Development of a Bifunctional PD-L1-Targeted Sialidase as a Novel Cancer Immunotherapeutic Approach

Jenny Che, Lihui Xu, Wayne Gatlin, Robert LeBlanc, Lizhi Cao, James Broderick, Li Peng
Palleon Pharmaceuticals, Waltham, MA, USA

LB221

Introduction

Sialoglycans have emerged as a critical glyco-immune checkpoint that impairs antitumor response by inhibiting innate and adaptive immunity. We have reported previously that Bi-Sialidase – an engineered human sialidase-Fc fusion – potentiates antitumor immune response by cleaving terminal sialic acids from sialoglycans (desialylating) on tumor cells and immune cells. Furthermore, we have shown that a bifunctional tumor-targeted sialidase, a heterodimeric molecule consisting of one chain of sialidase-Fc and a second chain of a HER2-targeting antibody (trastuzumab), leads to more efficient desialylation of tumor cells than the non-targeted Bi-Sialidase and demonstrates antitumor activity in trastuzumab-resistant and low HER2-expressing tumor models.

To evaluate the impact of targeted desialylation of both tumor cells and immune cells, we designed and characterized a bifunctional PD-L1-targeted sialidase in preclinical models, since PD-L1 is expressed on many types of tumor cells and immune cells. Furthermore, the bifunctional PD-L1-targeted sialidase simultaneously inhibits two orthogonal immune checkpoint pathways, immunosuppressive sialoglycans and the PD-1/PD-L1 axis. We generated a humanized anti-human PD-L1 antibody with potency of PD-1/PD-L1 blockade comparable to the existing anti-PD-L1 drugs, atezolizumab and avelumab. Subsequently, we constructed and screened multiple bifunctional PD-L1-targeted sialidases in various configurations and selected a heterodimeric molecule (E-705) as the lead, based on developability and potency.

The PD-L1-Targeted Sialidase (E-705) consists of one chain of sialidase-Fc from the third generation of engineered human sialidase (Neu2) and a second chain of the in-house generated anti-PD-L1 antibody. We further characterized E-705 using various biochemical assays in vitro and tested its antitumor activity in vivo using the transgenic CT26-human PD-L1 mouse tumor model expressing human PD-1 and PD-L1 in replace of the murine counterparts.

Degradation of Immunosuppressive Sialoglycans Using Human Sialidase-Based *EAGLE* Technology to Treat Cancer

Sialoglycans Suppress Immune Response Through Multiple Immune Receptors | **Palleon Approach: Sialoglycan Degradation** | **Desialylation Overcomes Redundancy and Enables Anti-Tumor Immunity**

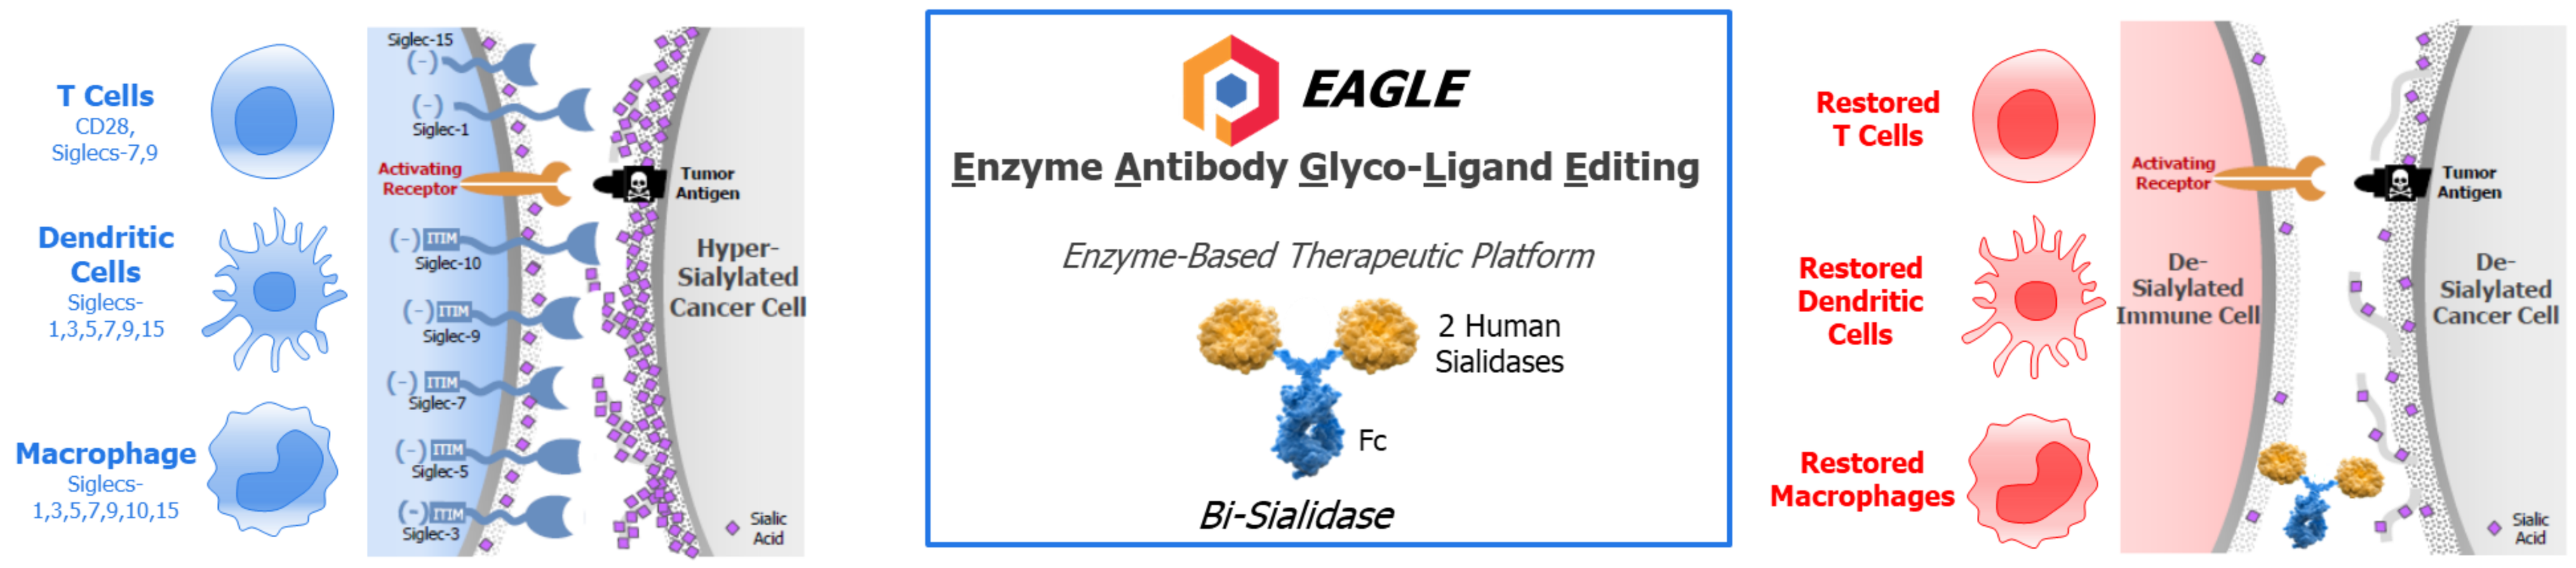
T Cells
CD28, Siglecs-7,9
Dendritic Cells
Siglecs-1,3,5,7,9,15
Macrophage
Siglecs-1,3,5,7,9,10,15
Activating Receptor
Tumor Antigen
Hyper-Sialylated Cancer Cell
Sialic Acid
EAGLE
Enzyme Antibody Glyco-Ligand Editing
Enzyme-Based Therapeutic Platform
2 Human Sialidases
Fc
Bi-Sialidase
Restored T Cells
Restored Dendritic Cells
Restored Macrophages
De-Sialylated Immune Cell
De-Sialylated Cancer Cell

- **Hypersialylation** is so immunosuppressive in cancer because it impacts multiple receptors concurrently
- Blocking an individual Siglec receptor cannot overcome this **redundancy**

- **Sialic acid** is the **single common dependency** of this highly redundant immune suppressive axis
- **Sialoglycan Degradation**: Sialidase enzymes remove sialic acids on both immune cell and cancer cell surfaces
- Impact: **Restored Anti-Tumor Immunity**

- Modest reduction of sialic acid density releases multiple inhibitory signals simultaneously (Siglecs, CD28 blockade, others)
- Impact: **Robust anti-tumor response** from both innate and adaptive immune cells

Figure 1. Schematic representation of the sialoglycan immune checkpoint axis and the *EAGLE* therapeutic platform. At the immunological synapse, a dense array of various sialoglycans interact with multiple sialic-acid sensing immune receptors, including Siglecs[1-3] (sialic acid-binding Ig-like lectins) and CD28[4] on immune cells, dampening innate and adaptive immune responses. The interactions between sialoglycans and sialic-acid-sensing receptors are "Velcro"-like with overlapping/promiscuous binding preferences and tremendous redundancy, which poses a challenge for therapeutic intervention of this axis because targeting a single receptor or ligand cannot overcome the redundancy of this biology. The engineered human sialidase-based *EAGLE* therapeutic platform overcomes this hurdle by removing terminal sialic acids, the common motif of various sialoglycans, to release sialoglycan-mediated immunosuppression.

Bifunctional and Targeted Sialidase Enables Dual Antitumor Mechanisms and More Efficient Desialylation of Targeted Cells Compared to Non-Targeted Bi-Sialidase

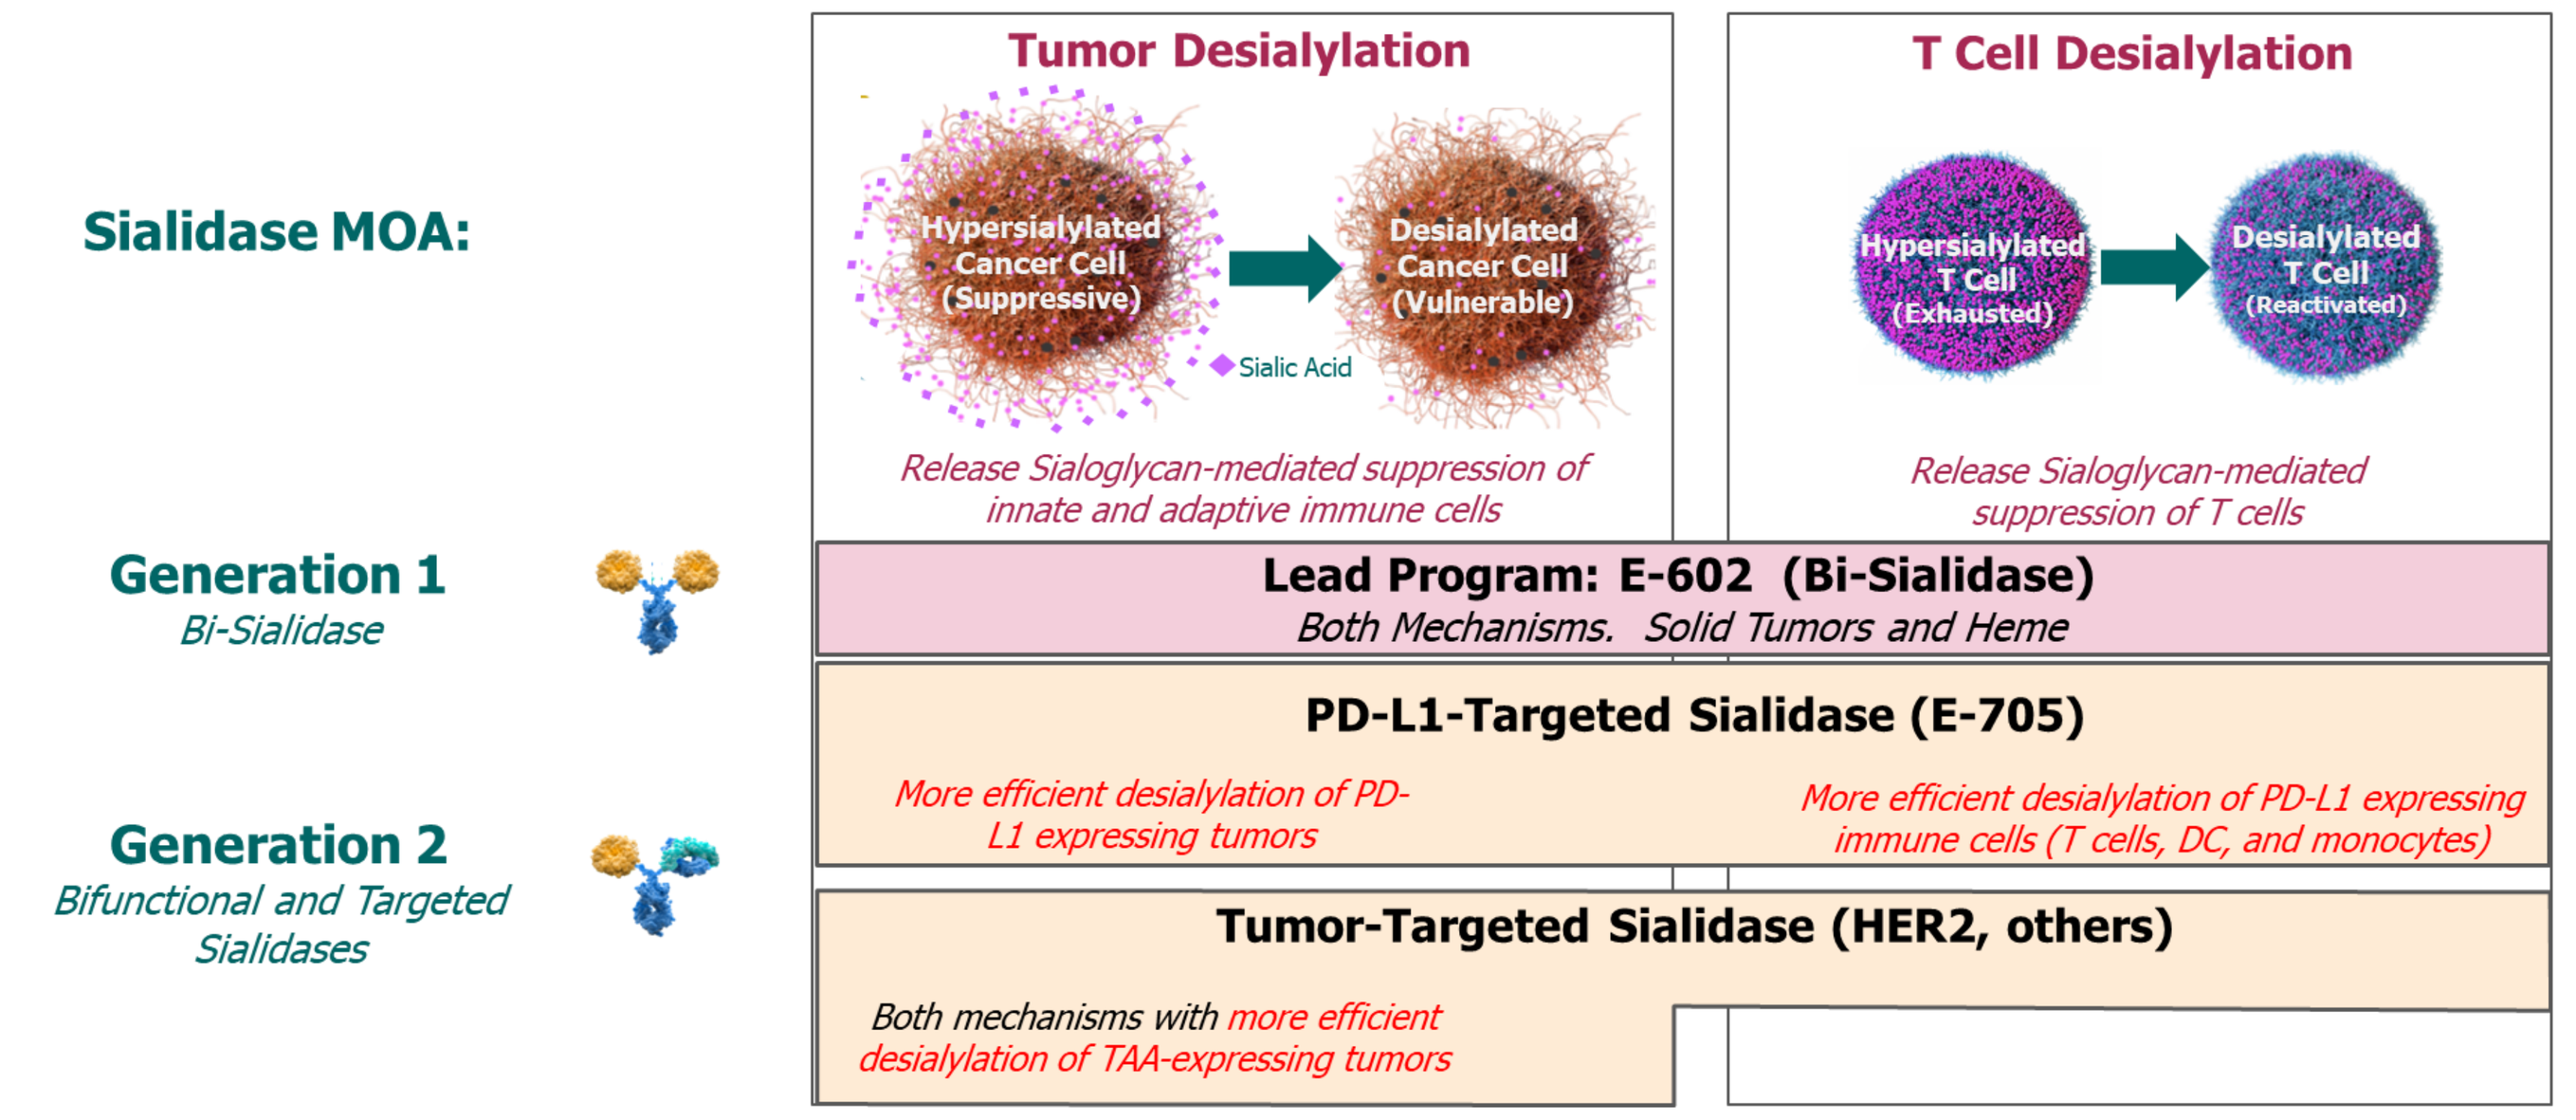
Sialidase MOA:
Tumor Desialylation
Hypersialylated Cancer Cell (Suppressive)
Desialylated Cancer Cell (Vulnerable)
Sialic Acid
Release Sialoglycan-mediated suppression of innate and adaptive immune cells
T Cell Desialylation
Hypersialylated T Cell (Exhausted)
Desialylated T Cell (Reactivated)
Release Sialoglycan-mediated suppression of T cells
Generation 1
Bi-Sialidase
Lead Program: E-602 (Bi-Sialidase)
Both Mechanisms. Solid Tumors and Heme
Generation 2
Bifunctional and Targeted Sialidases
PD-L1-Targeted Sialidase (E-705)
More efficient desialylation of PD-L1 expressing tumors
More efficient desialylation of PD-L1 expressing immune cells (T cells, DC, and monocytes)
Tumor-Targeted Sialidase (HER2, others)
Both mechanisms with more efficient desialylation of TAA-expressing tumors

Figure 2. Schematic representation of the primary MoAs of *EAGLE*, including 1) desialylation of tumor cells to release tumor- associated sialoglycan-mediated immunosuppression of innate and adaptive antitumor immunity, 2) desialylation of T cells to enhance sialoglycan-mediated suppression of T cell function. E-602 is a homodimer of the engineered human sialidase Neu2 (v2.0) genetically fused to a human IgG1 Fc region by an IgG1 hinge. Targeted-Sialidases are bifunctional molecules consisting of the sialidase moiety and a tumor-associated antigen (TAA) or an immune cell-targeted (such as PD-1) arm.

Results

Selection of the Bifunctional PD-L1-Targeted Sialidase (E-705) (Lead Identification and Lead Optimization)

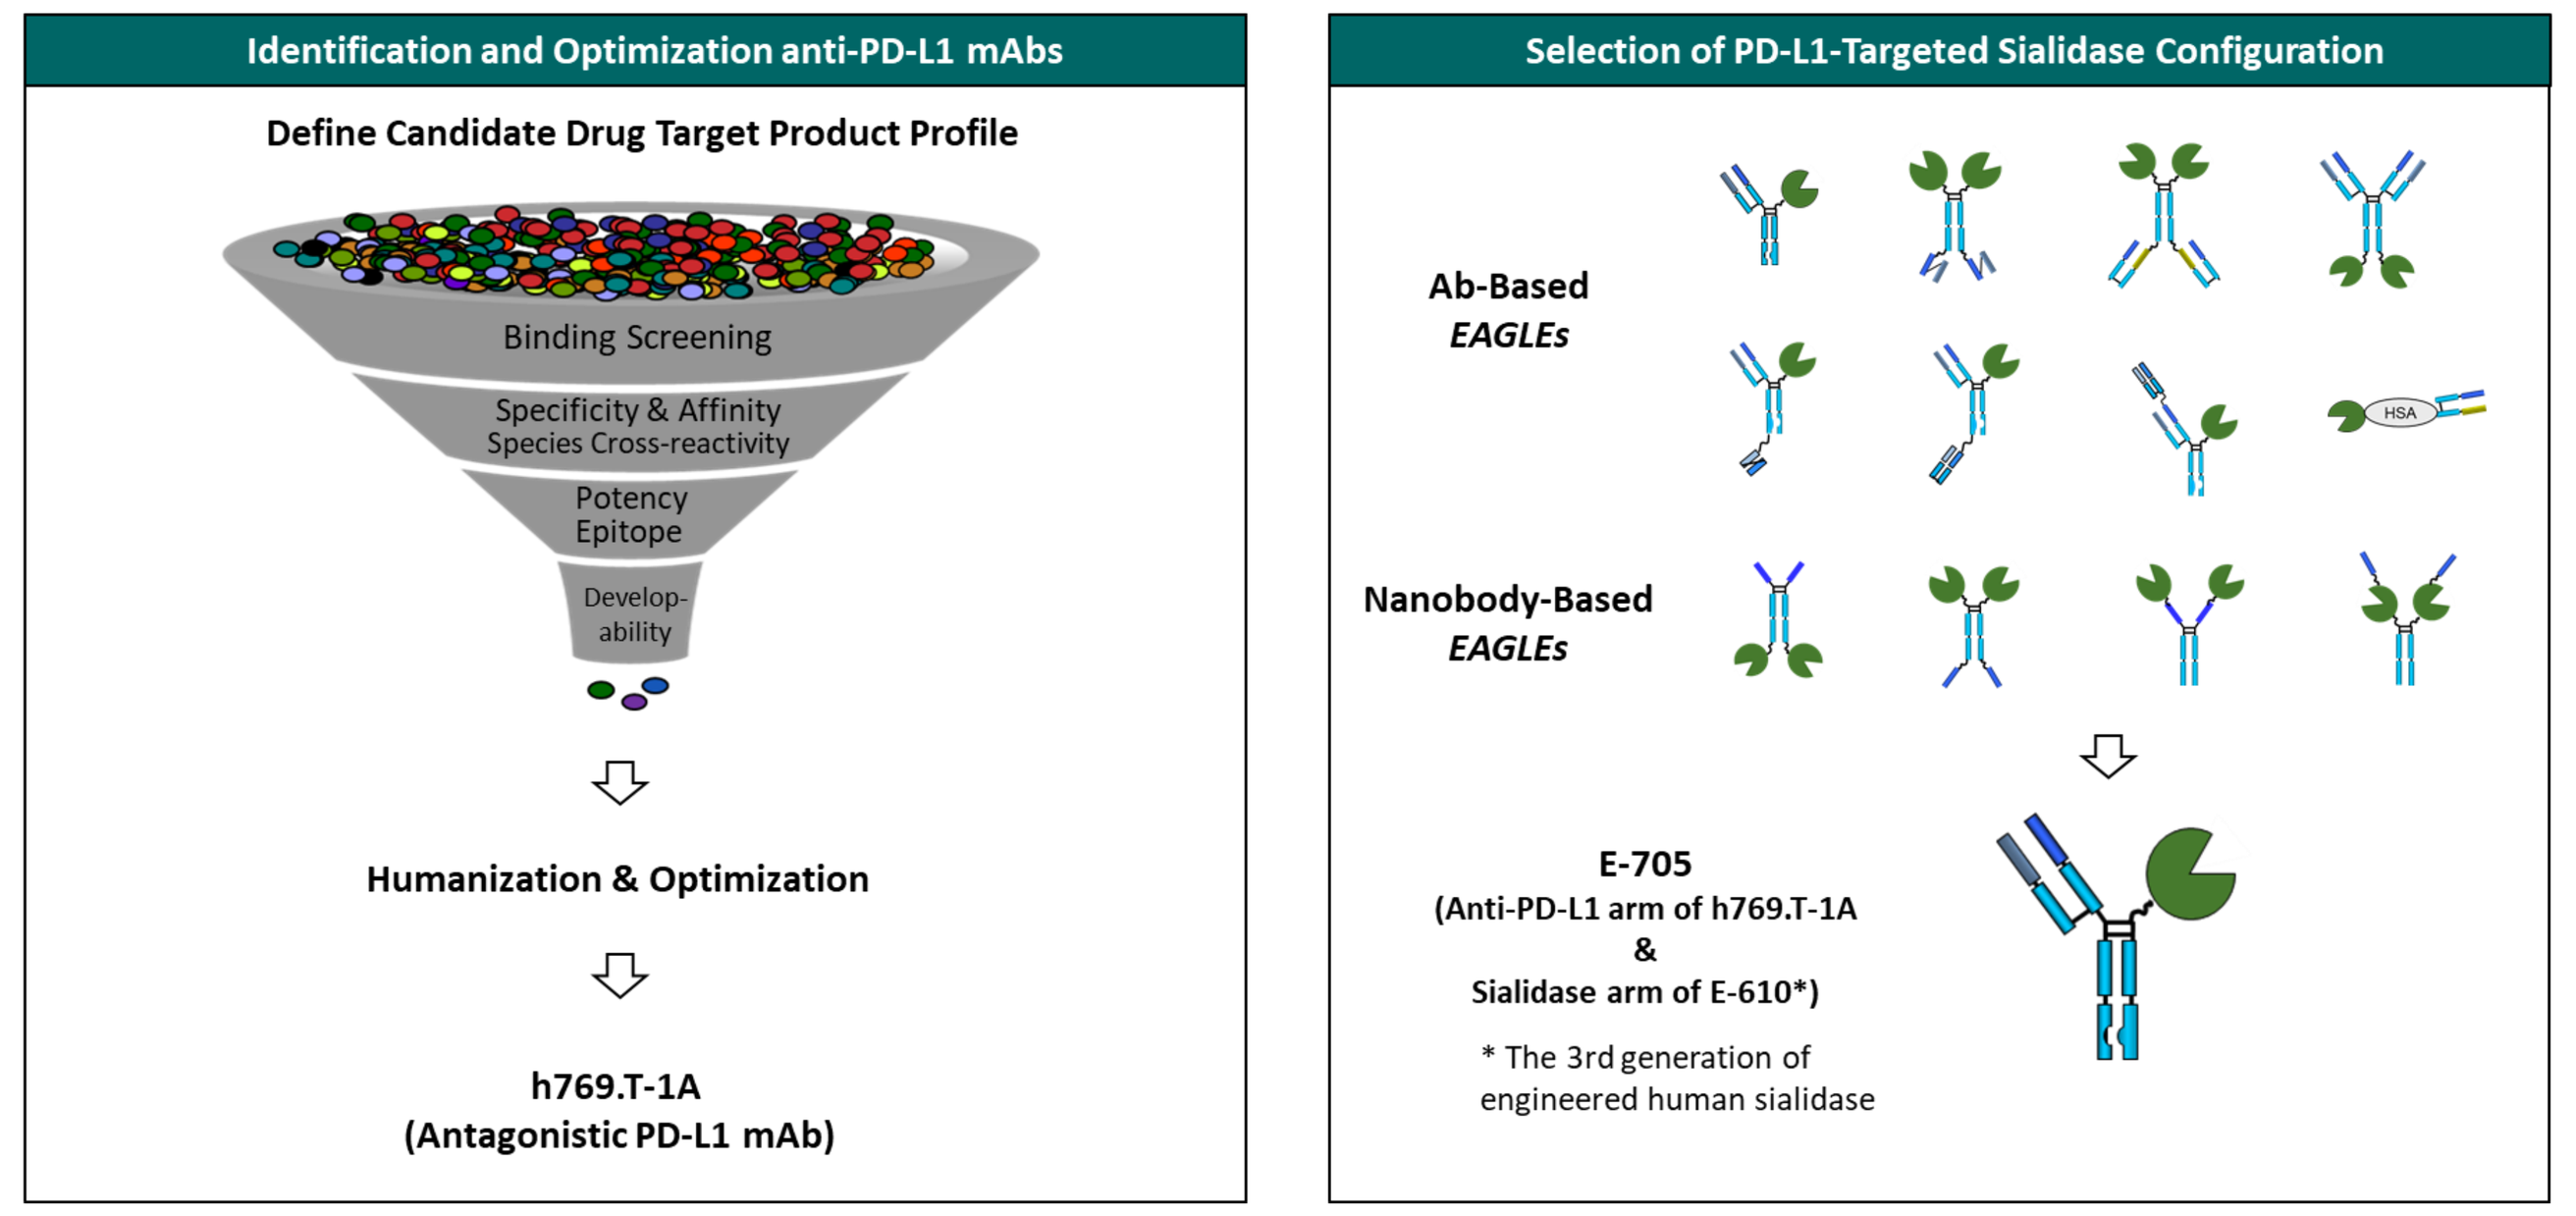
Identification and Optimization anti-PD-L1 mAbs
Define Candidate Drug Target Product Profile
Binding Screening
Specificity & Affinity
Species Cross-reactivity
Potency
Epitope
Develop-ability
Humanization & Optimization
h769.T-1A
(Antagonistic PD-L1 mAb)
Selection of PD-L1-Targeted Sialidase Configuration
Ab-Based EAGLEs
HSA
Nanobody-Based EAGLEs
E-705
(Anti-PD-L1 arm of h769.T-1A
&
Sialidase arm of E-610*)
* The 3rd generation of engineered human sialidase

Figure 3. The lead identification flowchart of anti-PD-L1 mAb (left) and schematic representation of various PD-L1-targeted sialidase configurations. The lead (E-705) was selected based on expression yield, stability, sialidase activity, and PD-1/PD-L1 inhibition potency.

E-705 Demonstrated Dual Functions of Cleaving Sialic Acids and Inhibiting PD-1/PD-L1 Interaction

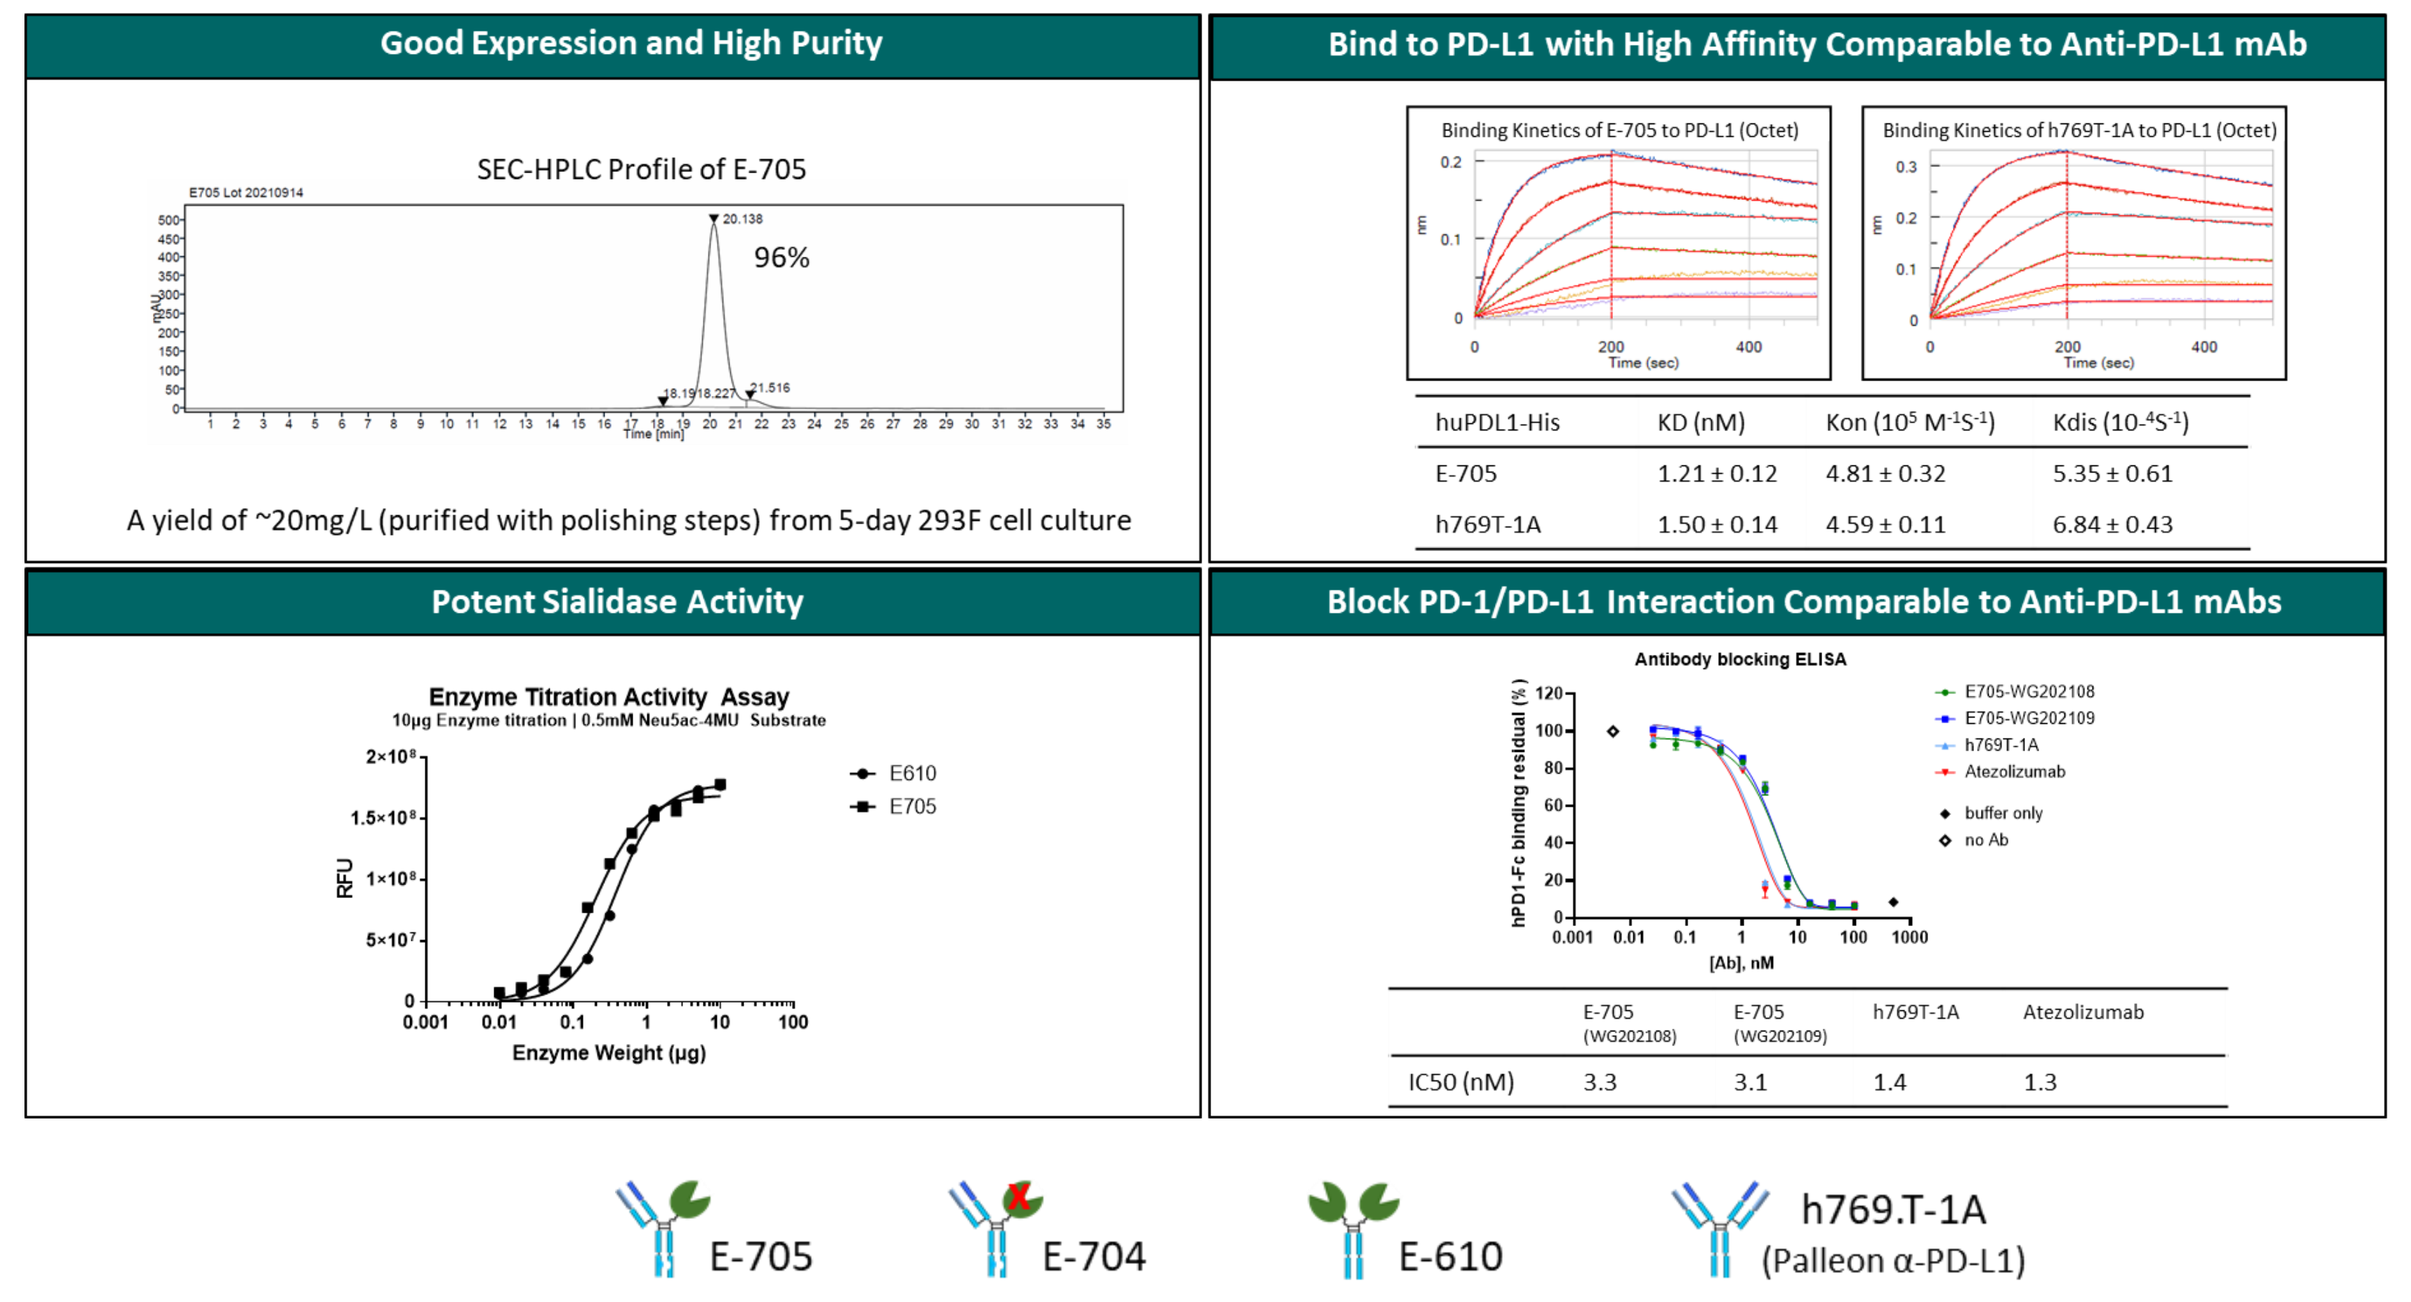
Good Expression and High Purity
SEC-HPLC Profile of E-705
96%
A yield of ~20mg/L (purified with polishing steps) from 5-day 293F cell culture
Bind to PD-L1 with High Affinity Comparable to Anti-PD-L1 mAb
Binding Kinetics of E-705 to PD-L1 (Octet)
Binding Kinetics of h769T-1A to PD-L1 (Octet)
Potent Sialidase Activity
Enzyme Titration Activity Assay
E610
E705
Enzyme Weight (μg)
Block PD-1/PD-L1 Interaction Comparable to Anti-PD-L1 mAbs
Antibody blocking ELISA
E-705
E-704
E-610
h769.T-1A
(Palleon α-PD-L1)

huPDL1-His	KD (nM)	Kon (10^5 $M^{-1}S^{-1}$)	Kdis ($10^{-4}S^{-1}$)
E-705	1.21 ± 0.12	4.81 ± 0.32	5.35 ± 0.61
h769T-1A	1.50 ± 0.14	4.59 ± 0.11	6.84 ± 0.43

	E-705 (WG202108)	E-705 (WG202109)	h769T-1A	Atezolizumab
IC50 (nM)	3.3	3.1	1.4	1.3

Figure 4. Biochemical and functional characterization of PD-L1-targeted sialidase (E-705). E-705 yielded 20mg/L with 96% purity from a 5-day transient 293F cell expression and a two-step purification of protein A and CHT chromatography. E-705 showed comparable desialylation potency to its parental sialidase-Fc (E-601) toward a fluorescently labeled sialic acid substrate (4-MU-Neu5Ac), confirming that pairing with the anti-PD-L1 arm didn't impact sialidase activity. In addition, E-705 demonstrated binding to PD-L1 and EC50s of inhibiting PD-1/PD-L1 interaction comparable to its parental anti-PD-L1 mAb (h769T-1A) or atezolizumab.

The PD-L1-Targeting Moiety of E-705 Enabled More Efficient Desialylation of PD-L1-Expressing Cells

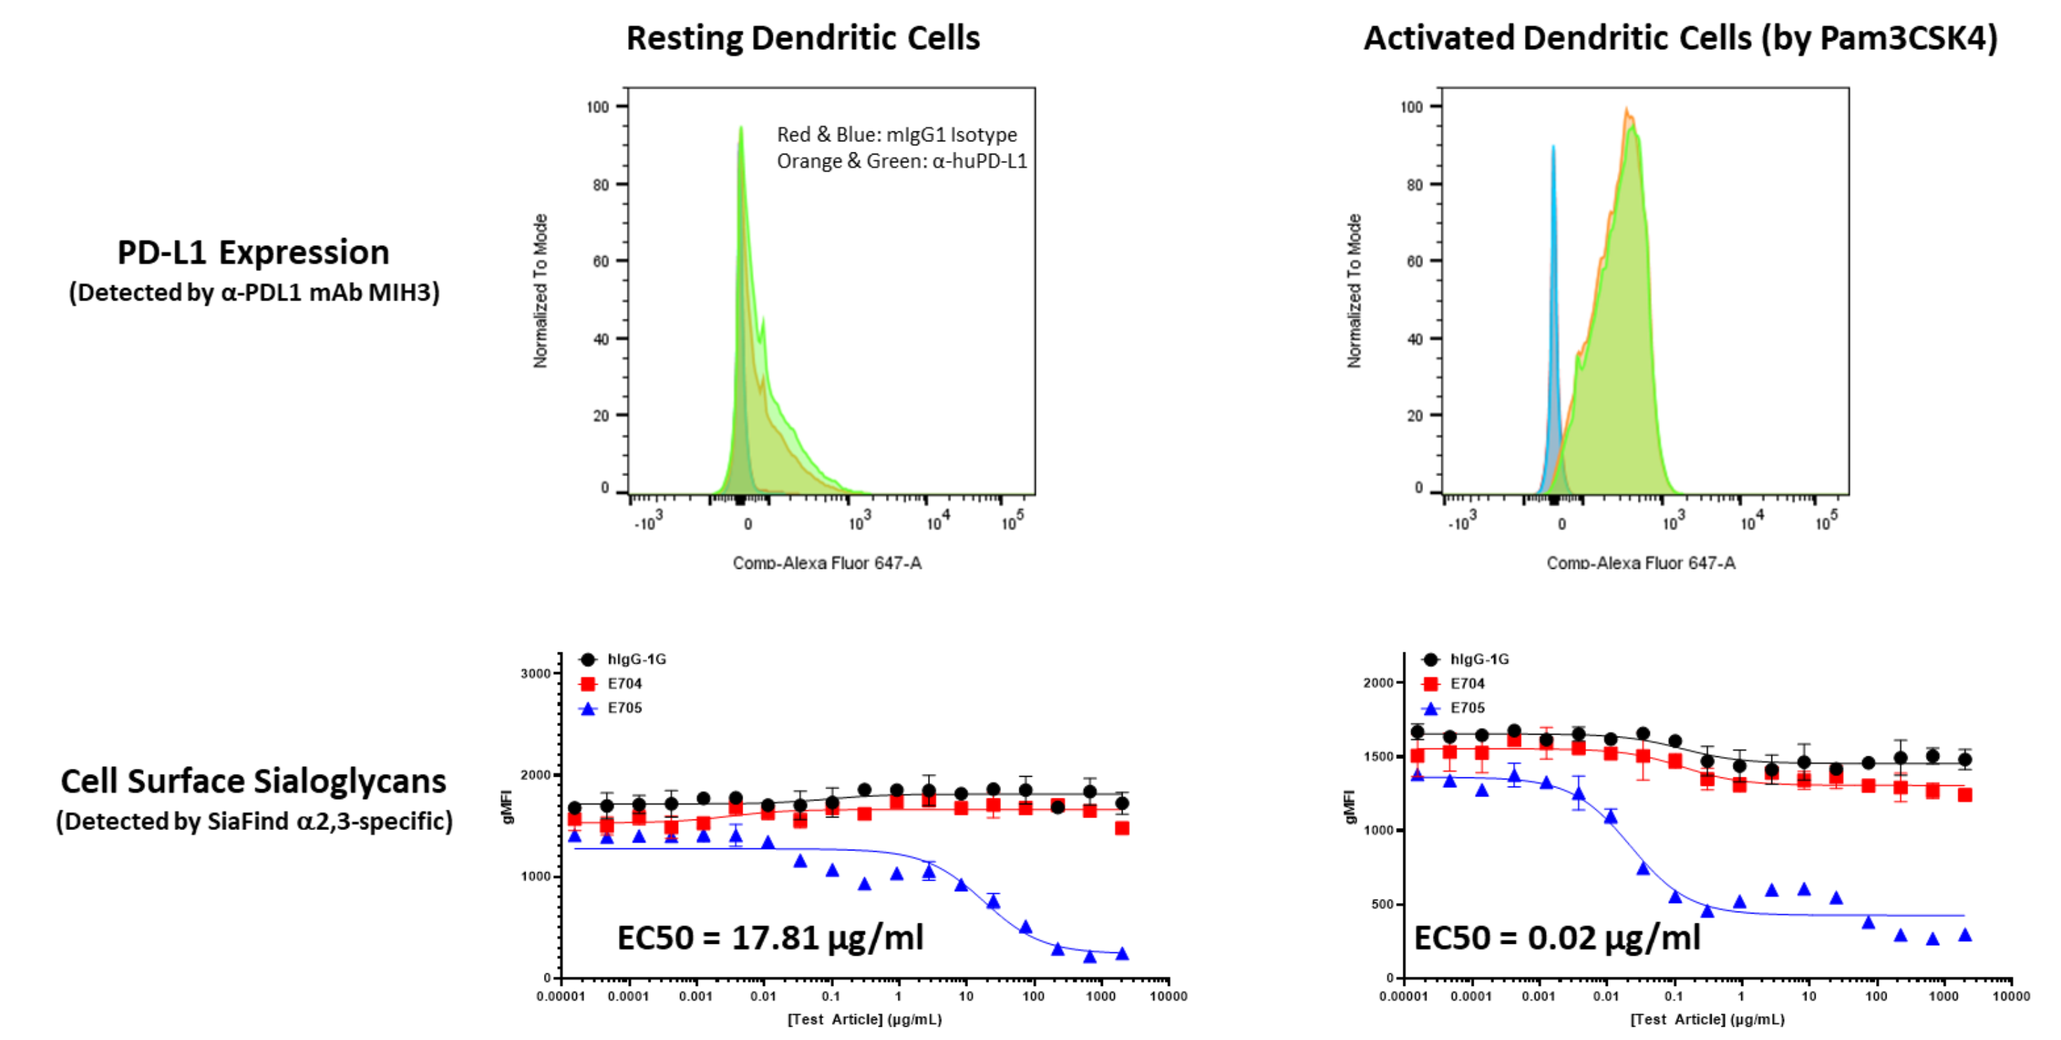
Resting Dendritic Cells
Activated Dendritic Cells (by Pam3CSK4)
PD-L1 Expression
(Detected by α-PDL1 mAb MIH3)
Red & Blue: mIgG1 Isotype
Orange & Green: α-huPD-L1
Comp-Alexa Fluor 647-A
Cell Surface Sialoglycans
(Detected by SiaFind α2,3-specific)
hIgG-1G
E704
E705
EC50 = 17.81 μg/ml
EC50 = 0.02 μg/ml
[Test Article] (μg/mL)

Figure 5. E-705 showed more efficient desialylation of activated DCs with abundant PD-L1 expression than resting DCs with low expression of PD-L1. Monocyte-derived dendritic cells (mDCs) were differentiated from peripheral CD14+ monocytes treated with GM-CSF and IL-4. Activated mDCs were simulated by TLR2/1 agonist Pam3CSK4, a synthetic triacylated lipopeptide. Resting and activated mDCs were treated with E-705, isotype, or silent-sialidase control (E-704), respectively. Activated mDCs showed more than 10-fold higher expression of PD-L1 than that of the resting mDCs. As expected, E-705 demonstrated higher potency (~900-fold) in desialylating activated mDCs (EC50 = 0.02 μg/ml) than resting mDCs (EC50 = 17.81 μg/ml).

E-705 Demonstrated Improved Antitumor Activity than PD-L1 Blockade or Sialidase in Transgenic Syngeneic CT26-hPD-L1 Mouse Tumor Model

MC38-hPD-L1 Transgenic Syngeneic Mouse Tumor Model Expressing Human PD-1 and PD-L1

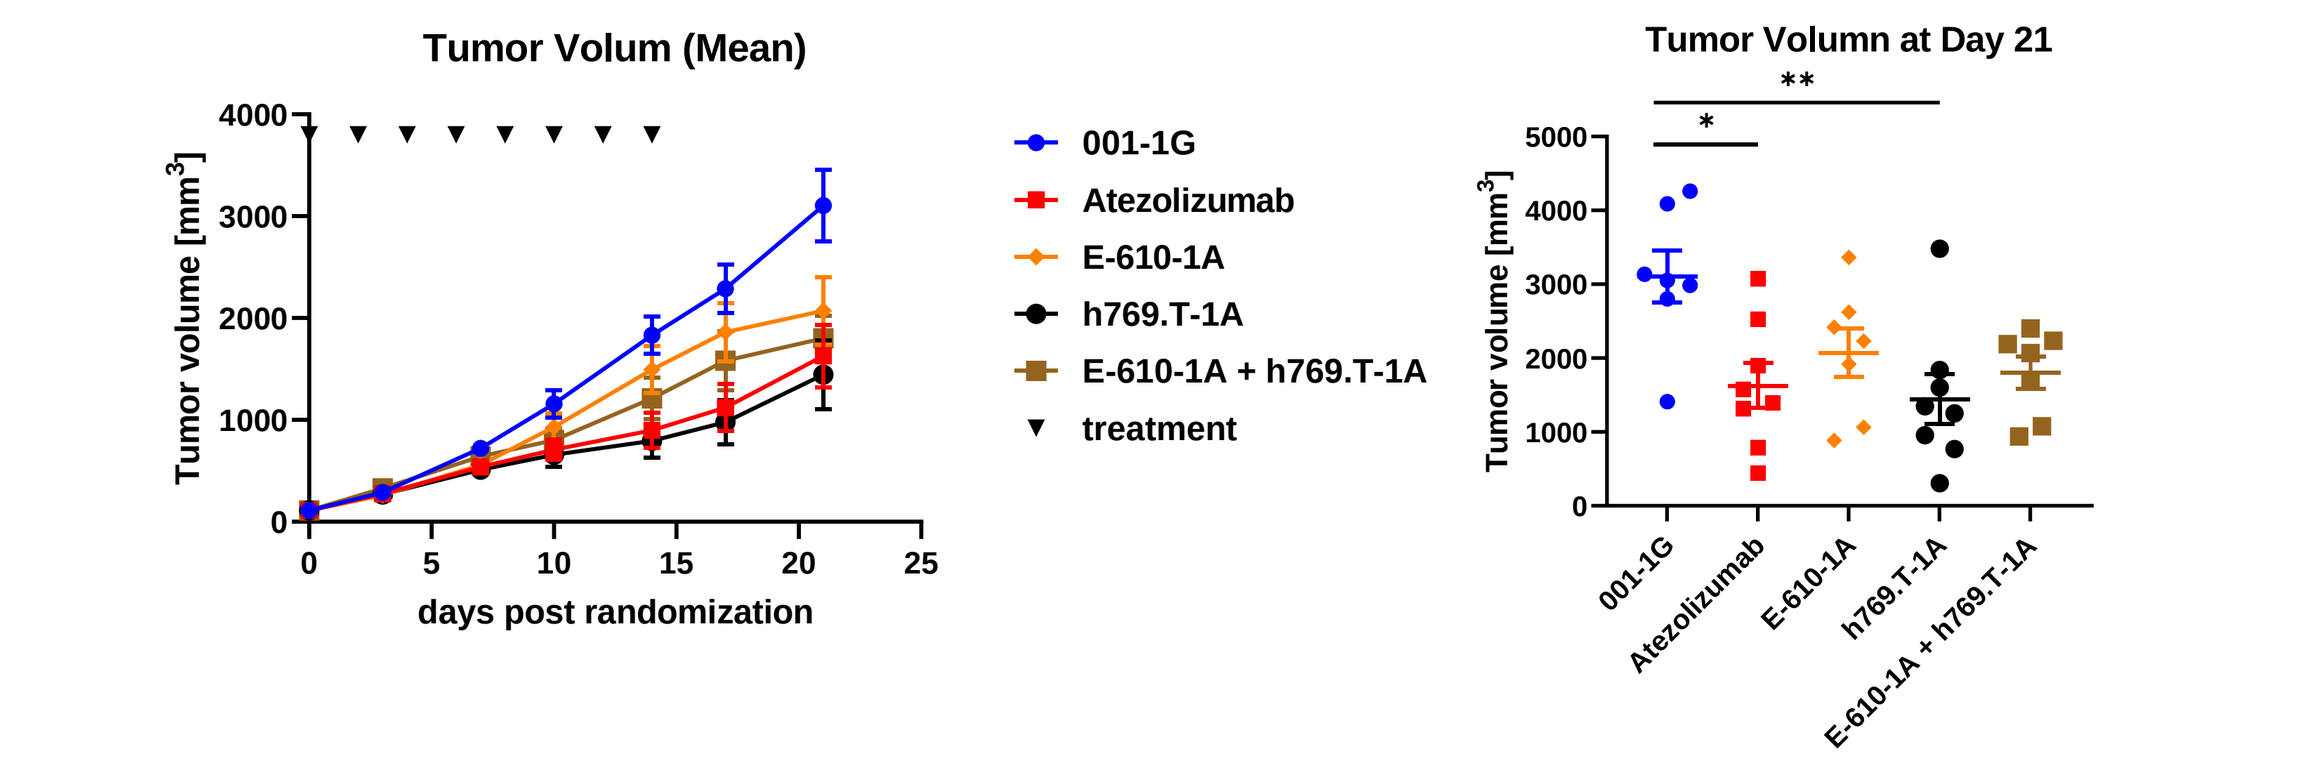
Tumor Volum (Mean)
Tumor volume [mm³]
days post randomization
001-1G
Atezolizumab
E-610-1A
h769.T-1A
E-610-1A + h769.T-1A
treatment
Tumor Volumn at Day 21

Figure 6. Efficacy study in MC38-hPD-L1 transgenic syngeneic mouse tumor model expressing human PD-1 and PD-L1 in replacement of mouse counterparts. Palleon's anti-PD-L1 mAb (h769.T-1A) showed comparable efficacy to atezolizumab. Furthermore, sialidase (E-610) also demonstrated antitumor activity in this PD-1/PD-L1 biology-focused transgenic mouse tumor model. However, the combination of h769.T-1A and E-610 did not show improved activity versus the single agent, suggesting the MC38 model is not suitable for studying the additive effect of combining desialylation and inhibition of PD1/PD-L1 axis.

CT26-hPD-L1 Transgenic Syngeneic Mouse Tumor Model Expressing Human PD-1 and PD-L1

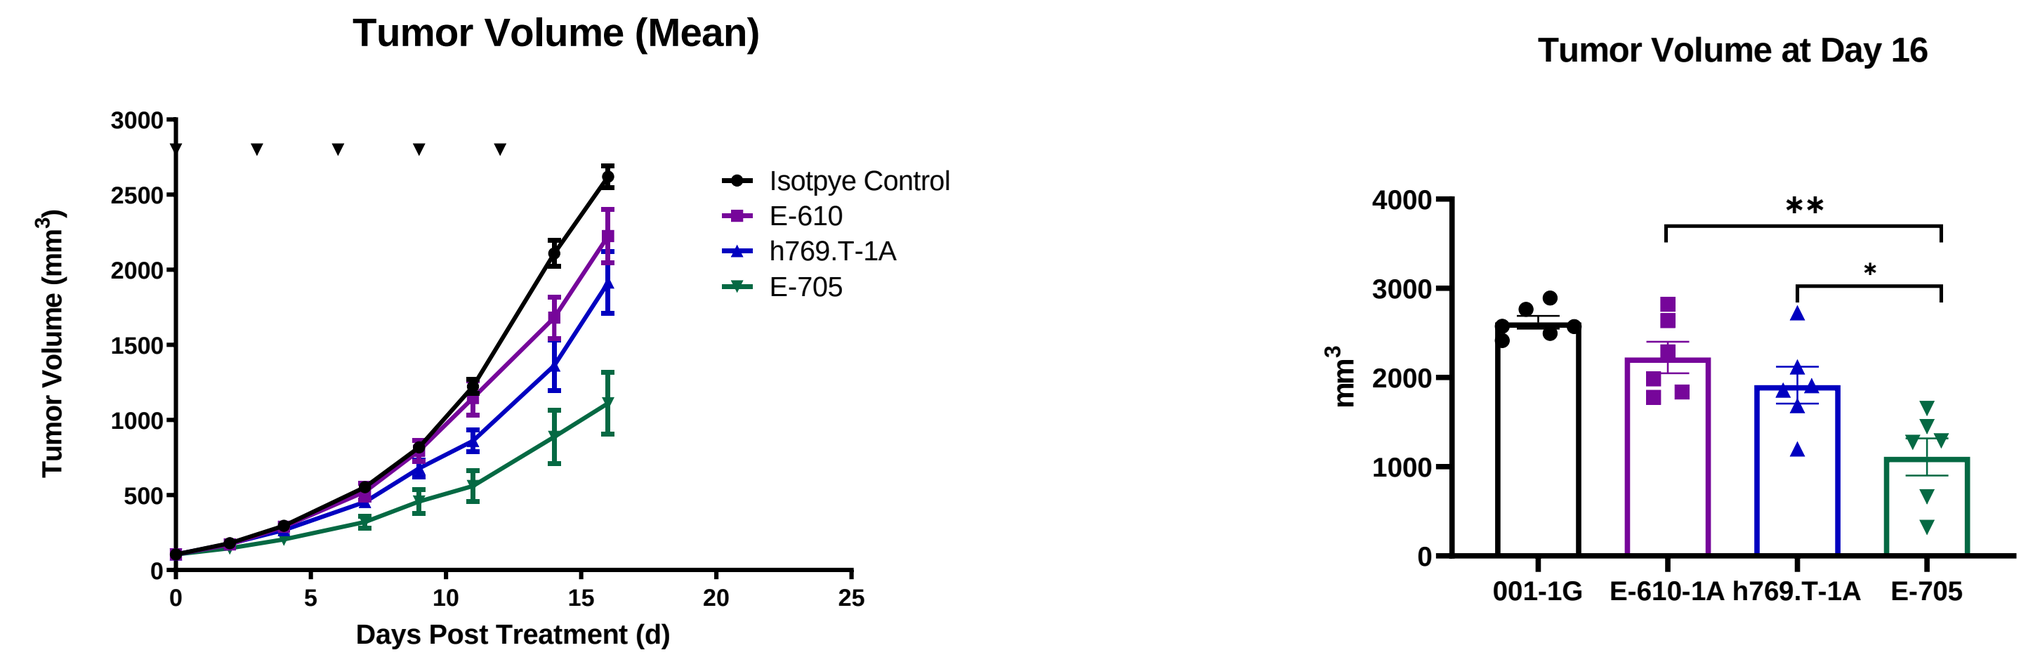
Tumor Volume (Mean)
Tumor Volume (mm³)
Days Post Treatment (d)
Isotpye Control
E-610
h769.T-1A
E-705
Tumor Volume at Day 16
001-1G
E-610-1A
h769.T-1A
E-705

Figure 7. Efficacy study in CT26-hPD-L1 transgenic syngeneic mouse tumor model expressing human PD-1 and PD-L1 in replacement of mouse counterparts. E-705 showed improved antitumor activity compared to its parental sialidase (E-610) or anti-PD-L1 mAb (h769.T-1A) alone in the CT26-hPD-L1 transgenic syngeneic mouse tumor model.

Pharmacodynamics Study of E-705-Mediated Immune Modulation in Tumor Microenvironment

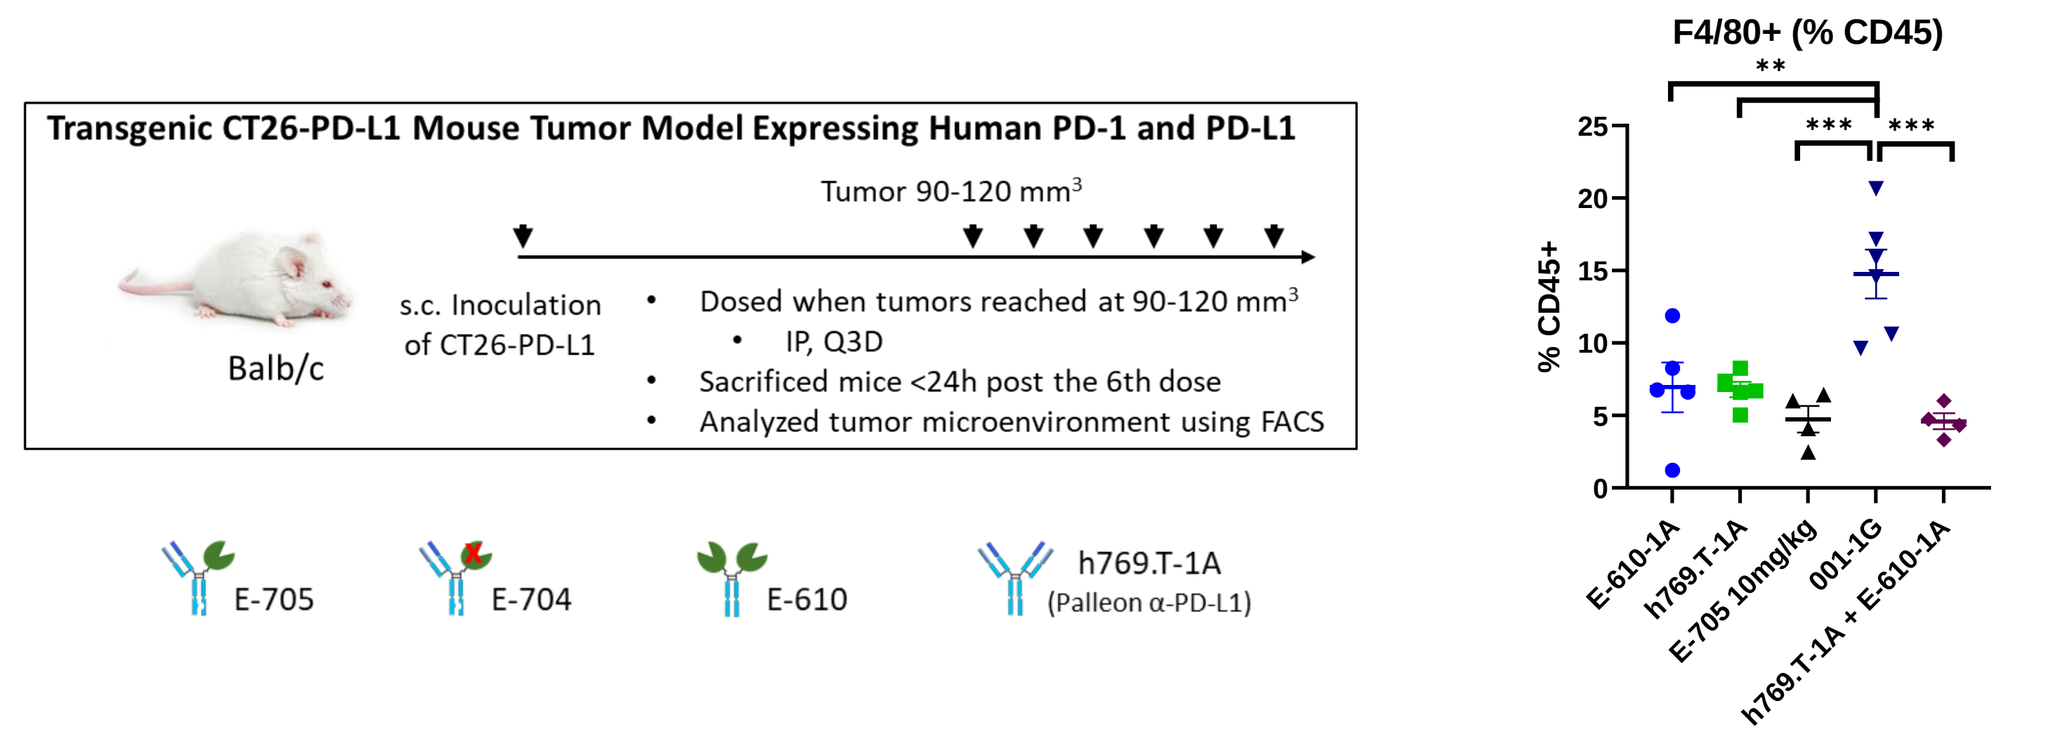
Transgenic CT26-PD-L1 Mouse Tumor Model Expressing Human PD-1 and PD-L1
Tumor 90-120 mm³
Balb/c
s.c. Inoculation of CT26-PD-L1
Dosed when tumors reached at 90-120 mm³
IP, Q3D
Sacrificed mice <24h post the 6th dose
Analyzed tumor microenvironment using FACS
E-705
E-704
E-610
h769.T-1A
(Palleon α-PD-L1)
F4/80+ (% CD45)
% CD45+
E-610-1A
h769.T-1A
E-705 10mg/kg
001-1G
h769.T-1A + E-610-1A

Figure 8. FACS analysis of E-705-mediated immune modulation in the tumor microenvironment. Tumors were harvested within 24 hours post the 6th dose of treatment and subjected to immunophenotyping using flow cytometry. A significant decrease of macrophages in the tumor microenvironment was observed from either desialylation (E-610) or PD-L1 blockade. In addition, E-705 and the combination E-610 and h769.T-1A led to slightly more reduced tumor-infiltrating macrophages compared to the corresponding single-agent treatments.

Conclusions

- Degradation of sialoglycans using human sialidase-based *EAGLE* platform technology can effectively release sialoglycan-mediated immunosuppression of antitumor immune response.
- The PD-L1-Targeted Sialidase (E-705) enabled more efficient desialylation of PD-L1-expressing immune cells and tumor cells and showed improved antitumor activity compared to its parental sialidase or anti-PD-L1 mAb in the CT26-hPD-L1 transgenic syngeneic mouse tumor model.
- The PD-L1-Targeted Sialidase (E-705) offers a novel immunotherapeutic approach of inhibiting two orthogonal checkpoint pathways by cleaving immunosuppressive sialic acids and blocking the PD-1/PD-L1 axis.

References:

1. Gray, M.A., Stanczak, M.A., Mantuano, N.R. et al. Targeted glycan degradation potentiates the anticancer immune response in vivo. Nat Chem Biol 2020, 16, 1376–1384.
2. Cao, L., Petrone, A., Gatlin, W. et al. AACR Annual Meeting 2019, DOI: 10.1158/1538-7445.
3. Crocker PR, Varki A. Siglecs in the immune system. Immunology. 2001;103:137-45.
4. Edgar LJ, Thompson AJ, Vartabedian VF. et al. Sialic acid ligands of CD28 suppress co-stimulation of T cells. ACS Cent Sci. 2021, 7:1508-1515.