Melanoma patients with multi-Siglec ligands as profiled by HYDRA technology are refractory to PD1 blockade

491

Jillian M. Prendergast[1], Dennie T. Frederick [2], Adam Petrone [1], Adam Shoemaker [1], James Broderick [1], Karl Normington [1], Genevieve Boland [2], Li Peng [1].

Affiliation: [1] Palleon Pharmaceuticals, Waltham, MA; [2] Department of Surgery, Massachusetts General Hospital, Boston, MA

Introduction

Abstract (#491) : PD1/PD-L1 and CTLA-4 checkpoint blockade have revolutionized cancer therapy and led to cures in metastatic melanoma, but most patients develop primary and acquired resistance to these therapies. Treating this refractory population requires the discovery of new immune escape mechanisms. Sialic acid–binding immunoglobulin-type lectins (Siglecs) are expressed on the majority of white blood cells of the immune system, play critical roles in immune cell signaling and serve as immune checkpoints to prevent unwanted immune responses. Sialic acid is a ligand for inhibitory Siglecs; hypersialyation is a hallmark of poor prognosis and is believed to help tumors escape from immune surveillance. However, the role of hypersialylation in resistance to immune checkpoint therapies remains unexplored. To study if hypersialylation drives immune escape in melanoma, we profiled the immunosuppressive sialoglycans using Siglec-based high-affinity sialoglycan-binding constructs called 'HYDRAs'. The current study focuses on understanding Siglec-3, -7 and -9 sialoglycan ligand expression on tumors using the HYDRA-3, -7 and -9 platform, because these Siglecs are the major inhibitory Siglecs on both innate and adaptive immune cells among the fourteen Siglecs in humans. Serial sections from melanoma tumors and healthy tissues were stained with HYDRA-3, -7 or -9 and scored using the semi-qualitative H-score method by a blinded pathologist. HYDRA IHC on healthy and cancerous human tissues demonstrate unique binding patterns with melanomas having high signals for HYDRA-3, -7 and -9. A pre-treatment checkpoint inhibitor therapy cohort (n=53), which contained responders (n=30) and non-responders (n=23) to either aPD1 or aPD1 and aCTLA-4 combination therapy was further studied. Serial sections from each patient was stained with HYDRA-3, -7 or -9 and scored using the semi-qualitative H-score method by our blinded pathologist. Cutoffs were determined in an unbiased manner for each HYDRA individually and each possible HYDRA combination to obtain correlations with patient progression-free and overall survival. A significant tumor H-score cutoff of a combined HYDRA-3 and -7 correlated with poor outcomes. This HYDRA-3 and -7 cutoff did not correlate with other melanoma biomarkers such as BRAF-mutation, liver metastases, PD-L1, nor TILs, suggesting a unique biology independent of these markers. We discovered that melanoma patients with multi-Siglec ligands as profiled by HYDRAs tend to be resistant to PD-1 checkpoint blockade and can be candidates for novel treatments targeting the Siglec-Sialoglycan axis. A larger cohort and longitudinal study are currently underway to examine the Siglec-Sialoglycan axis of immunosuppression in melanoma and late-breaking results will be included in this poster.

Glyco-Immune Checkpoints Suppress Innate and Adaptive Immunity

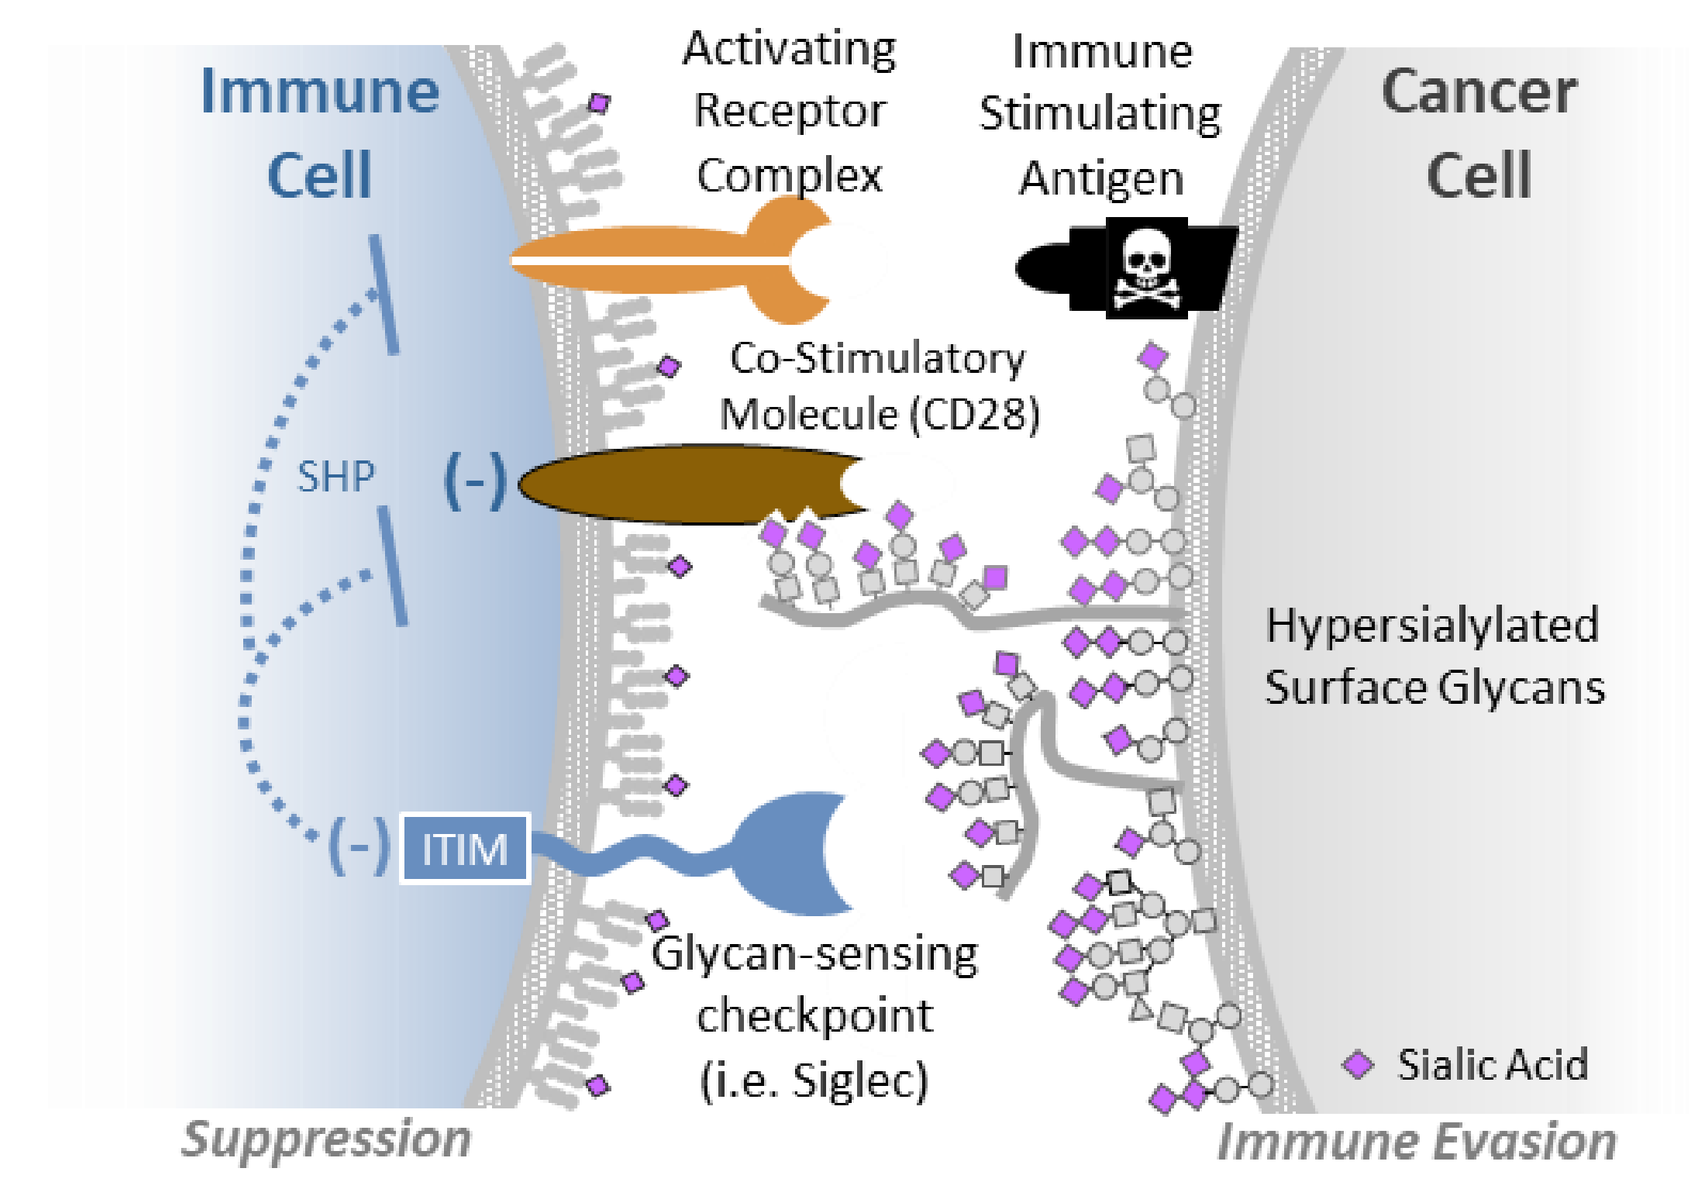
Immune Cell
Activating Receptor Complex
Immune Stimulating Antigen
Cancer Cell
Co-Stimulatory Molecule (CD28)
SHP
(-)
ITIM
Hypersialylated Surface Glycans
Glycan-sensing checkpoint (i.e. Siglec)
Sialic Acid
Suppression
Immune Evasion

Figure 1: Schematic representation of glyco-immune checkpoints axis. At the immunological synapse, sialoglycans on immune cells and tumor cells interact with Siglecs (sialic acid-binding Ig-like lectins[1]), expressed on macrophages and monocytes DC, and NK cells, and other immune receptors (such as CD28 on T cells[2]) to suppress anti-cancer immunity.

The HYDRA Translational Platform: Quantifying an Immunosuppressive Sialoglycan Signature in Patients

Challenge

- Tumors present heterogeneous glycan structures on their surfaces
- How to best quantify the tumor surface glycocalyx?

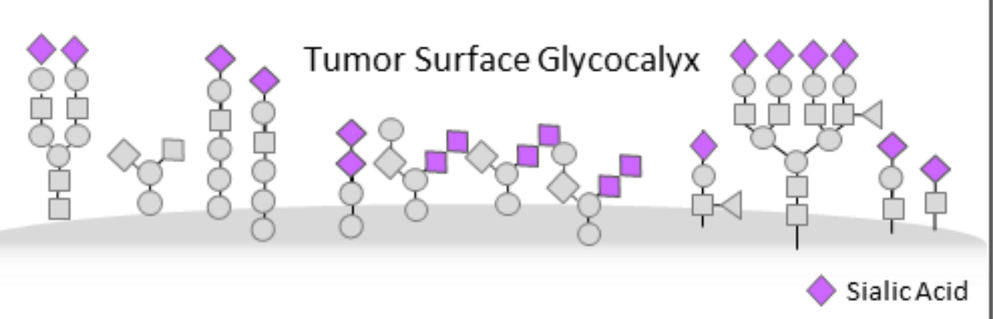
Tumor Surface Glycocalyx
Sialic Acid

Solution

- *HYDRA* – Proprietary IHC reagents based on multimeric Siglec receptors that specifically detect immunosuppressive sialoglycans
- Provide a ***functional signature*** of immunosuppressive tumor sialoglycans overcoming the chemical structural heterogeneity of sialoglycans
- H Score 0-300 scale: IHC score (0-3) x % cells

Representative HYDRA structure:

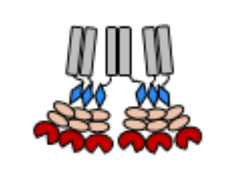
HYDRA-9: Detects Siglec-9 ligands

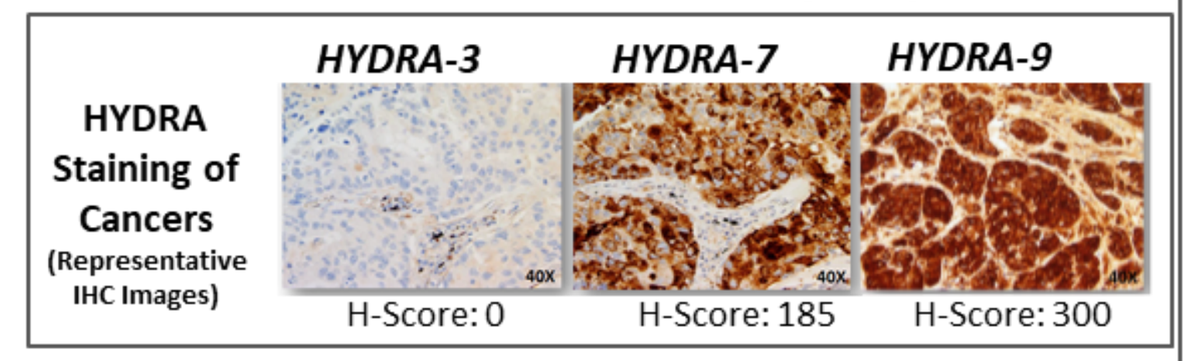
HYDRA Staining of Cancers (Representative IHC Images)
HYDRA-3
HYDRA-7
HYDRA-9
H-Score: 0
H-Score: 185
H-Score: 300

Applications

Precision Medicine: Tools to Guide Clinical Development

- Indication Prioritization
- Enrichment of patient segments based on *HYDRA*-stained biopsies (tumor sialylation)
- PK/PD relationship to guide optimal dosing (tumor and immune surface desialylation)

Results

Melanomas Have Increased HYDRA Scores Compared to Normal Skin

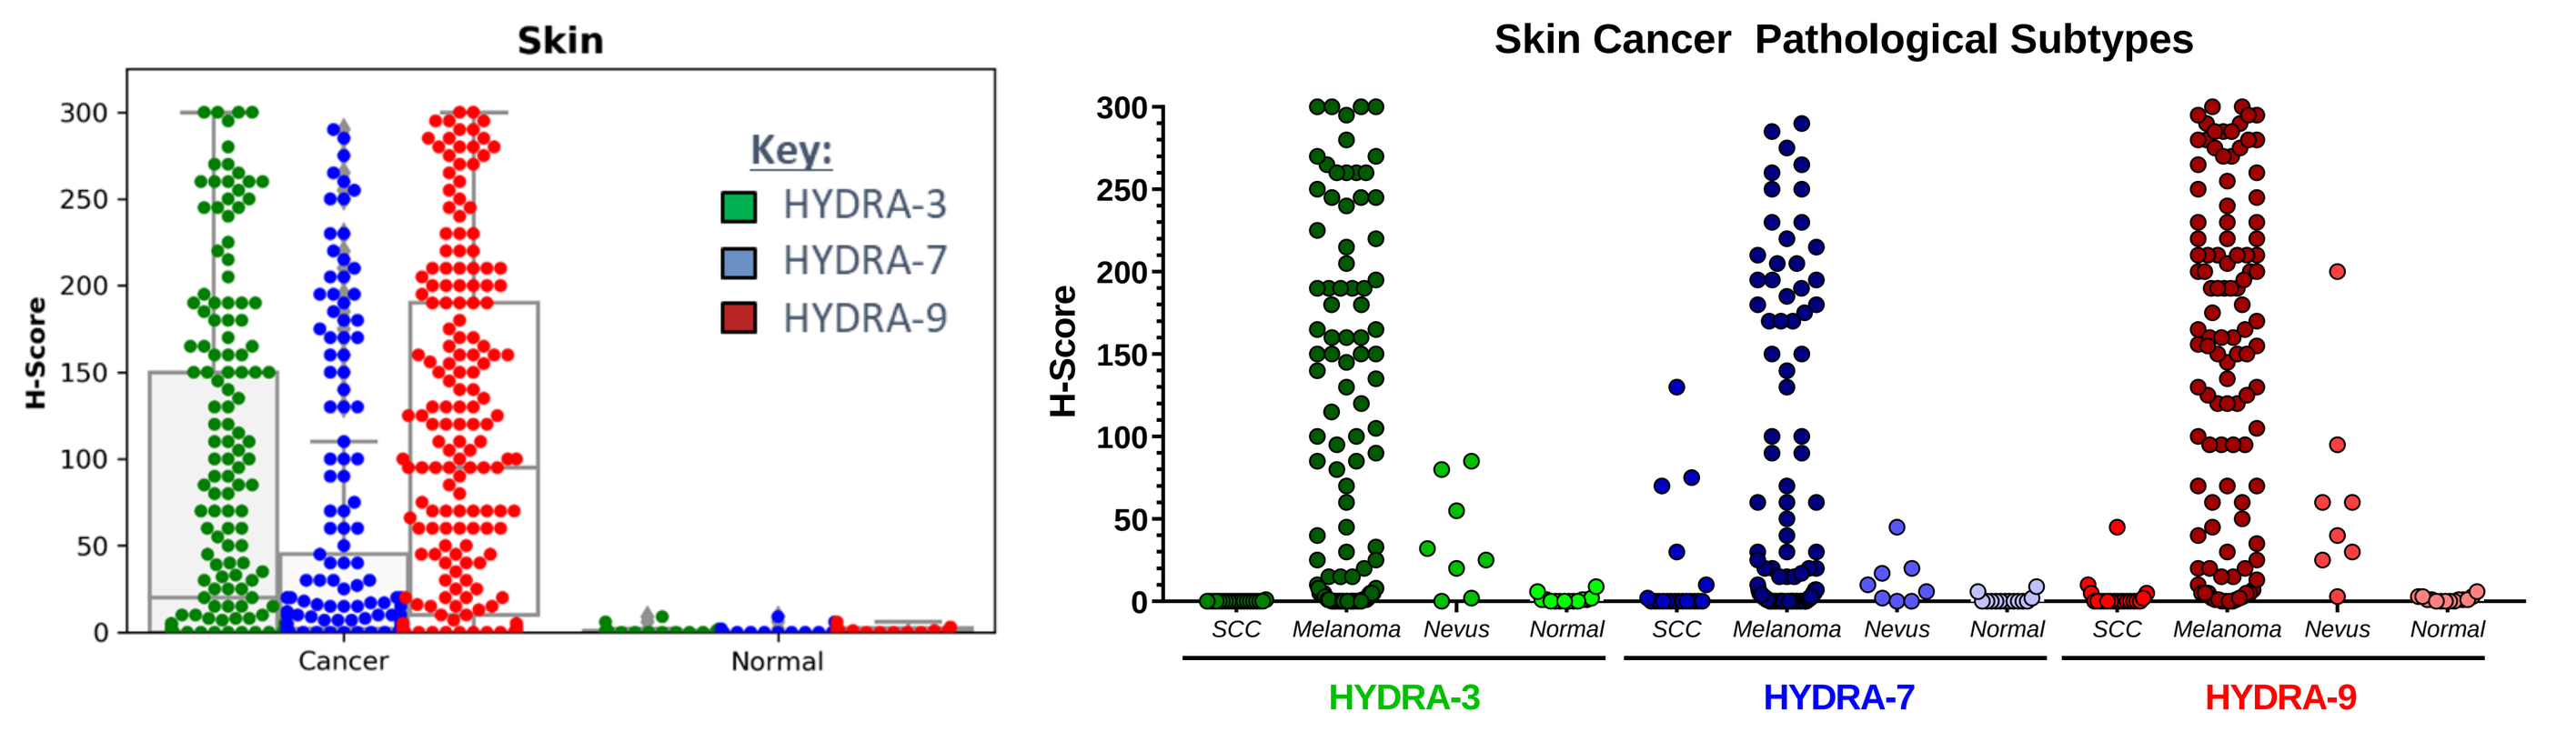
Skin
Key:
HYDRA-3
HYDRA-7
HYDRA-9
H-Score
Cancer
Normal
Skin Cancer Pathological Subtypes
SCC
Melanoma
Nevus
Normal

Figure 2: HYDRA profiling of skin cancers and normal skins. HYDRA staining was performed using commercially available tumor tissue microarray containing skin cancers (melanoma, squamous cell carcinoma (SCC)), nevus samples, and normal skin samples. High HYDRA-3, -7, and -9 scores were observed in most melanoma samples, while normal skin has very low or no HYDRA signals, suggesting that hypersialylation (a high density of sialoglycans) may play a role in melanoma pathogenesis.

Overview of the MGH Melanoma Cohort Received PD1 Therapies

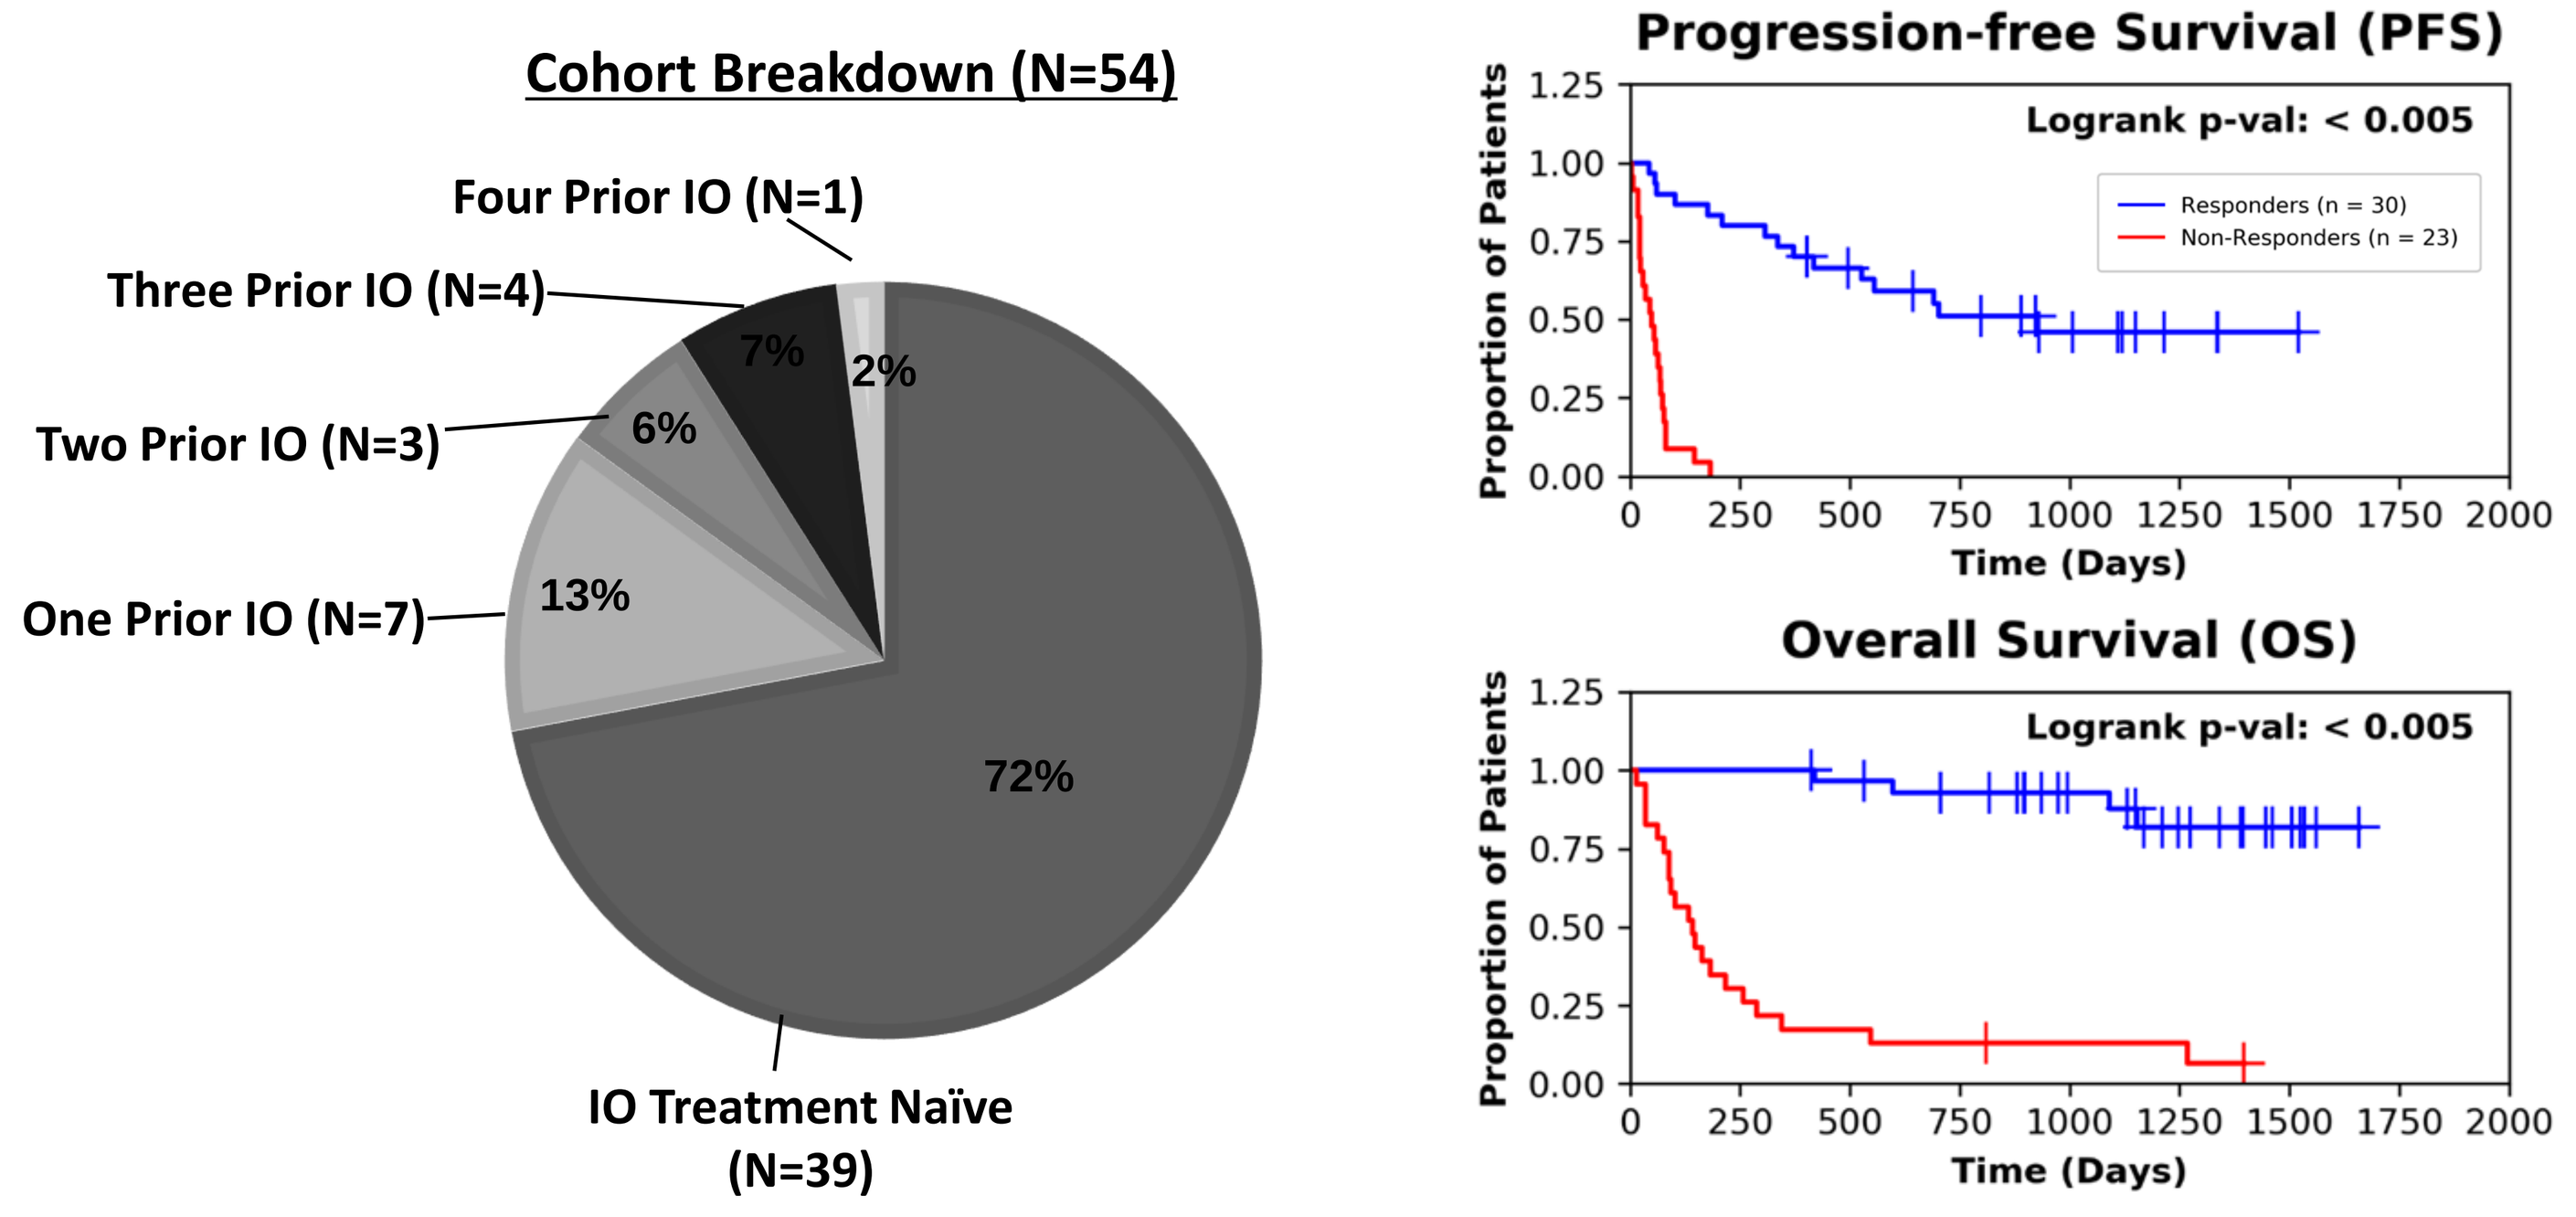
Cohort Breakdown (N=54)
Four Prior IO (N=1)
Three Prior IO (N=4)
Two Prior IO (N=3)
One Prior IO (N=7)
IO Treatment Naïve (N=39)
72%
13%
6%
7%
2%
Progression-free Survival (PFS)
Logrank p-val: < 0.005
Responders (n = 30)
Non-Responders (n = 23)
Overall Survival (OS)
Logrank p-val: < 0.005
Proportion of Patients
Time (Days)

Figure 3: Overview of the Melanoma Cohort. A clinically rich cohort was designed by Drs. Boland and Frederick to explore HYDRA expression in stage IV full melanoma resections. All patients received PD1 IO therapies. (A) Pie chart representation of the clinical cohort. 72% of patients are IO naïve patients and have tumor biopsies before the PD1 therapies. 28% patients received prior IO treatments and the number of prior IO treatments was denoted in the pie chart. (B) Kaplan-Meier curves of the PFS and OS of the cohort.

HYDRA IHC in Melanoma

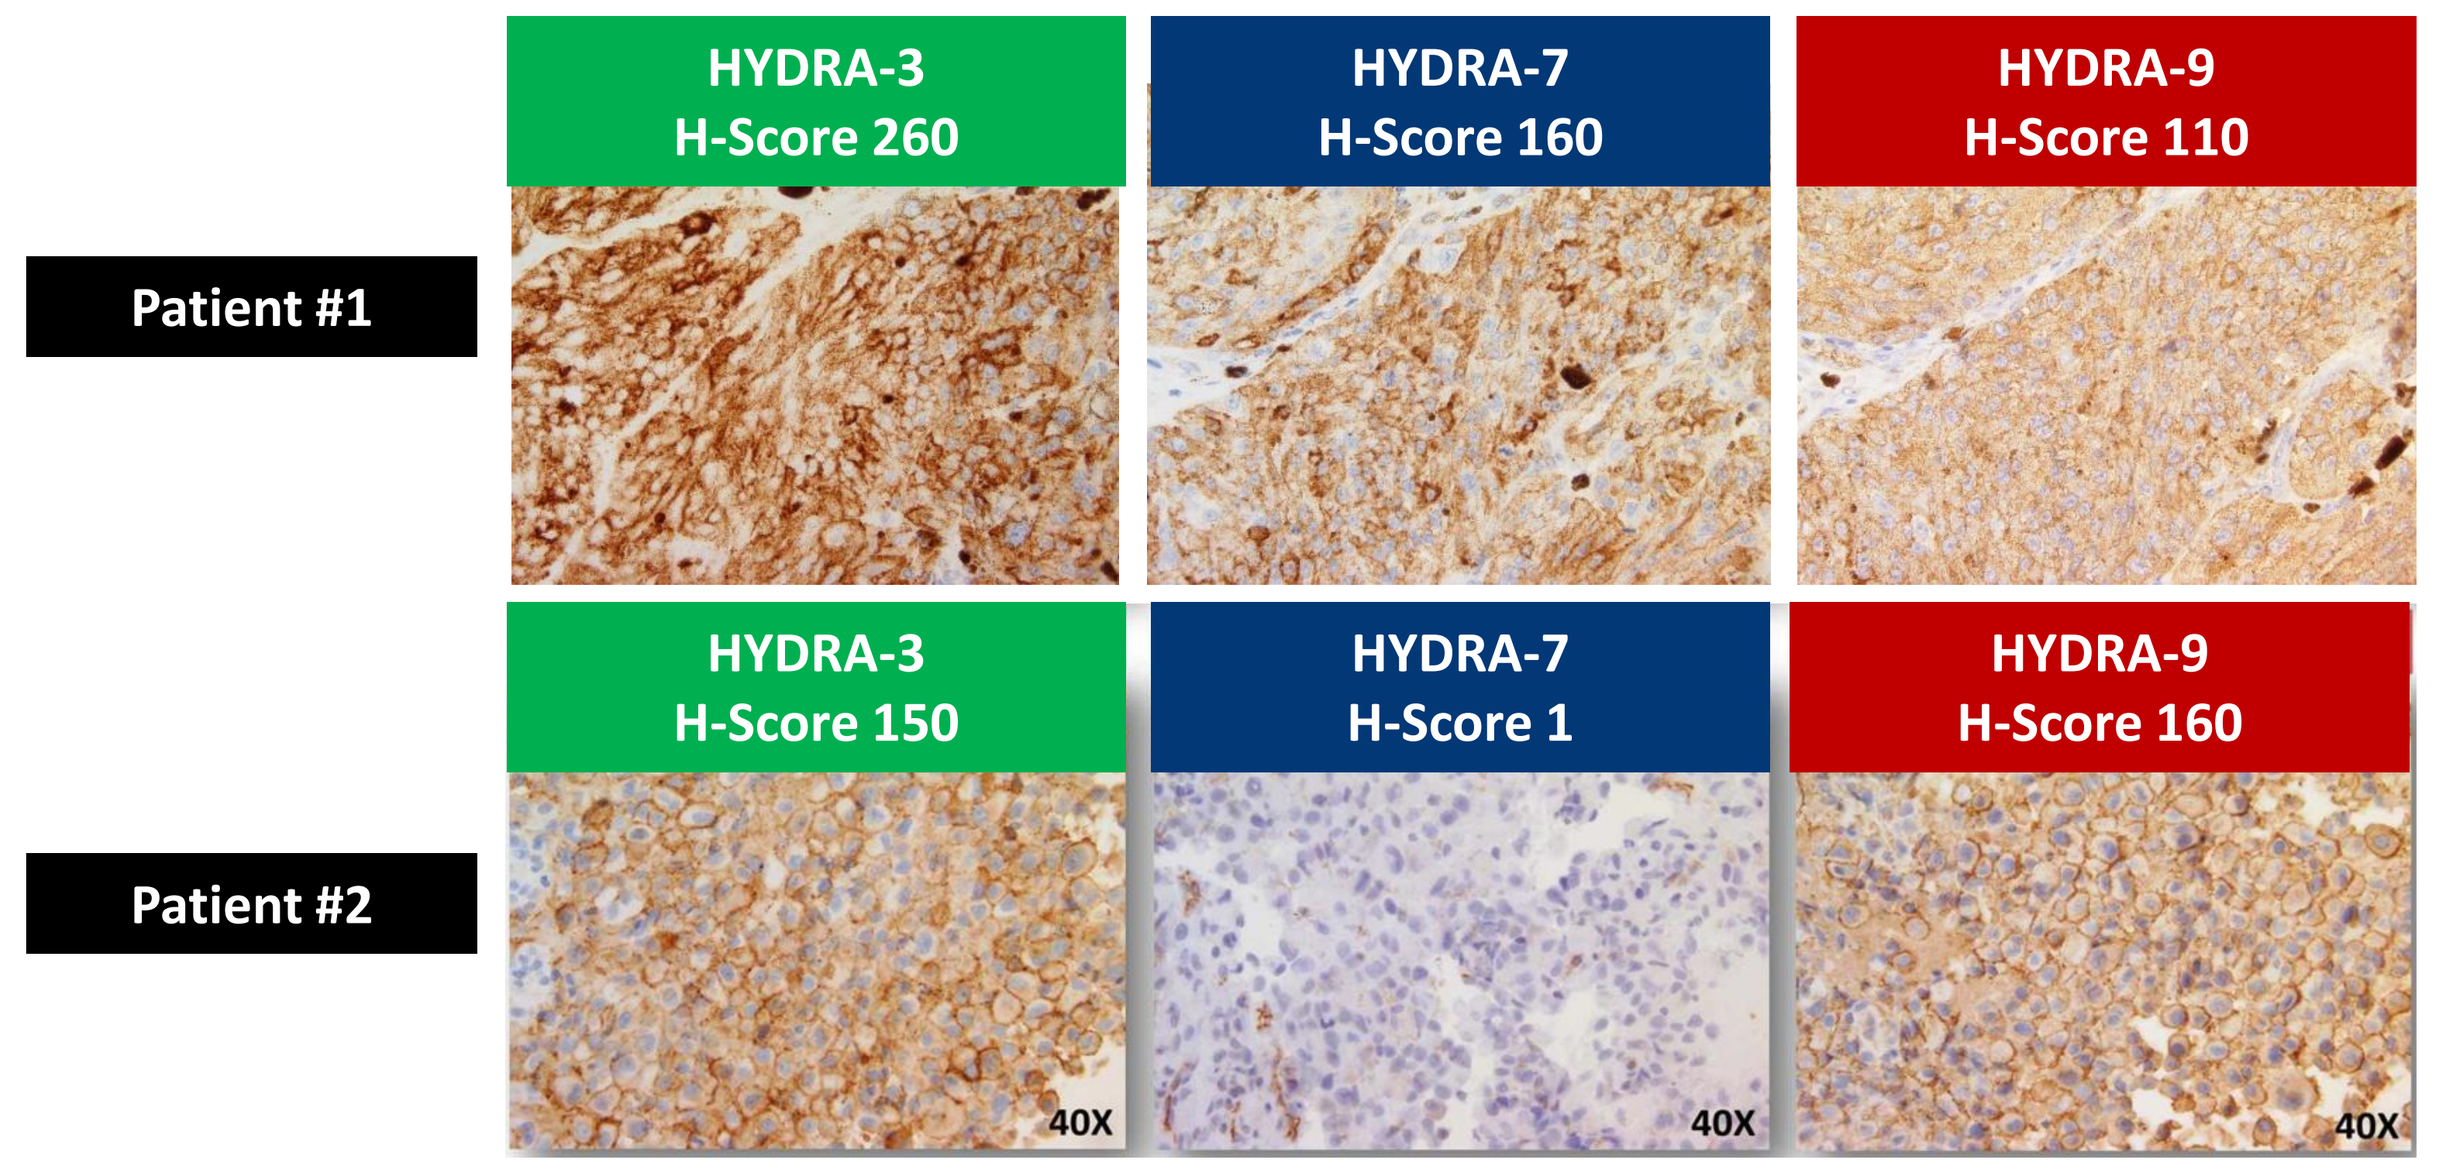
Patient #1
Patient #2
HYDRA-3
H-Score 260
HYDRA-7
H-Score 160
HYDRA-9
H-Score 110
HYDRA-3
H-Score 150
HYDRA-7
H-Score 1
HYDRA-9
H-Score 160
40X

Figure 4:Representative Images of the Melanoma Cohort. Representative images of two patients HYDRA IHC. Patient 1 (A-C) demonstrates robust HYDRA binding at 40x; Patient 2 (D-E) demonstrates robust HYDRA-3 and -9 binding but weak HYDRA-7 at 40x.

Patients With Multi-Siglec Ligand Correlate With Poor PFS and OS

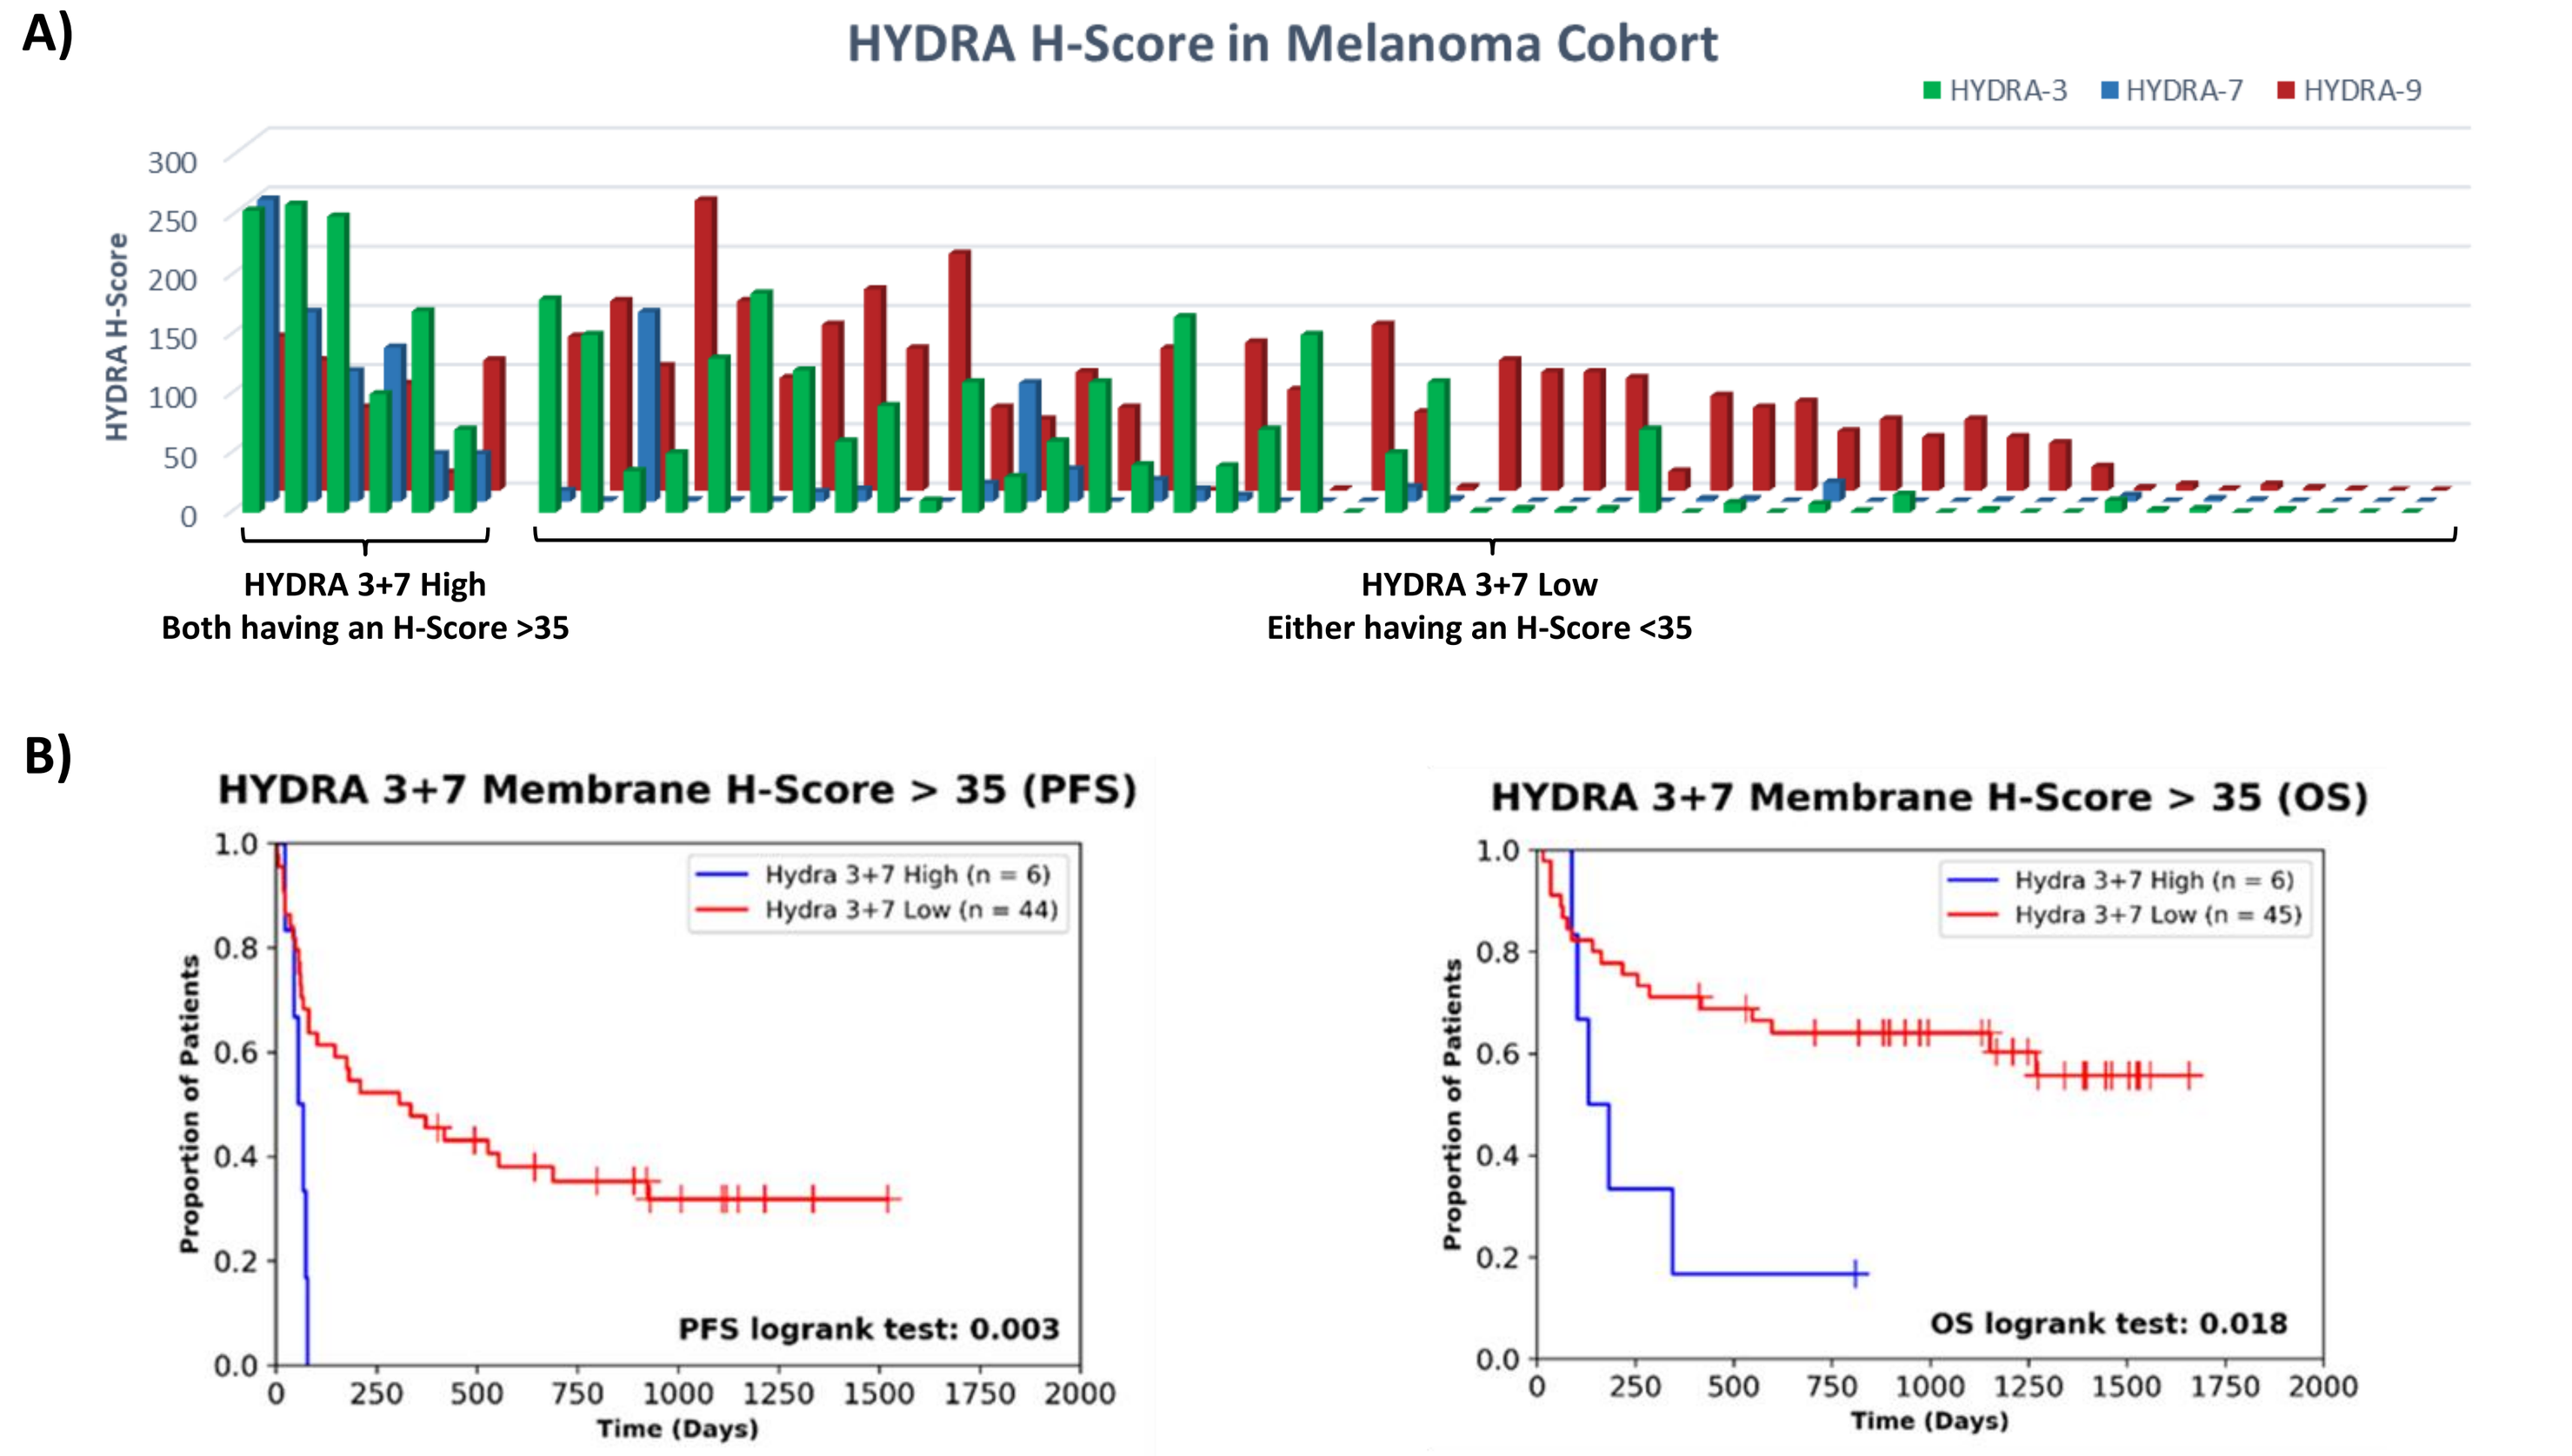
A)
HYDRA H-Score in Melanoma Cohort
HYDRA-3
HYDRA-7
HYDRA-9
HYDRA H-Score
HYDRA 3+7 High
Both having an H-Score >35
HYDRA 3+7 Low
Either having an H-Score <35
B)
HYDRA 3+7 Membrane H-Score > 35 (PFS)
Hydra 3+7 High (n = 6)
Hydra 3+7 Low (n = 44)
PFS logrank test: 0.003
HYDRA 3+7 Membrane H-Score > 35 (OS)
Hydra 3+7 High (n = 6)
Hydra 3+7 Low (n = 45)
OS logrank test: 0.018
Proportion of Patients
Time (Days)

Figure 5: HYDRA Profiling of this Melanoma Cohort and Correlation Analysis of HYDRAs with Clinical Outcome to PD1 Therapy (A) HYDRA profiling of the pre-treatment samples showed that most patients in this melanoma cohort have at least one HYDRA positivity with a similar HYDRA patten as the commercial melanoma TMAs. (B) Using unbiased univariate Kaplan-Meier curves, we binned each HYDRA alone and in combination to determine correlations with PFS and OS. We found that patients with HYDRA-3 and -7 (> 35) correlate with worse outcome of PD1 therapies in this cohort. Patients with HYDRA-3 and -7 tended to also have the HYDRA-9 signature.

Melanoma Biomarkers of Liver Metastasis, TIL, BRAF, and PDL1 Status Do Not Correlate with PFS or OS In This Cohort

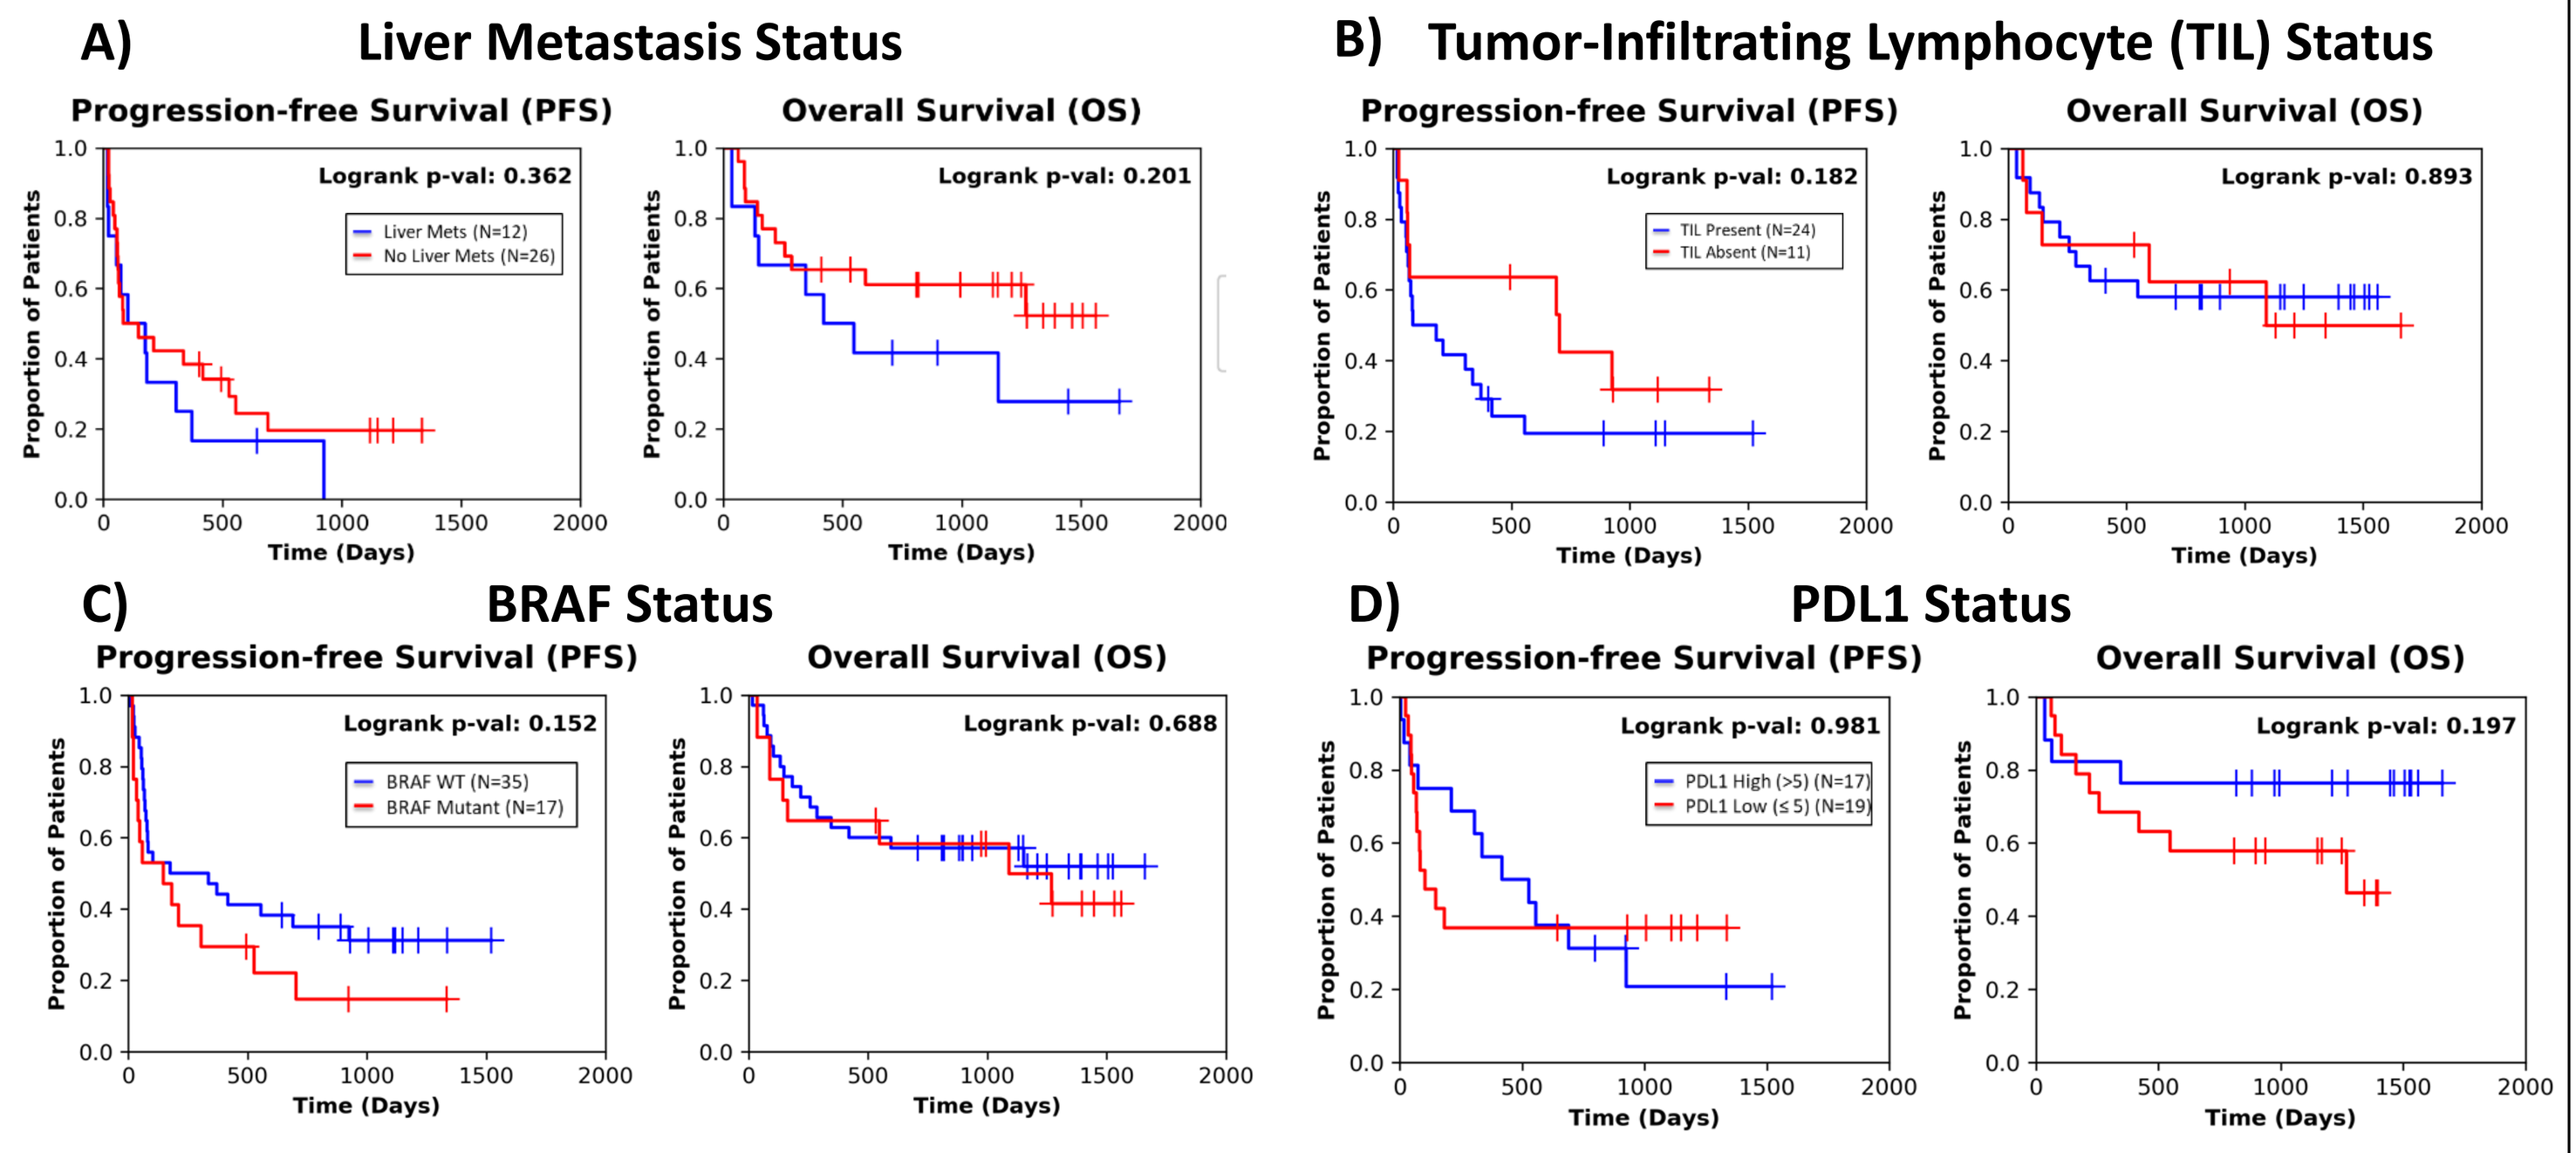
A) Liver Metastasis Status
Progression-free Survival (PFS)
Logrank p-val: 0.362
Liver Mets (N=12)
No Liver Mets (N=26)
Overall Survival (OS)
Logrank p-val: 0.201
B) Tumor-Infiltrating Lymphocyte (TIL) Status
Logrank p-val: 0.182
TIL Present (N=24)
TIL Absent (N=11)
Logrank p-val: 0.893
C) BRAF Status
Logrank p-val: 0.152
BRAF WT (N=35)
BRAF Mutant (N=17)
Logrank p-val: 0.688
D) PDL1 Status
Logrank p-val: 0.981
PDL1 High (>5) (N=17)
PDL1 Low (≤ 5) (N=19)
Logrank p-val: 0.197
Proportion of Patients
Time (Days)

Figure 6: Correlation Analysis of Melanoma Biomarkers (including Liver Mets, TILs, BRAF and PDL1) With Clinical Outcome. Typical clinical melanoma markers, independent of HYRDAs, do not correlate with PFS nor OS by Kaplan–Meier univariate testing. A) The presence of liver metastases has been correlated with poorer outcome in the literature[3]. However, in this cohort liver metastasis status showed a trend with poor OS but was not statically significant. B) The presence or absence of TILs in the primary melanoma lesion, C) BRAF wild type or any mutation, and D) PDL1 did not correlate with clinical outcome in this cohort.

HYDRA Signatures Are Independent of Liver Metastasis, TIL and BRAF Biomarkers But HYDRA-9 Correlates With PDL1

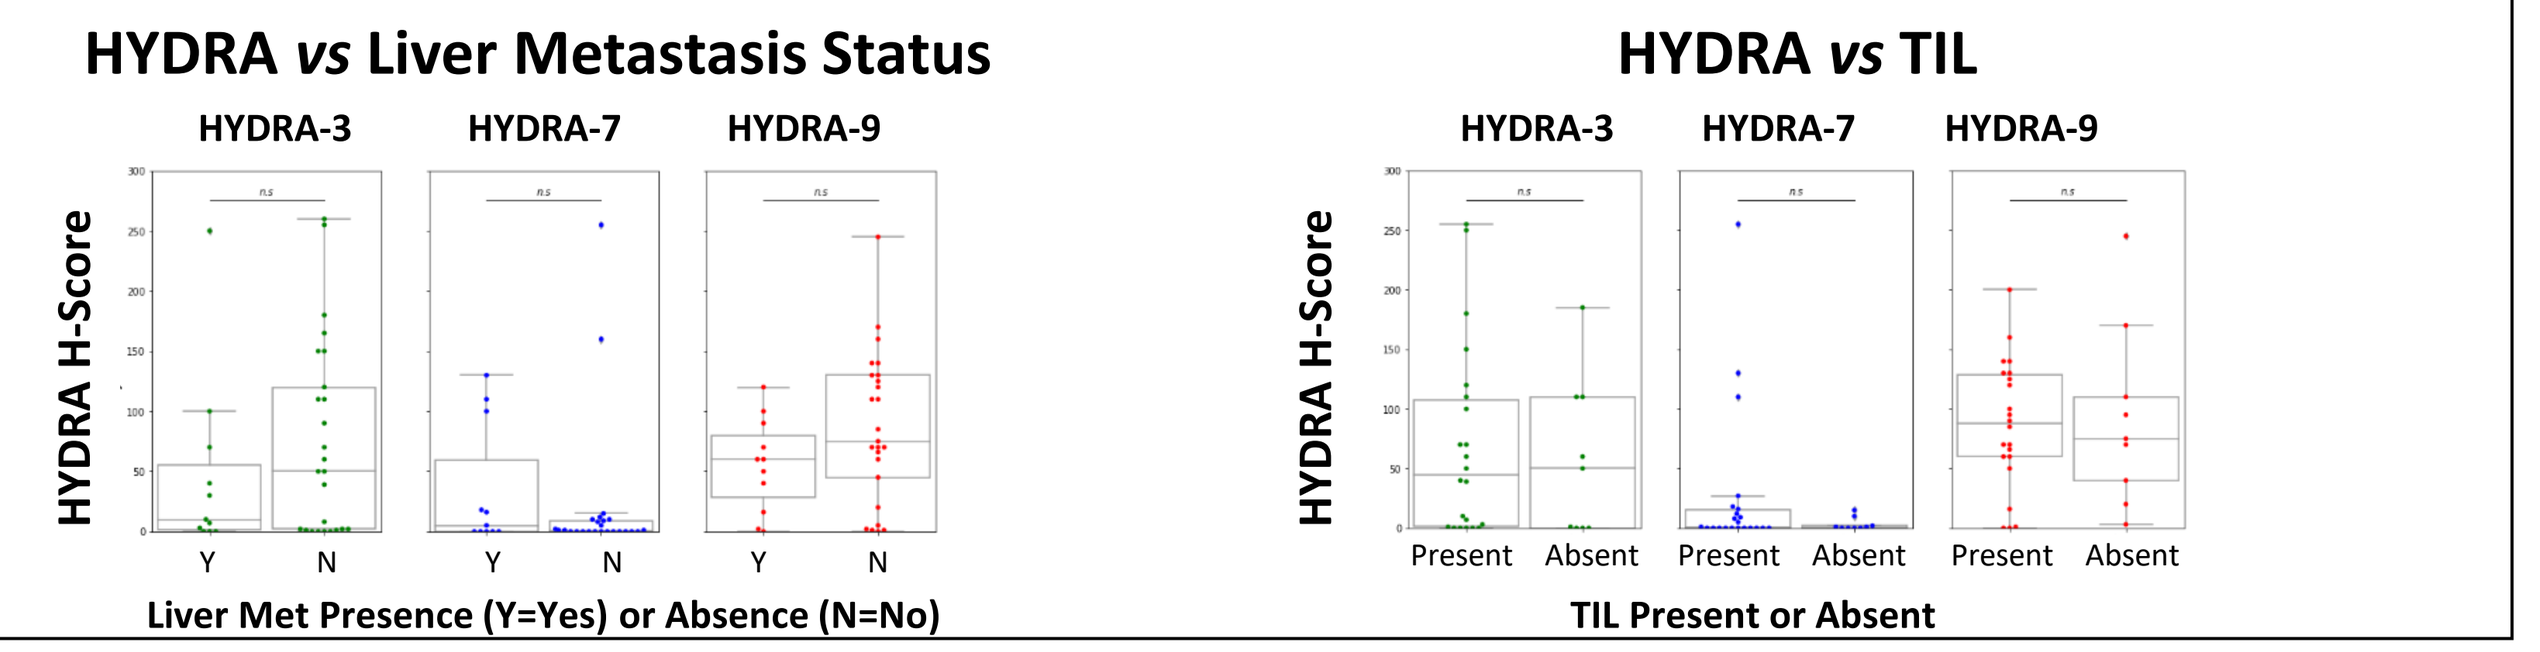
HYDRA vs Liver Metastasis Status
HYDRA-3
HYDRA-7
HYDRA-9
HYDRA H-Score
Y
N
Liver Met Presence (Y=Yes) or Absence (N=No)
HYDRA vs TIL
Present
Absent
TIL Present or Absent

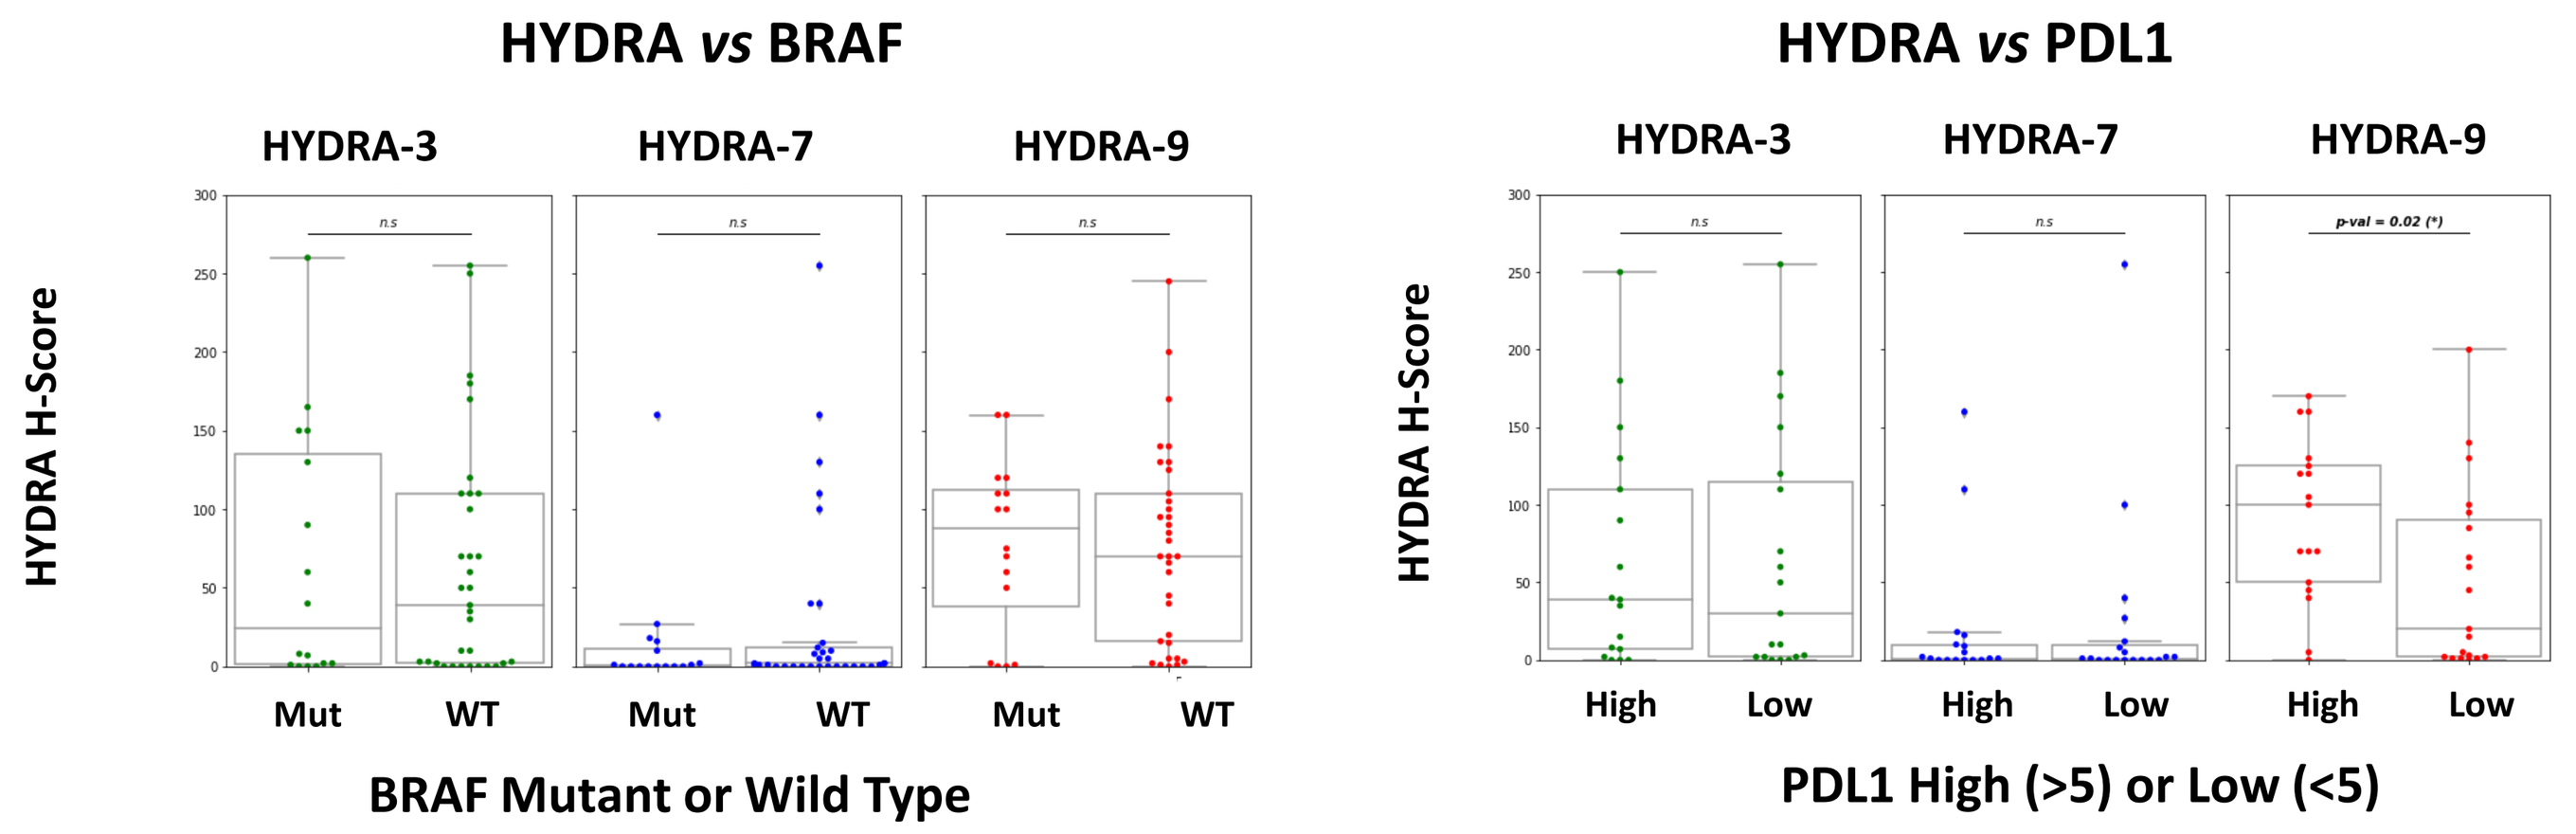
HYDRA vs BRAF
HYDRA-3
HYDRA-7
HYDRA-9
HYDRA H-Score
Mut
WT
BRAF Mutant or Wild Type
HYDRA vs PDL1
High
Low
PDL1 High (>5) or Low (<5)

Figure 7 Correlation Analysis Between HYDRAs and Biomarkers of Liver Metastasis Status, TIL, BRAF, and PDL1. HYDRAs are independent of liver metastasis status, TIL and BRAF. HYDRA-9 was significantly higher in PDL1 high patients by Mann Whitney-U Test.

Ongoing Longitudinal Study

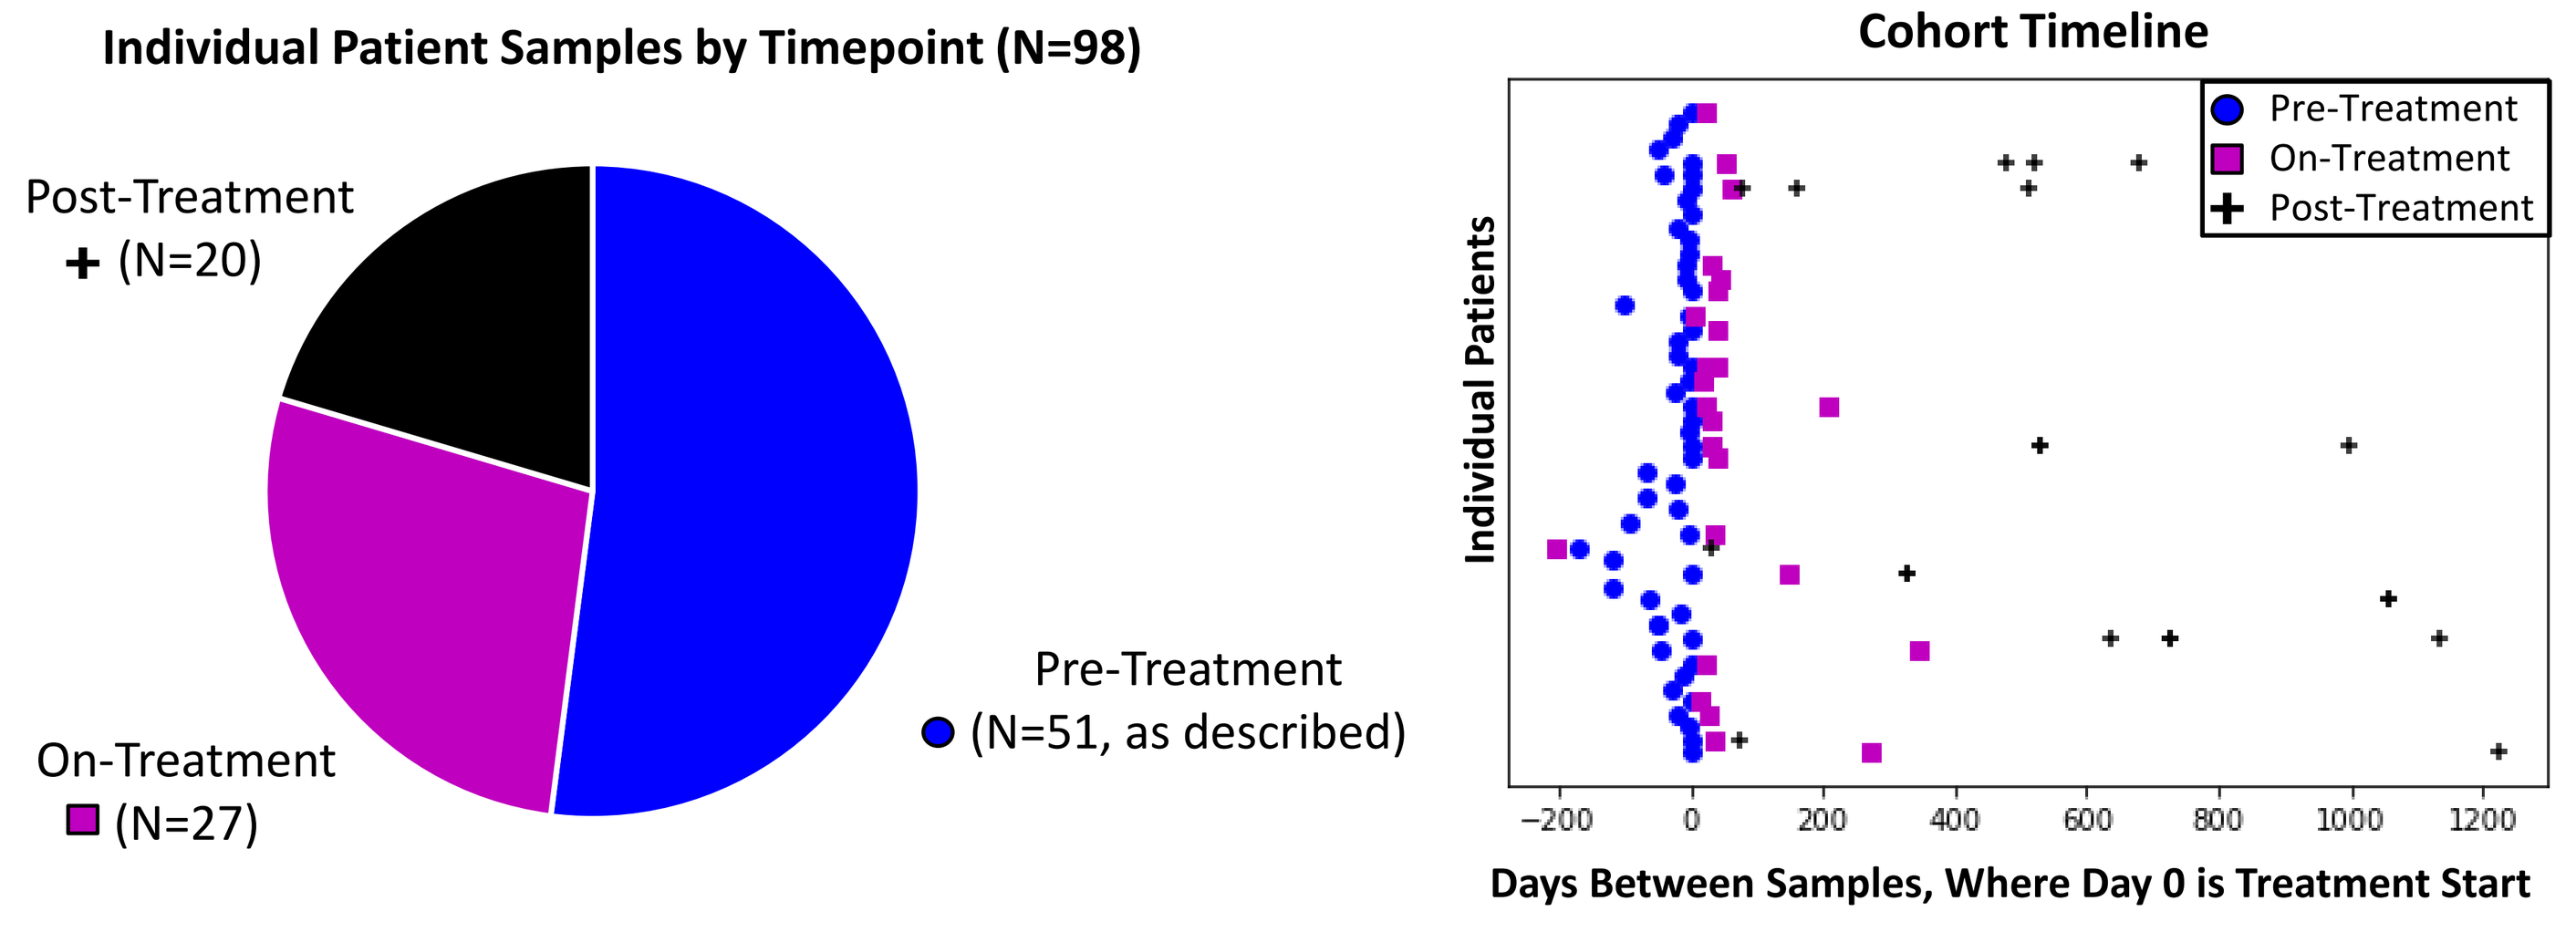
Individual Patient Samples by Timepoint (N=98)
Post-Treatment
(N=20)
On-Treatment
(N=27)
Pre-Treatment
(N=51, as described)
Cohort Timeline
Pre-Treatment
On-Treatment
Post-Treatment
Individual Patients
Days Between Samples, Where Day 0 is Treatment Start

Figure 8 Overview of Longitudinal Cohort In Progress: Visualization of the longitudinal cohort whose HYDRA staining is currently ongoing. (Left) Displays total patient count in each treatment category and sample number in total. In addition to the Pre-Treatment cohort analyzed here (n=51), this new cohort includes a matching set of 27 on-treatment samples and 20 post-treatment samples from depicted patients. (Right) Displays cohort timeline with days between collected samples where day 0 is treatment initiation. Pre-treatment samples are denoted in blue circles, on-treatment in magenta squares, and post-treatment in black plus signs. Some patients have multiple samples collected at these treatment time points. Status of pre/on/post are decided upon a desired fixed point in their clinical history; not all patients are treatment naïve.

Conclusions

- Sialoglycans are substantially upregulated in melanoma compared to normal skin as profiled by sialoglycan-binding Siglec-based HYDRA-3, -7 and -9
- Pretreatment patient samples with multiple HYDRA signatures tended to be refractory to PD1 therapies in this cohort of stage IV melanoma.
- Most HYDRA signatures are independent of liver metastases, TILs and BRAF biomarkers, but HYDRA-9 correlated with PDL1 expression in this cohort.

Acknowledgements

We would like to thank Brian Quattrochi for assistance with pathology and feedback during analysis process; Adriano Piris for pathologic scoring of PDL1; Hongli Chen for assistance with 40x IHC images; Robert Sikorski for his clinical insight and advice; and Lihui Xu for her guidance during HYDRA production.

References

1: Smith, B., & Bertozzi, C. R. (2021). The clinical impact of glycobiology: targeting selectins, Siglecs and mammalian glycans. Nature reviews. Drug discovery, 20(3), 217–243. https://doi.org/10.1038/s41573-020-00093-1

2: Edgar LJ et. al. Sialic acid ligands of CD28 block co-stimulation of T cells bioRxiv 2021.02.22.432333; https://doi.org/10.1101/2021.02.22.432333

3: Tumeh PC et al. Liver Metastasis and Treatment Outcome with Anti-PD-1 Monoclonal Antibody in Patients with Melanoma and NSCLC. Cancer Immunol Res. 2017;5(5):417-424. doi:10.1158/2326-6066.CIR-16-0325