3)
Tumor Antigen Targeted Sialidase
Enhanced Anti-Tumor Immunity
Innate Immune Cell
Innate Immune:
↑ Tumor killing
(ADCP, ADCC, CDC)
↓ Pro-fibrotic CD163+ M2 macrophages
T Cell
Adaptive Immune:
↑ T cell priming
↑ Tumor killing
↑ Immune memory

Therapeutic Sialidase: Proof-of-Mechanism Established, Next Generation Engineered to Address Key Limitations

- Enzymatic removal of terminal sialic acid residues overcomes Siglec-sialoglycan redundancy, restoring innate and adaptive anti-tumor immunity
 - E-602, an untargeted human sialidase-Fc dimer, demonstrated single-agent activity in multiple syngeneic tumor models and a wide safety window in non-human primates
 - First-in-human E-602 study in cancer confirmed mechanism: tumor desialylation, immune modulation (reduced CD163+ M2 macrophages), and early efficacy in hypersialylated patients
- Key limitations in cancer identified: ~2-day desialylation durability insufficient to sustain immune activation, and no ADCC/ADCP functionality; drug repositioned for autoimmunity (to reduce CD163+ M2 macrophages)
- E-688/HLX316: B7-H3 targeting concentrates sialidase in the TME, extending desialylation to 5–7 days and increasing potency 5,000–10,000-fold; functional IgG1 Fc adds ADCP/ADCC

E-688/HLX316: A First-in-Class Targeted Sialidase Coupling Tumor-Selective Desialylation with B7-H3-Directed Killing

E-688/HLX316 Uses Tandem VHH Domains to Direct Sialidase Activity to B7-H3-Expressing Tumors

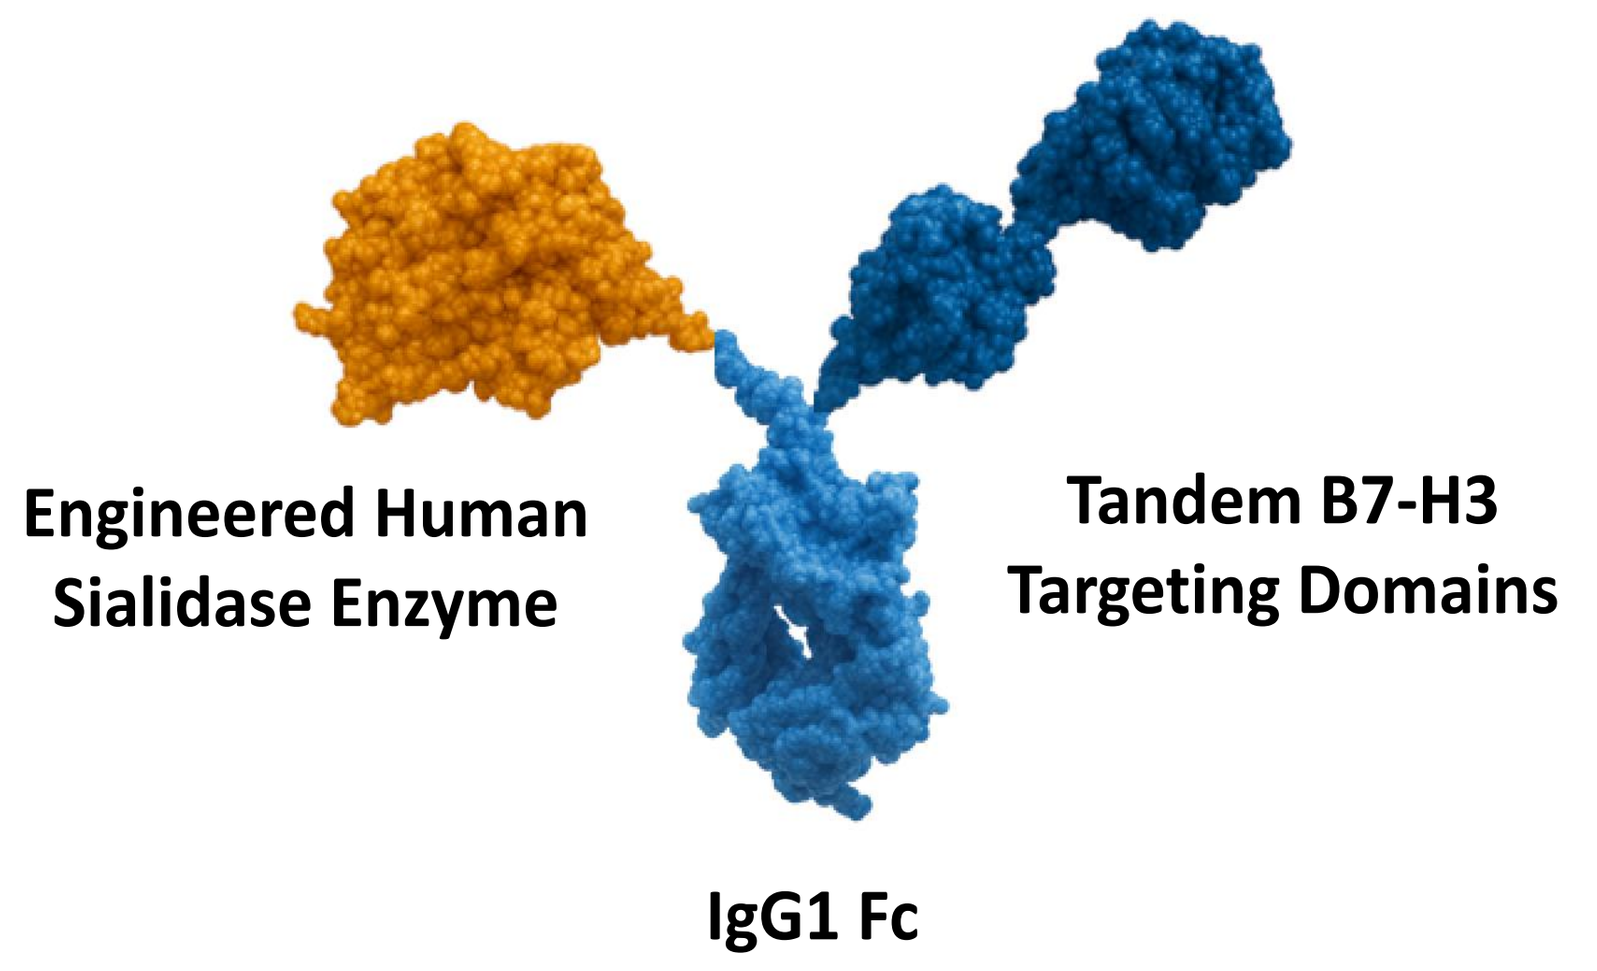
Engineered Human Sialidase Enzyme
Tandem B7-H3 Targeting Domains
IgG1 Fc

- B7-H3-directed delivery concentrates sialidase within the tumor microenvironment, maximizing desialylation depth and extending duration to 5-7 days
- Functional IgG1 Fc mediates direct tumor killing via ADCC and ADCP

E-688/HLX316 Achieves 5,000–10,000-Fold Greater Tumor Desialylation Potency Compared To Untargeted Sialidase

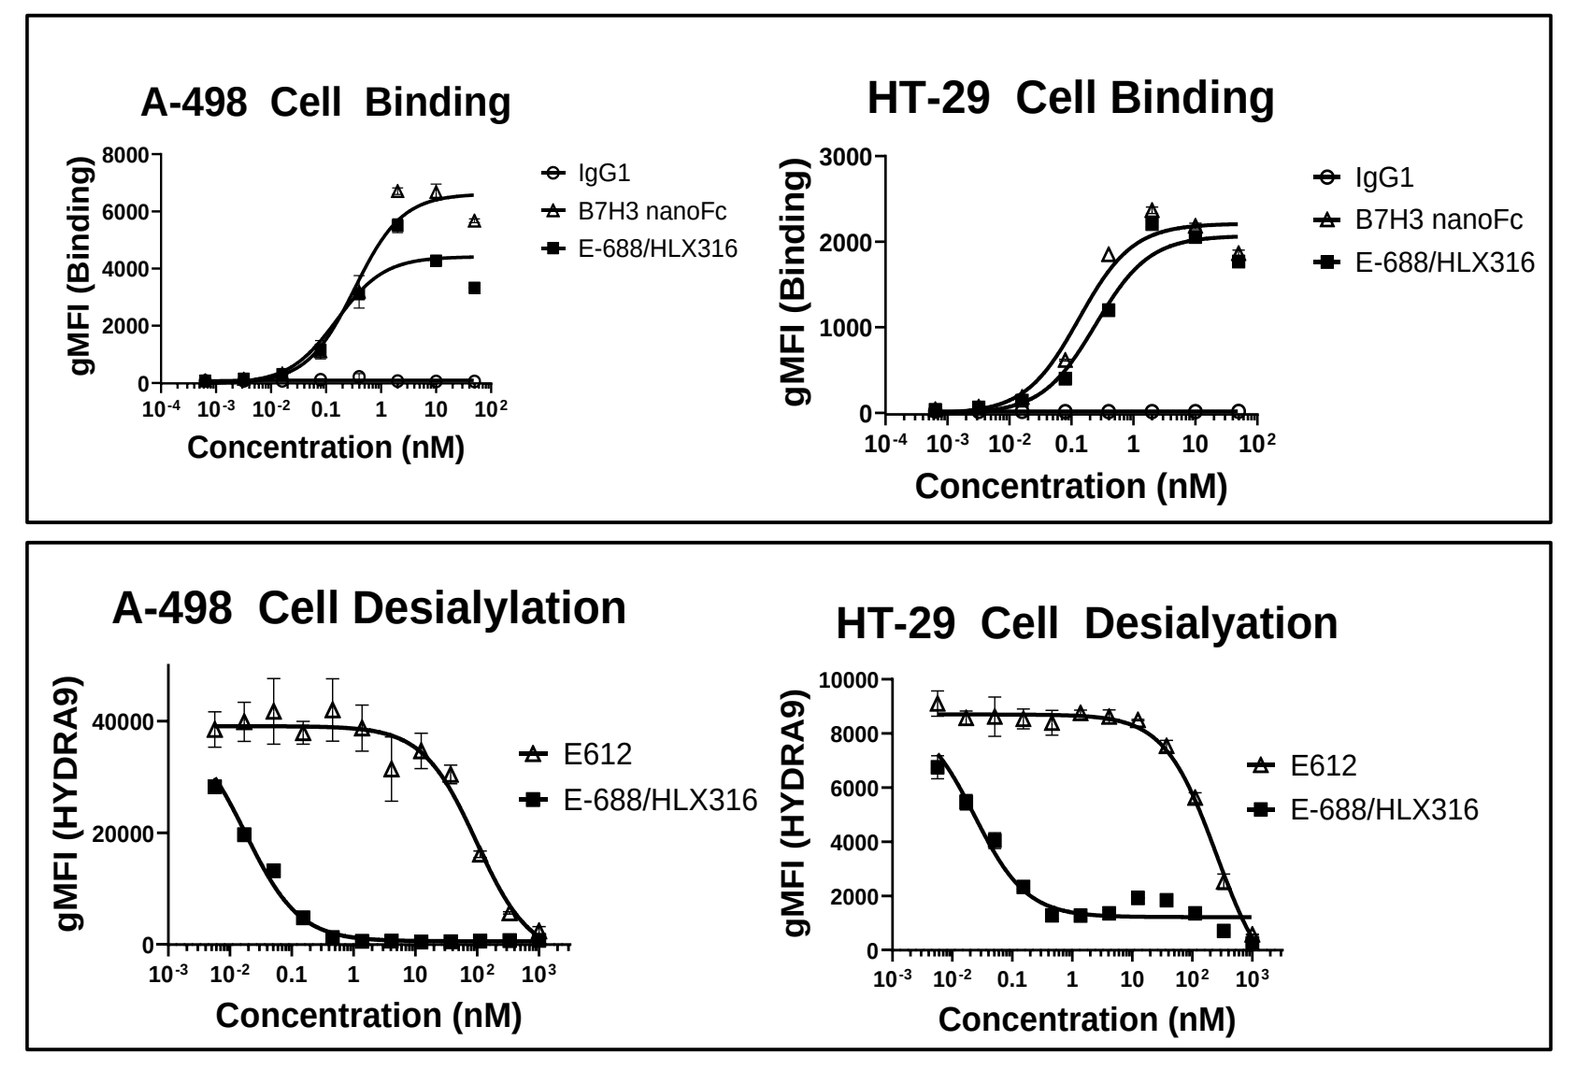
A-498 Cell Binding
HT-29 Cell Binding
gMFI (Binding)
Concentration (nM)
IgG1
B7H3 nanoFc
E-688/HLX316
A-498 Cell Desialylation
HT-29 Cell Desialylation
gMFI (HYDRA9)
E612
E-688/HLX316

- B7-H3 binding affinity is preserved relative to bivalent VHH-Fc alone, confirming that sialidase incorporation does not compromise tumor target engagement
- Tumor-directed delivery drives 5,000–10,000-fold greater desialylation potency versus untargeted sialidase (E-612)

Figure 1: E-688/HLX316 achieves greater than 5,000-fold improvement in desialylation potency over untargeted sialidase E-612, while retaining comparable sub-nanomolar B7-H3 binding affinity. Top: E-688/HLX316 binds B7-H3-expressing A-498 and HT-29 cancer cell lines with EC50 values of 0.156 nM and 0.256 nM respectively — comparable to the bivalent anti-B7-H3 VHH-Fc homodimer, confirming no loss of binding from the single-arm format. Bottom: E-688/HLX316 desialylation EC50 values of 0.018 nM (A-498) and 0.023 nM (HT-29) represent greater than 5,000-fold improvement over untargeted sialidase E-612 (96.8 nM and 251.1 nM respectively).

B7-H3-Targeted Desialylation Enhances ADCC and ADCP In Vitro

- E-688/HLX316 significantly enhances NK cell-mediated ADCC killing of B7-H3-expressing tumor cell lines, confirmed across two independent healthy donors
- E-688/HLX316 promotes macrophage-mediated ADCP by both M1 and M2 subtypes, engaging pro-inflammatory and alternative activation states
- Enhanced ADCC and ADCP are sialidase-dependent, confirmed by loss-of-function control (E-688 LoF)

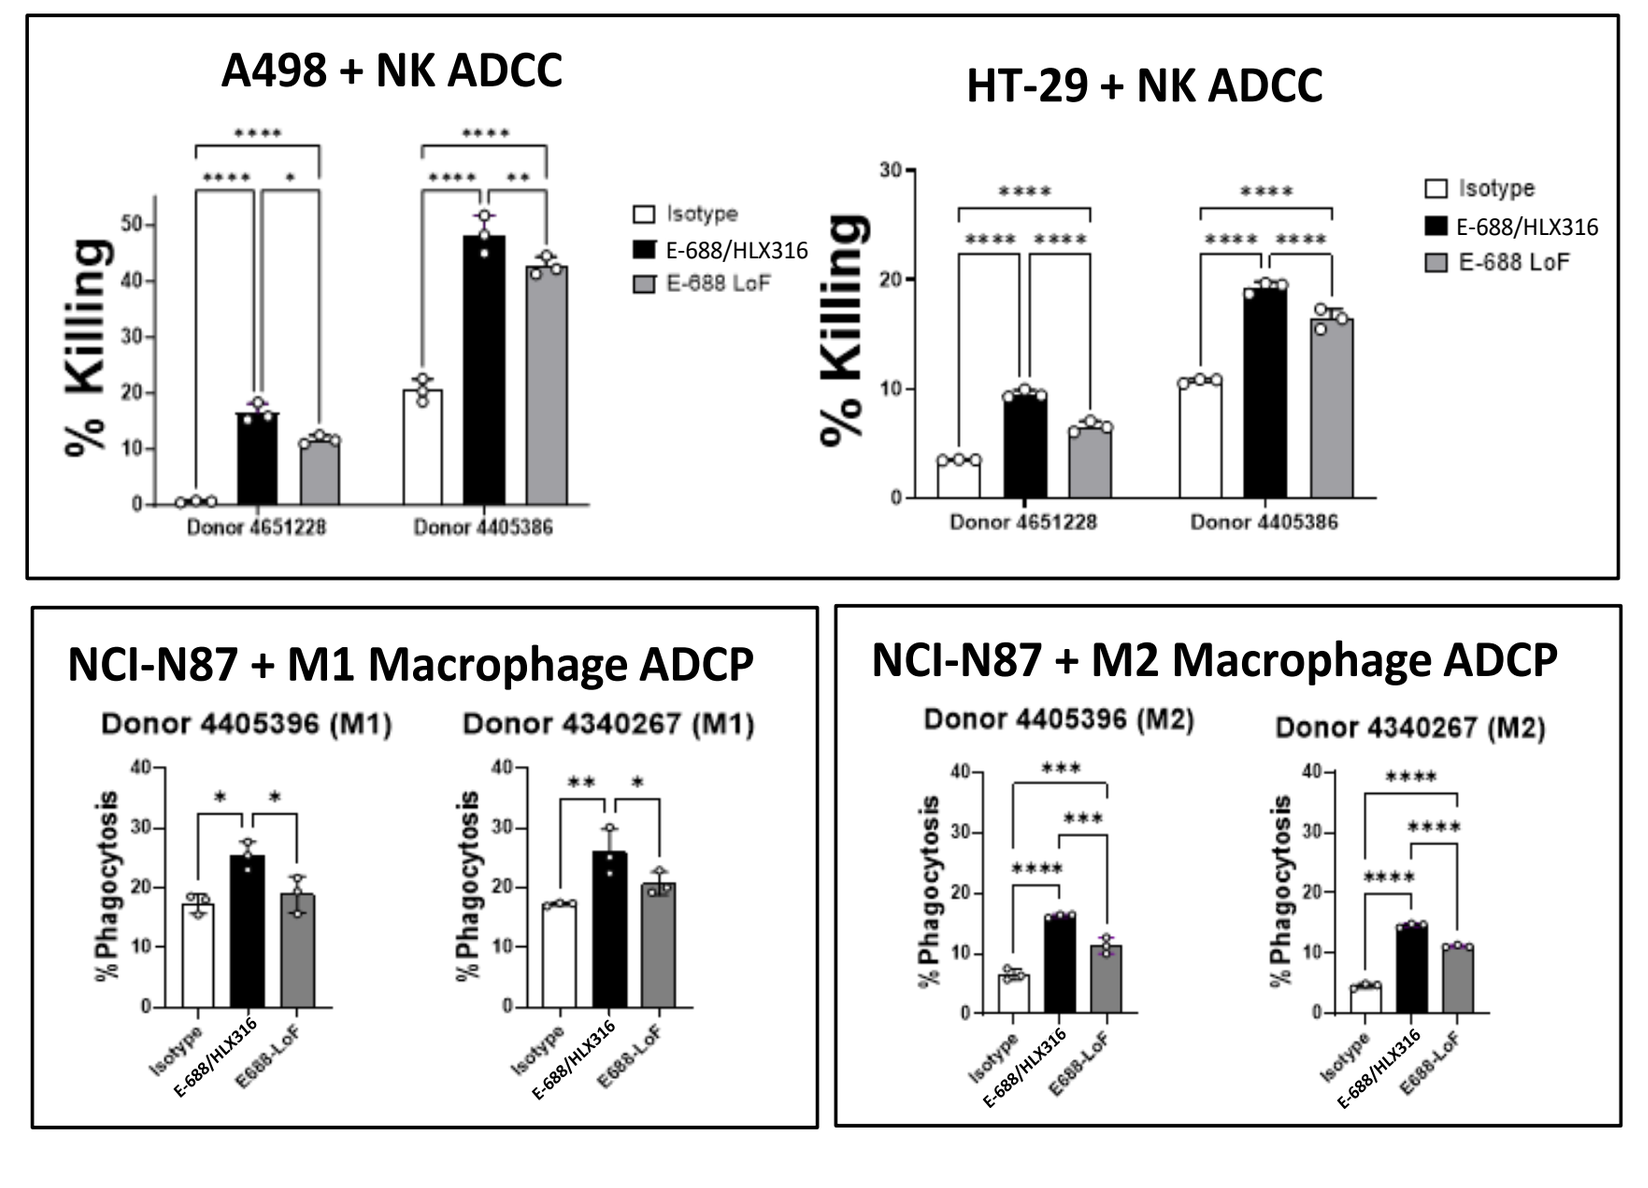
A498 + NK ADCC
HT-29 + NK ADCC
% Killing
Donor 4651228
Donor 4405386
Isotype
E-688/HLX316
E-688 LoF
NCI-N87 + M1 Macrophage ADCP
Donor 4405396 (M1)
Donor 4340267 (M1)
NCI-N87 + M2 Macrophage ADCP
Donor 4405396 (M2)
Donor 4340267 (M2)
% Phagocytosis
Isotype
E-688/HLX316
E688-LoF

Figure 2: E-688/HLX316 drives both NK cell-mediated ADCC and macrophage-mediated ADCP; loss-of-function of the sialidase arm (E-688 LoF) significantly reduces both activities, confirming that enzymatic desialylation is central to the mechanism of action — not antibody binding alone.
Top: E-688/HLX316 promotes statistically higher ADCC killing of A-498 and HT-29 tumor cells by NK cells from two healthy donors, reported as percentage of dead cells following treatment. Bottom: E-688/HLX316 promotes higher ADCP of NCI-N87 cells by monocyte-derived M1 and M2 macrophages from two healthy donors compared to E-688 LoF and isotype controls.

In Vivo Models Illustrate Efficacy in Mouse Models and Safety in GLP Cyno Monkey Toxicology Studies

E-688/HLX316 Demonstrates Single-Agent Efficacy Superior to Anti-PD-1 in Preclinical Tumor Models

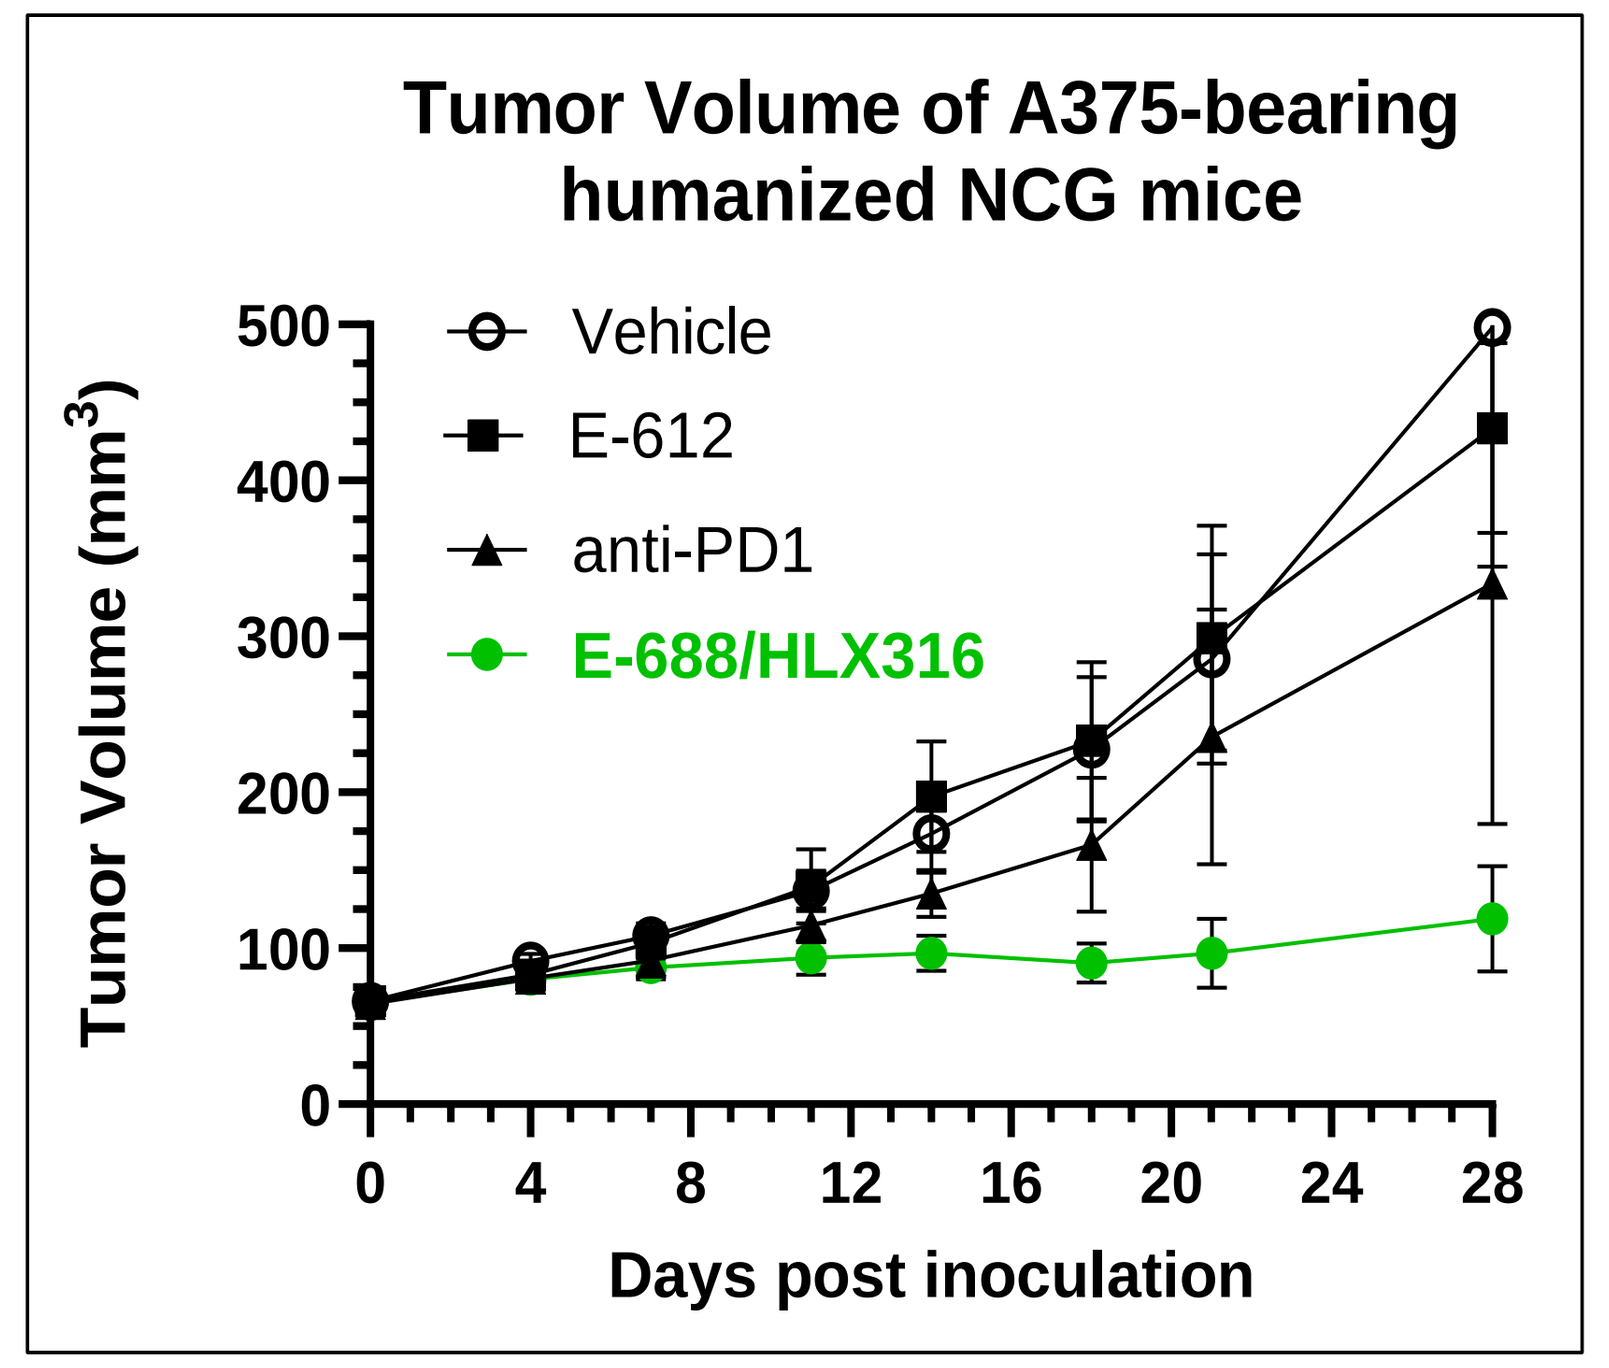
Tumor Volume of A375-bearing humanized NCG mice
Tumor Volume (mm³)
Vehicle
E-612
anti-PD1
E-688/HLX316
Days post inoculation

- In a humanized A375 xenograft model, E-688/HLX316 achieved 87.5% tumor growth inhibition (TGI) as a single agent — more than twice the TGI of anti-PD-1 in the same model
- Outperformed all comparators: untargeted sialidase E-612 (15% TGI) and anti-PD-1 (38% TGI)
- Single-agent efficacy confirmed across multiple additional tumor models (data not shown)

Figure 3 E-688/HLX316 achieves 87.5% tumor growth inhibition (TGI) as a single agent — outperforming anti-PD-1 (38%), and untargeted sialidase E-612 (15%) in a clinically relevant humanized mouse model. TGI was measured at Day 28 in an A375 xenograft model with human PBMC reconstitution in NCG mice and calculated as percent inhibition relative to vehicle control.

B7-H3 Targeting Dramatically Improves Potency and Durability of Tumor Desialylation Compared to an Untargeted Sialidase (E-612)

- E-688/HLX316 extends tumor desialylation to 5–7 days versus ~2 days with untargeted sialidase E-612 — directly addressing the key limitation identified in first-generation clinical experience
- Deeper and more sustained in vivo desialylation signal, consistent with the 5,000–10,000-fold in vitro potency advantage over E-612

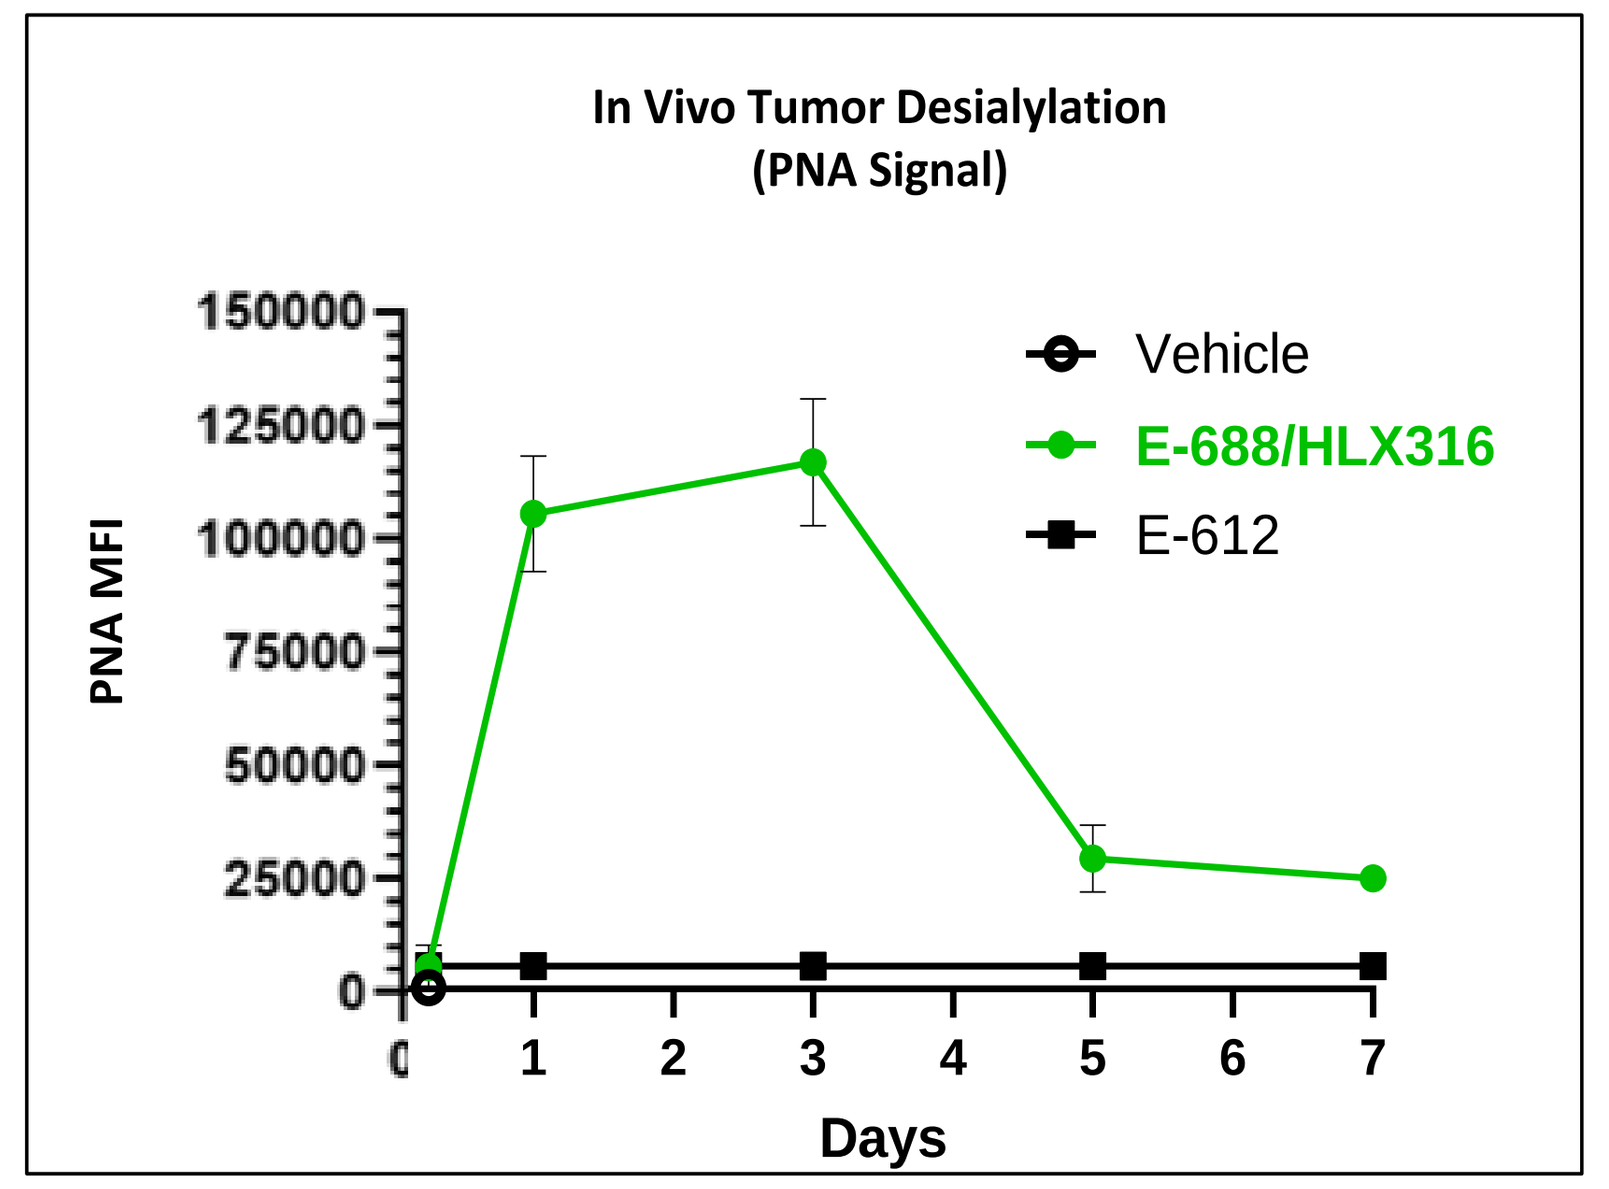
In Vivo Tumor Desialylation
(PNA Signal)
PNA MFI
Vehicle
E-688/HLX316
E-612
Days

Figure 4: E-688/HLX316 extends tumor desialylation durability to 5–7 days compared to untargeted sialidase E-612. Tumor desialylation was measured by PNA flow cytometry at Days 0, 1, 3, 5, and 7 in replicate mice bearing A498 tumors, and reported as fold-increase in desialylation signal over E-612.

E-688/HLX316 Demonstrates a Favorable Safety and Tolerability Profile

- First generation sialidase E-602 established human sialidase as a safe therapeutic modality: 90+ subjects dosed across two clinical trials (NCT05259696, NCT07038382)
- E-688/HLX316 was well tolerated in GLP one-month repeat-dose NHP toxicology study:
 - NOAEL 150 mg/kg/week — providing a 150-fold safety margin over the anticipated Phase 1 starting dose
 - Tissue cross-reactivity consistent with known B7-H3 expression profiles
 - No RBC hemolysis observed
 - Cytokine release profile consistent with sialidase mechanism of action

E-688/HLX316 Phase 1 Clinical Trial With Expansion to Ovarian Cancer is Planned for 2026

B7-H3 and Hypersialylation Are Broadly Co-Expressed Across Solid Tumors

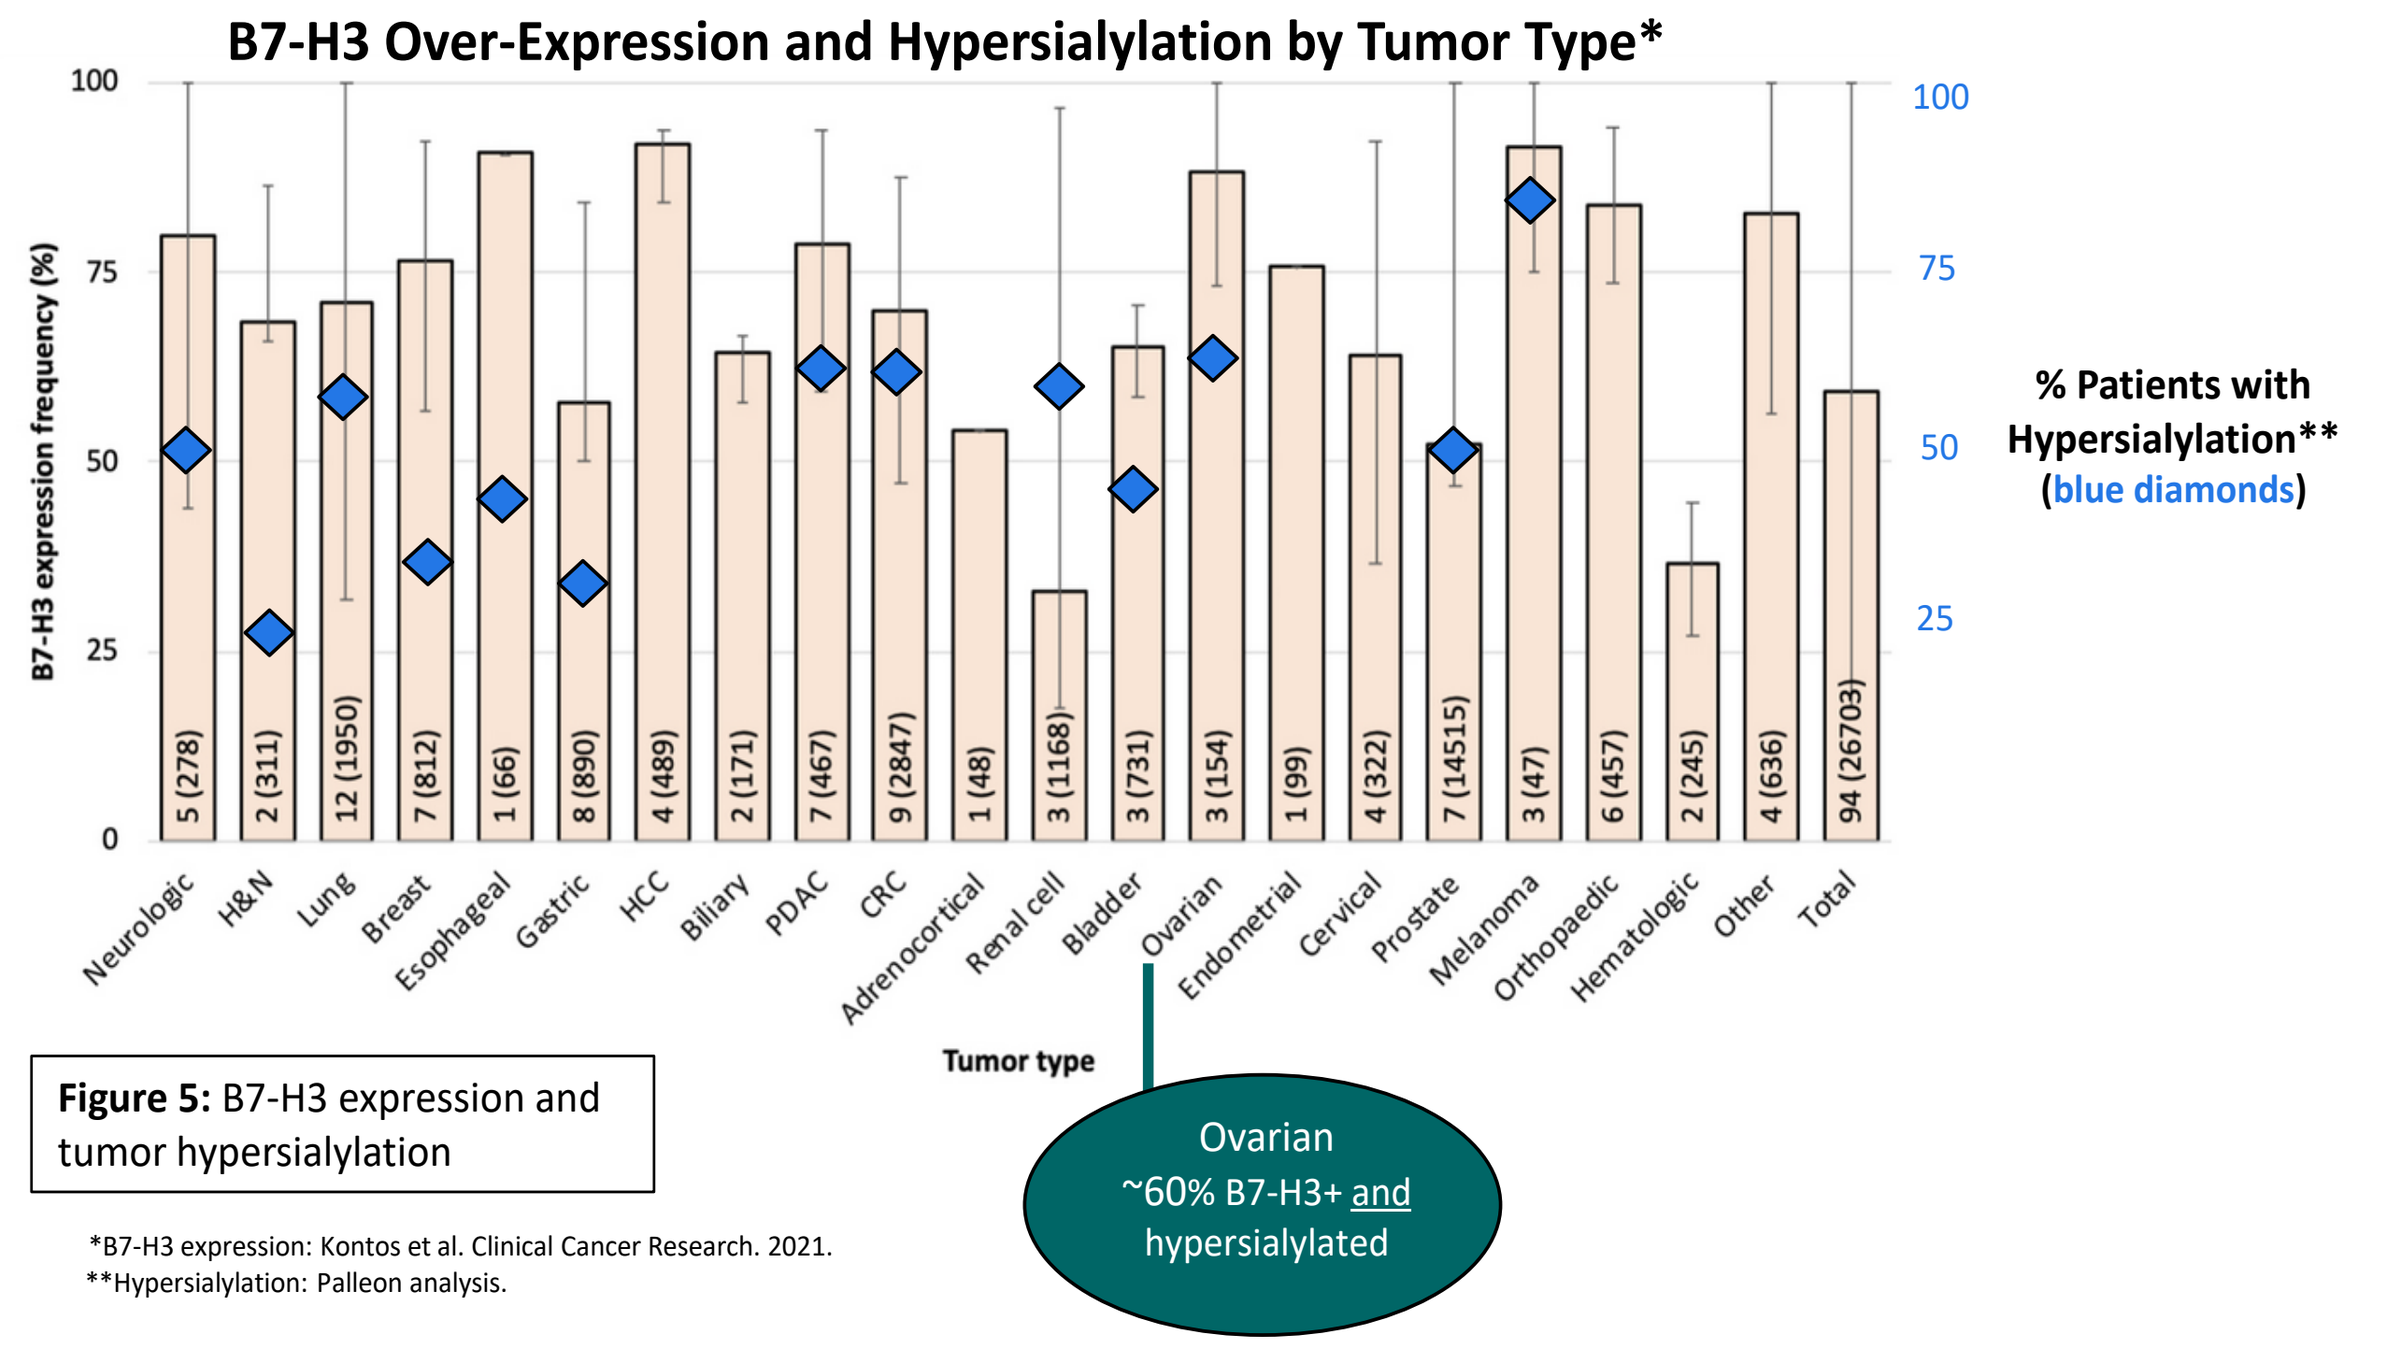
B7-H3 Over-Expression and Hypersialylation by Tumor Type*
B7-H3 expression frequency (%)
% Patients with Hypersialylation**
(blue diamonds)
Neurologic
H&N
Lung
Breast
Esophageal
Gastric
HCC
Biliary
PDAC
CRC
Adrenocortical
Renal cell
Bladder
Ovarian
Endometrial
Cervical
Prostate
Melanoma
Orthopaedic
Hematologic
Other
Total
Tumor type
Ovarian
~60% B7-H3+ and hypersialylated

Figure 5: B7-H3 expression and tumor hypersialylation

*B7-H3 expression: Kontos et al. Clinical Cancer Research. 2021.
**Hypersialylation: Palleon analysis.

- B7-H3 is broadly overexpressed across solid tumors; elevated expression correlates with advanced stage and metastatic disease
- Hypersialylation co-occurs with B7-H3 overexpression across multiple tumor types and independently correlates with poor clinical outcome in over a dozen indications
- Priority indications where both targets are highly co-expressed:
 - Ovarian cancer: ~90% B7-H3 expression; ~65% hypersialylated → first-in-human indication
 - NSCLC, SCLC, and CRPC: high co-expression rates → priority expansion indications
- Ovarian cancer is particularly well-suited to this approach: elevated B7-H3 and sialoglycan expression versus healthy tissue; hypersialylation linked to tumor growth, immune evasion, chemoresistance, and poor prognosis

E-688/HLX316 Phase 1 Trial Initiating in 2026: Dose Escalation with Expansion in Platinum-Resistant Ovarian Cancer

- Phase 1a: dose escalation beginning at 1 mg/kg sentinel patient, escalating through 3, 10, 20, and 30 mg/kg (≥3 patients/cohort) to establish DLT and MTD; backfill cohorts of 7–10 patients at the two highest dose levels
- Phase 1b: two parallel expansion cohorts (20 patients each) in platinum-resistant ovarian cancer at two selected dose levels; additional expansion cohort planned per positive efficacy signal; primary endpoint ORR; secondary endpoints PFS, DCR, DOR, TTR, OS, and safety/AE profile

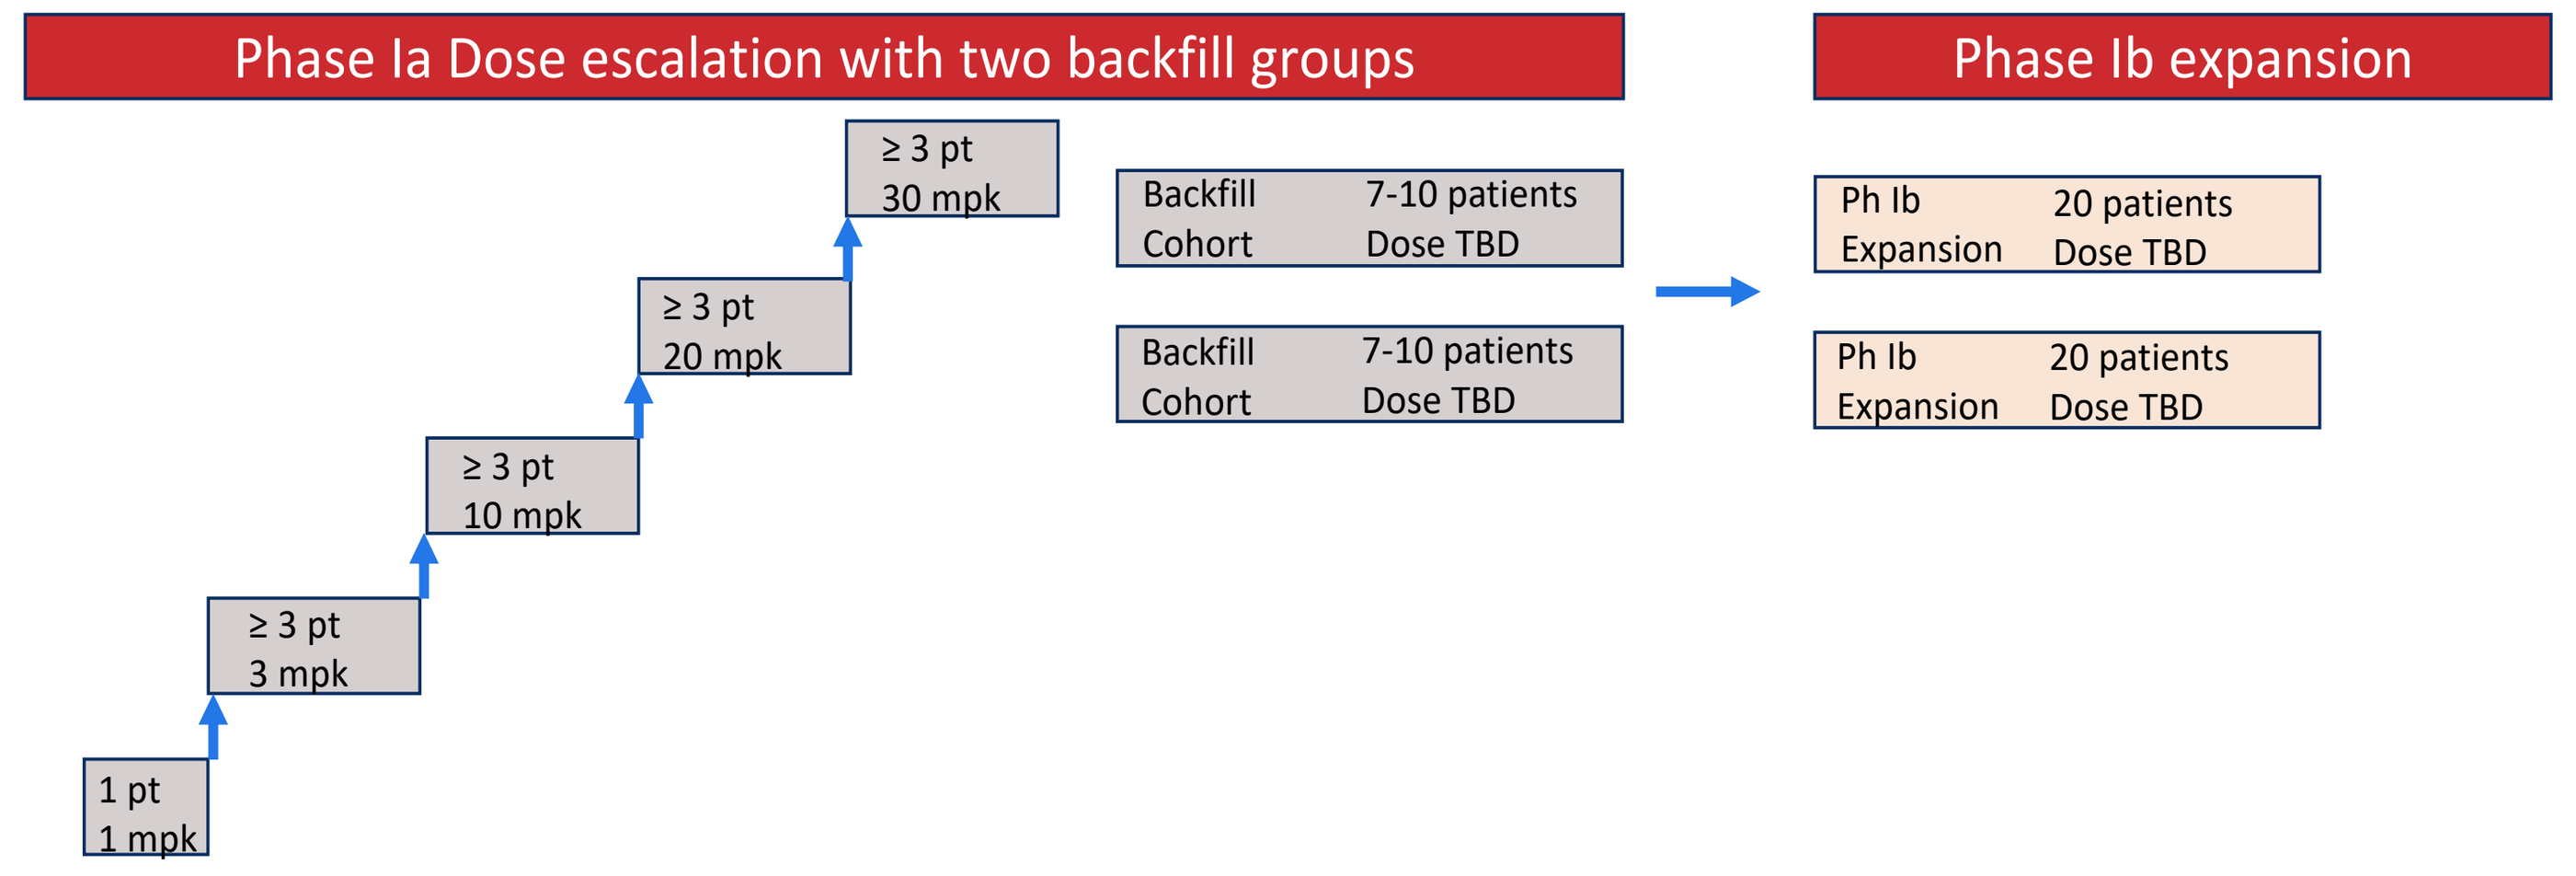
Phase Ia Dose escalation with two backfill groups
1 pt
1 mpk
≥ 3 pt
3 mpk
≥ 3 pt
10 mpk
≥ 3 pt
20 mpk
≥ 3 pt
30 mpk
Backfill Cohort 7-10 patients Dose TBD
Backfill Cohort 7-10 patients Dose TBD
Phase Ib expansion
Ph Ib Expansion 20 patients Dose TBD
Ph Ib Expansion 20 patients Dose TBD

Figure 6 E-688/HLX316 Phase 1a/b trial design: dose escalation to establish safety and MTD (Phase 1a) followed by two parallel expansion cohorts at selected dose levels in platinum-resistant ovarian cancer patients to seek efficacy signal (Phase 1b). Phase 1a begins with a sentinel patient at 1 mg/kg, escalating through 3, 10, 20, and 30 mg/kg (3 patients per cohort) with backfill cohorts at the two highest dose levels. Phase 1b enrolls 20 patients per cohort at two dose levels, with additional expansion cohorts planned following positive efficacy signal. Key eligibility criteria, primary endpoints (DLT/MTD in Phase 1a; ORR in Phase 1b), and secondary endpoints are detailed in the adjacent panel.