A Novel Therapeutic Modality of Inhibiting the Glyco-Immune Checkpoint Axis to Treat Cancer

LB-109

Lizhi Cao[1], Adam Petrone[1], Wayne Gatlin[1], Jenny Che[1], Abhishek Das[1], Robert LeBlanc[1], Zakir Siddiquee[1], Sujata Nerle[1], Michal Stanczak[2], Michele Mayo[1], Lihui Xu[1], Karl Normington[1], Jeff Brown[1], Wei Yao[1], Carolyn Bertozzi[3], James Broderick[1], Heinz Läubli[2], Li Peng[1]

[1]Palleon Pharmaceuticals, Waltham, MA, USA, [2] Cancer Immunology Laboratory, University Hospital Basel, Switzerland, [3] Department of Chemistry, Stanford University, Stanford, CA, USA

Introduction

Cancer therapy has been revolutionized by the recent developments of immune-checkpoint inhibitors to harness the power of the immune system in fighting cancer. However, the majority of patients fail to have durable responses or become resistant to immuno-oncology drugs, highlighting the need to identify new mechanisms of immune evasion in cancer and to develop new therapeutic modalities. Recently, the glyco-immune checkpoint axis (sialoglycan/Siglec pathway) has emerged as a novel mechanism of immune regulation involving both innate and adaptive immunity and an important mechanism of cancer immune escape[1-3]. Upon ligation of sialylated glycans to ITIM-containing Siglecs on immune cells, this pathway plays a previously unrecognized role in regulating functions of NK cell, macrophages, dendritic cells, monocytes and T-cells in the tumor microenvironment. It suppresses multiple facets of anti-cancer immunity, including cancer antigen release, cancer antigen presentation, priming and activation of anti-cancer T-cell immunity, which may represent a novel mechanism of resistance to current immunotherapy. Here, we described a novel therapeutic modality, a multi-functional antibody-like molecule named *EAGLE* (Enzyme-Antibody Glyco-Ligand Editing), to inhibit the glyco-immune checkpoint axis in the tumor microenvironment by selectively removing the terminal sialic acids of sialoglycans on tumor cells.

Glyco-Immune Checkpoints Suppress Innate and Adaptive Immunity

Glyco-Immune Checkpoints:

- Glycan-sensing receptors that block immune activation; play healthy role in self vs non-self discrimination
- Major receptor families: Siglecs (10 unique to human), some CTLs
- Broad expression across various innate and adaptive immune cells
- Exploited by cancer for immune evasion

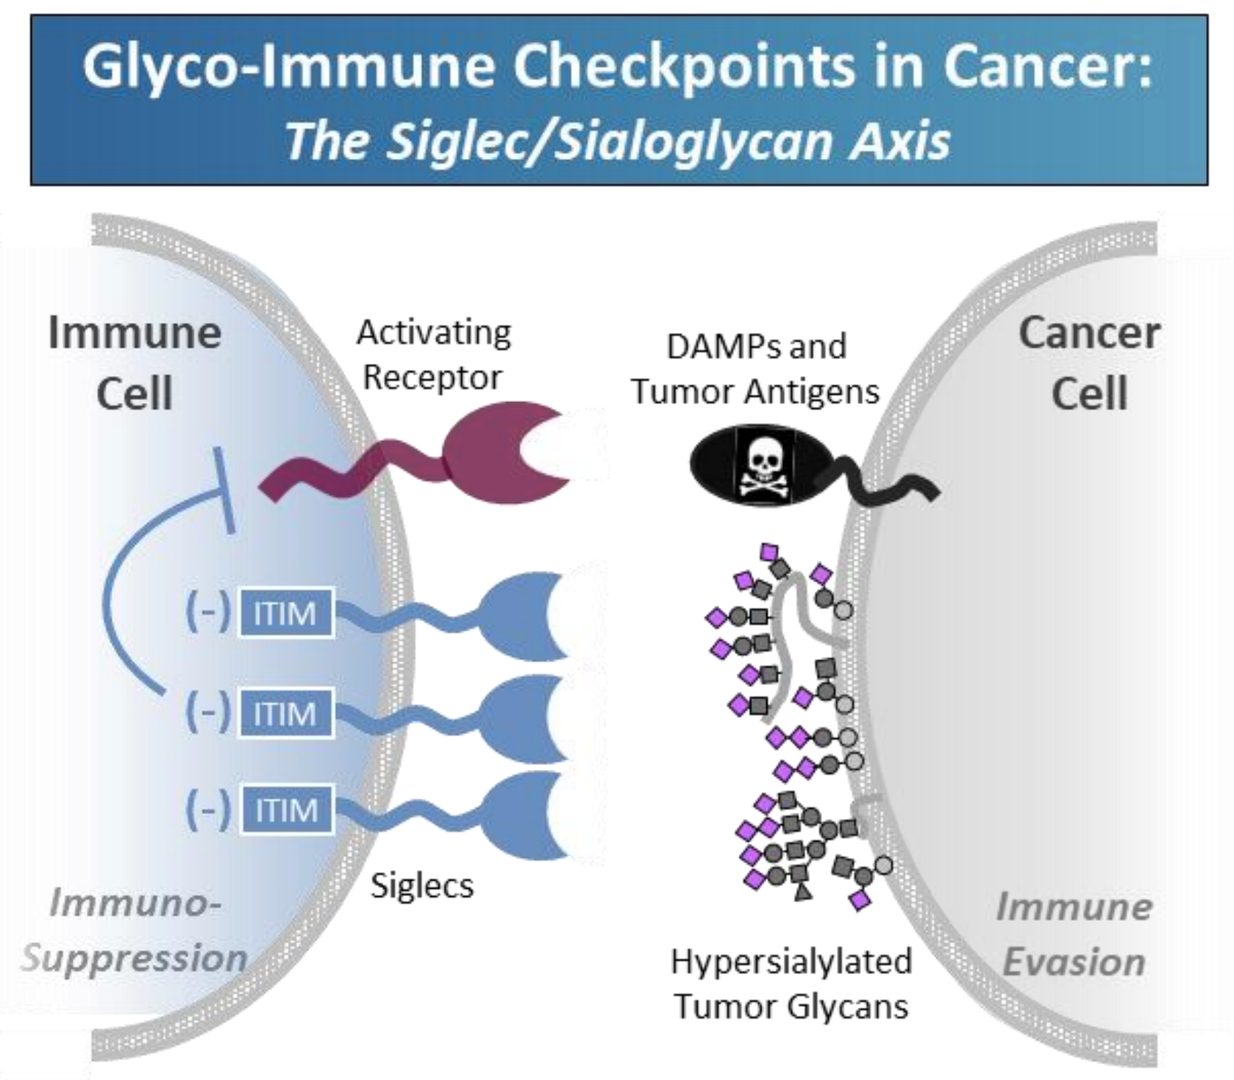
Glyco-Immune Checkpoints in Cancer:
The Siglec/Sialoglycan Axis
Immune Cell
Activating Receptor
DAMPs and Tumor Antigens
Cancer Cell
(-) ITIM
(-) ITIM
(-) ITIM
Siglecs
Immuno-Suppression
Hypersialylated Tumor Glycans
Immune Evasion

Tumor Glycan Evolution:

- Tumors evolve and cloak themselves with functionally immunosuppressive glycan patterns
- Several common tumor glycans: ST, STn, SLe^x, SLe^a, GM2, GD3, PSA
- Recurrent pattern: **Hypersialylation**
- On tumor cell surface glyco-proteins, mucins, and glycolipids

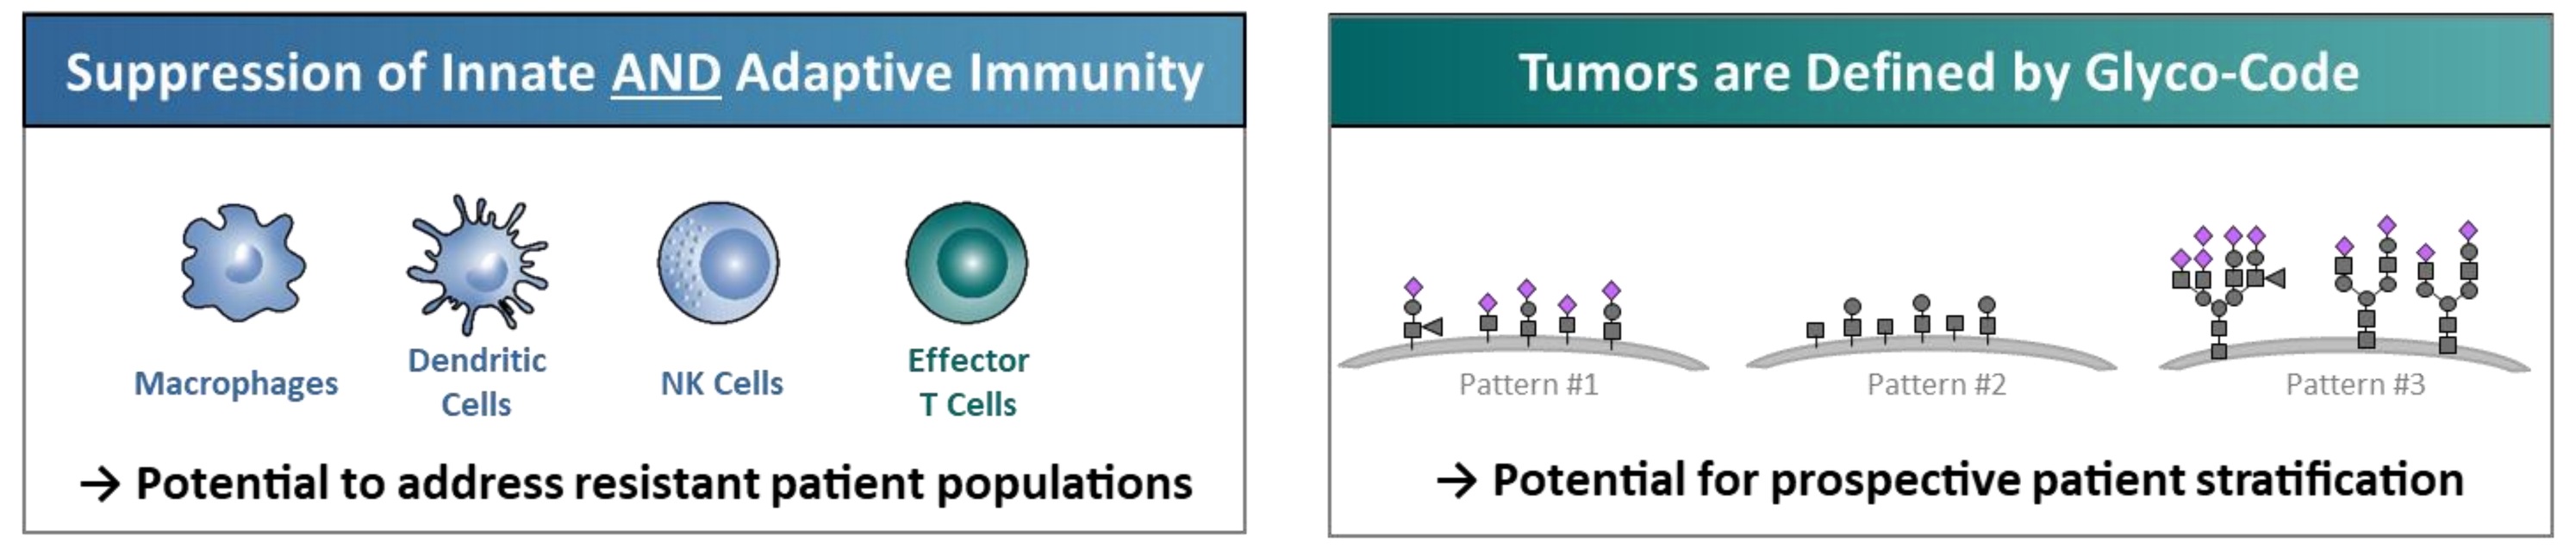
Suppression of Innate AND Adaptive Immunity
Macrophages
Dendritic Cells
NK Cells
Effector T Cells
→ Potential to address resistant patient populations
Tumors are Defined by Glyco-Code
Pattern #1
Pattern #2
Pattern #3
→ Potential for prospective patient stratification

Figure 1. Schematic representation of glyco-immune checkpoints axis. At the immunological synapse, Siglecs (sialic acid-binding Ig-like lectins) on immune cells interact with tumor associated sialoglycans and recruit SHP phosphatases, dampening immune responses. Siglecs are expressed on macrophages, monocytes, DC, and NK cells, recognizing a variety of tumor associated glycans which contain a terminal sialic acid. The Siglec/Sialoglycan plays important roles in cancer cell killing by NK cells and macrophages, cancer antigen presentation, T cell priming and activation, and cancer cell killing by T effector cells.

Results

Development of *EAGLE* (Enzyme-Antibody Glyco-Ligand Editing) Platform

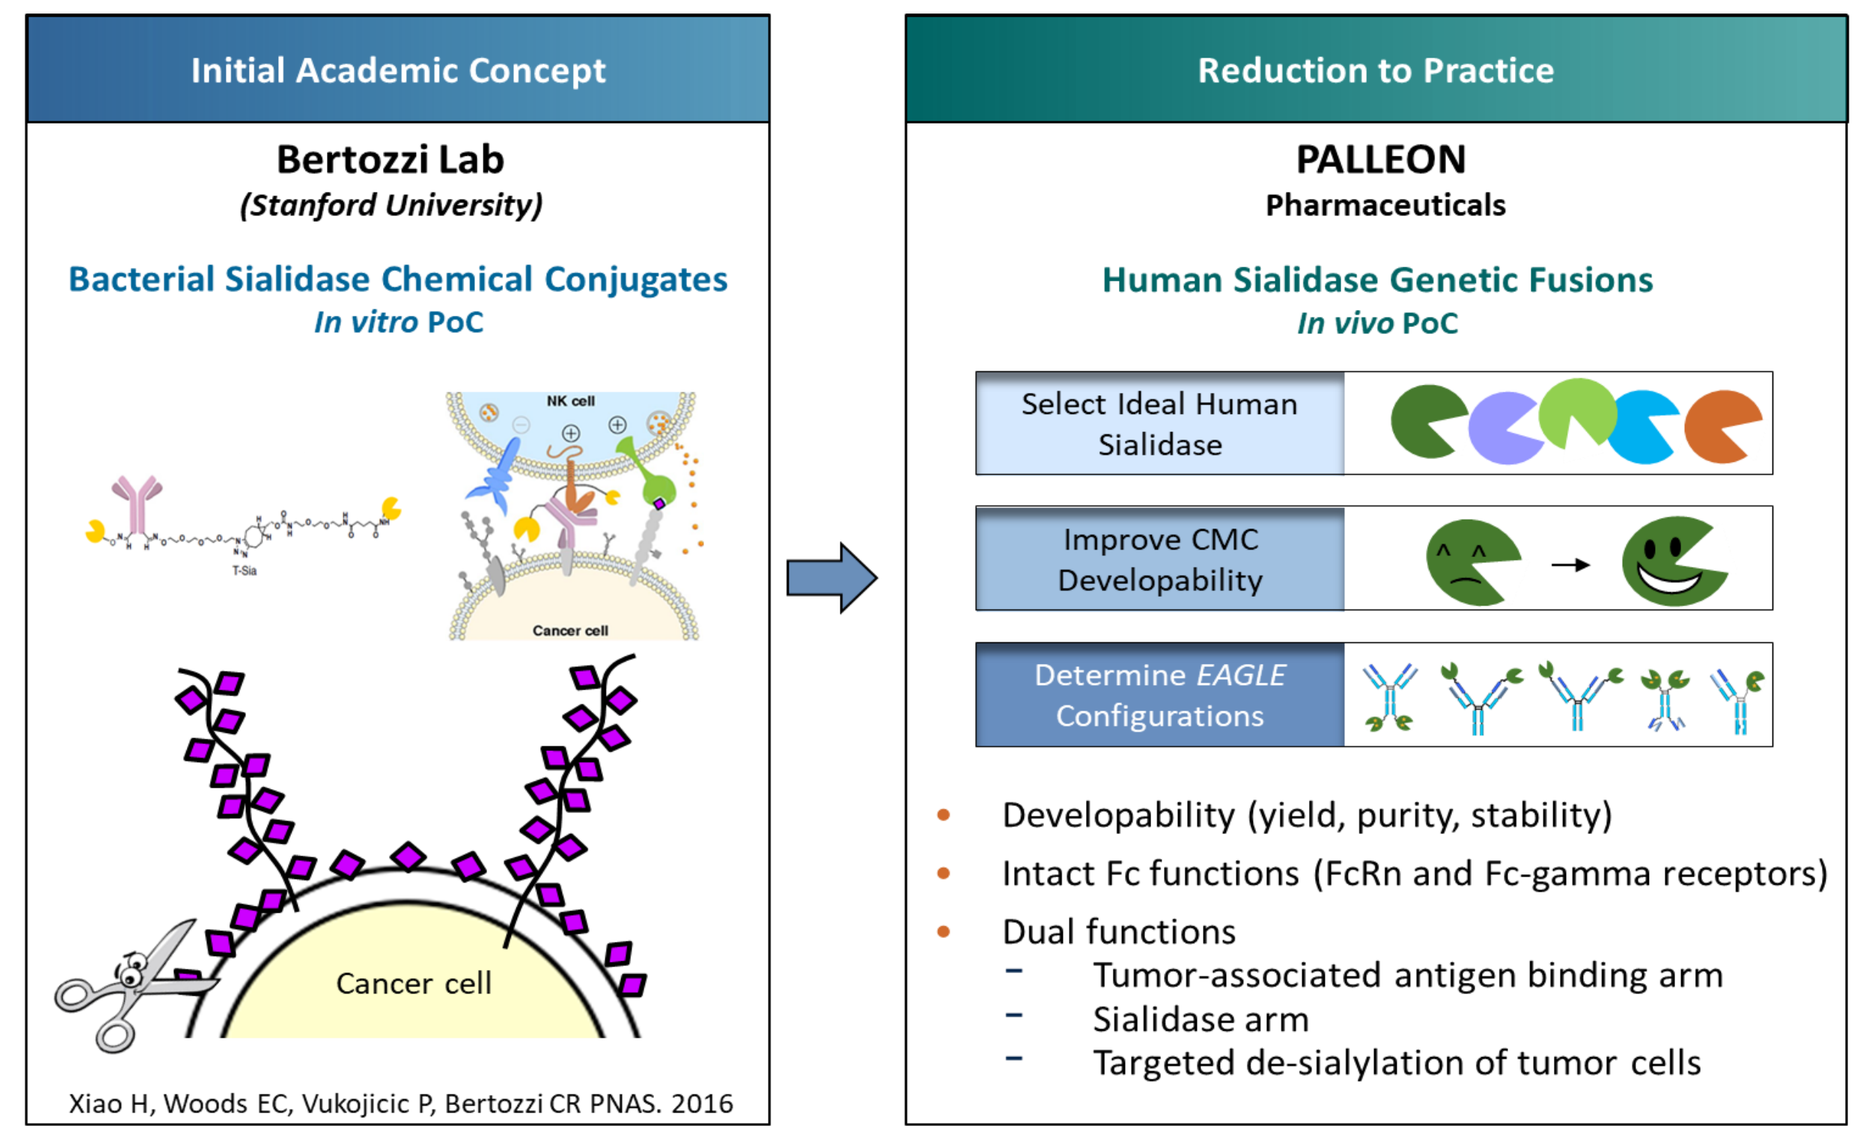
Initial Academic Concept
Bertozzi Lab
(Stanford University)
Bacterial Sialidase Chemical Conjugates
In vitro PoC
Cancer cell
Xiao H, Woods EC, Vukojicic P, Bertozzi CR PNAS. 2016
Reduction to Practice
PALLEON
Pharmaceuticals
Human Sialidase Genetic Fusions
In vivo PoC
Select Ideal Human Sialidase
Improve CMC Developability
Determine EAGLE Configurations
Developability (yield, purity, stability)
Intact Fc functions (FcRn and Fc-gamma receptors)
Dual functions
Tumor-associated antigen binding arm
Sialidase arm
Targeted de-sialylation of tumor cells

Figure 2. Evolution of *EAGLE* platform. (Left) Schematic representation of antibody-sialidase chemical conjugates using bacterial sialidase. Conjugating bacterial sialidase to antibodies which recognize tumor-associated antigens enables targeted removal of sialic acids of sialoglycans on tumor cells[4]. (Right) Development of *EAGLE* platform. *EAGLE* is a bi-functional antibody-sialidase fusion protein consisting of an engineered human sialidase with improved developability and enzyme activity.

EAGLEs Showed Targeted Cleavage of Sialic Acids from Her2+ Tumor Cells

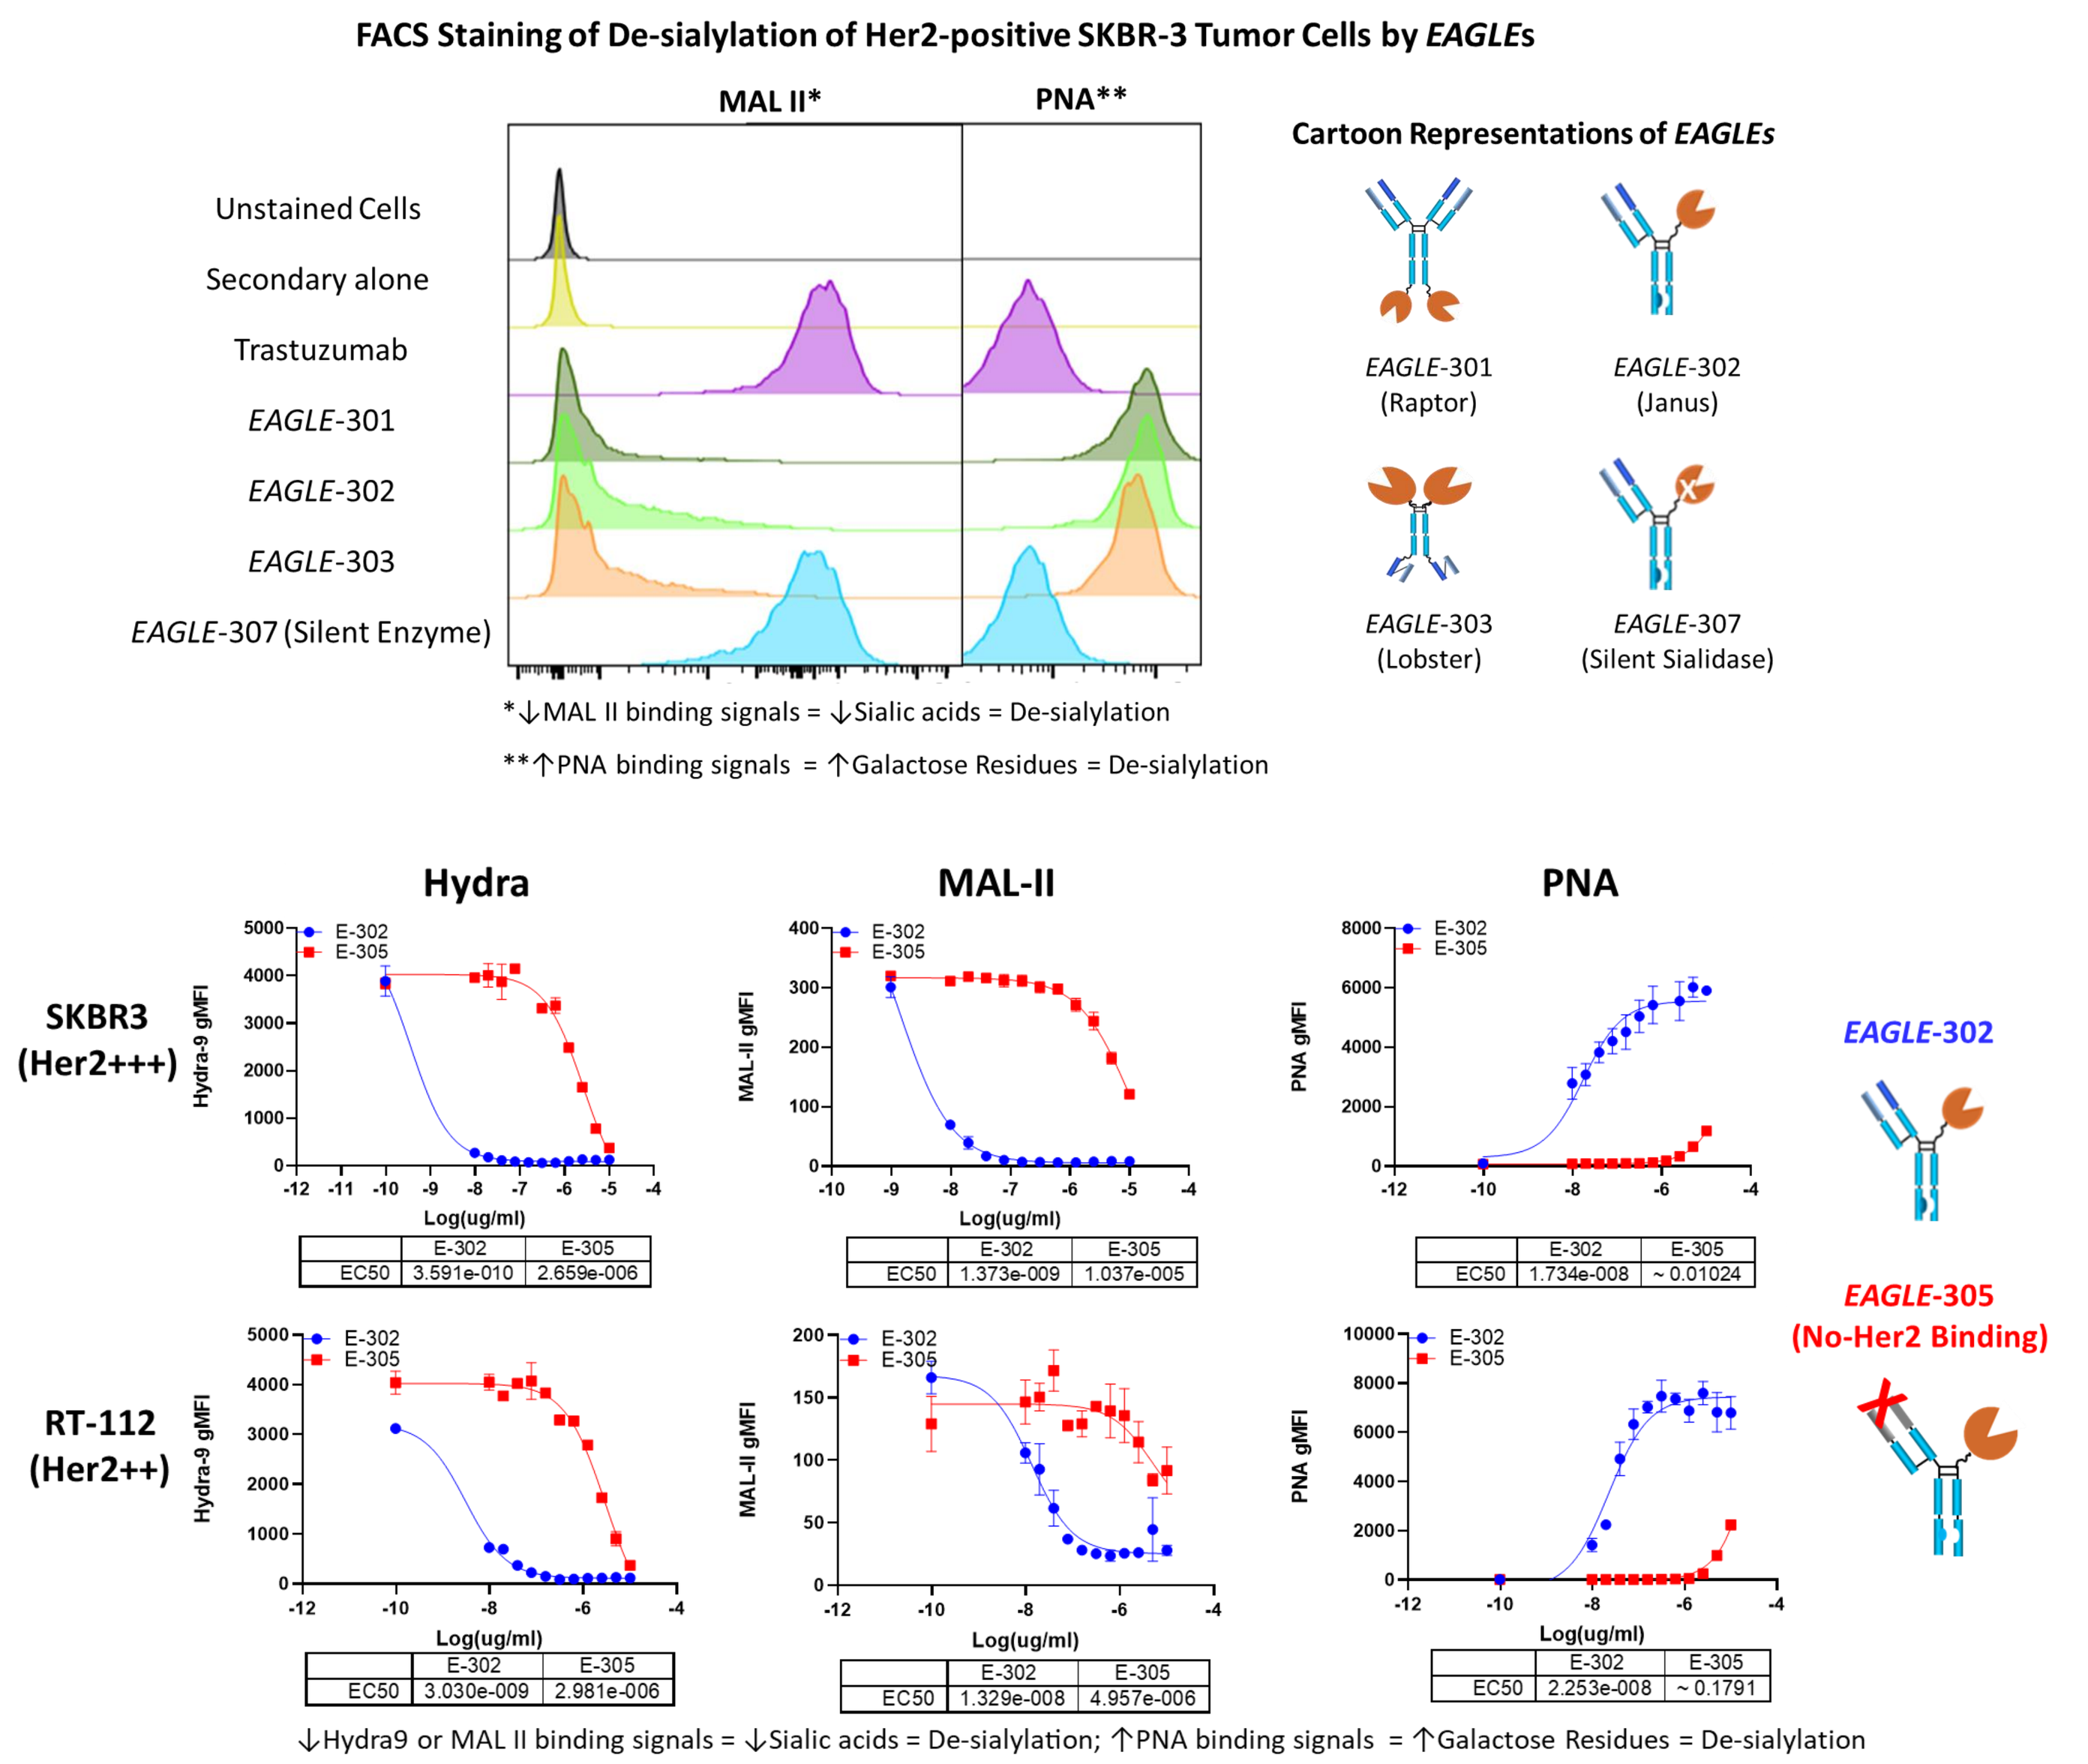
FACS Staining of De-sialylation of Her2-positive SKBR-3 Tumor Cells by EAGLEs
MAL II*
PNA**
Unstained Cells
Secondary alone
Trastuzumab
EAGLE-301
EAGLE-302
EAGLE-303
EAGLE-307 (Silent Enzyme)
*↓MAL II binding signals = ↓Sialic acids = De-sialylation
**↑PNA binding signals = ↑Galactose Residues = De-sialylation
Cartoon Representations of EAGLEs
EAGLE-301 (Raptor)
EAGLE-302 (Janus)
EAGLE-303 (Lobster)
EAGLE-307 (Silent Sialidase)
Hydra
MAL-II
PNA
SKBR3 (Her2+++)
RT-112 (Her2++)
E-302
E-305
Log(ug/ml)
EC50 3.591e-010 2.659e-006
EC50 1.373e-009 1.037e-005
EC50 1.734e-008 ~ 0.01024
EC50 3.030e-009 2.981e-006
EC50 1.329e-008 4.957e-006
EC50 2.253e-008 ~ 0.1791
EAGLE-302
EAGLE-305 (No-Her2 Binding)
↓Hydra9 or MAL II binding signals = ↓Sialic acids = De-sialylation; ↑PNA binding signals = ↑Galactose Residues = De-sialylation

Figure 3. FACS staining of desialylation of tumor cells treated with *EAGLEs*. (Top) Compared to the control group of trastuzumab treatment, various *EAGLEs* (301, 302, and 303) decreased sialic acid levels on Her2-positive cancer cells. (Bottom) Her2-targeting moiety of *EAGLE* enabled more efficient de-sialylation of tumor cells. *EAGLE*-302 with a trastuzumab arm demonstrated 1,000 – 10,000 folds higher potency than non-targeting *EAGLE*-305 consisting of a dummy arm.

EAGLE Showed Sialidase-Dependent Efficacy, Induced Anti-Tumor Immunological Memory, and Increased Immune Cell Infiltration

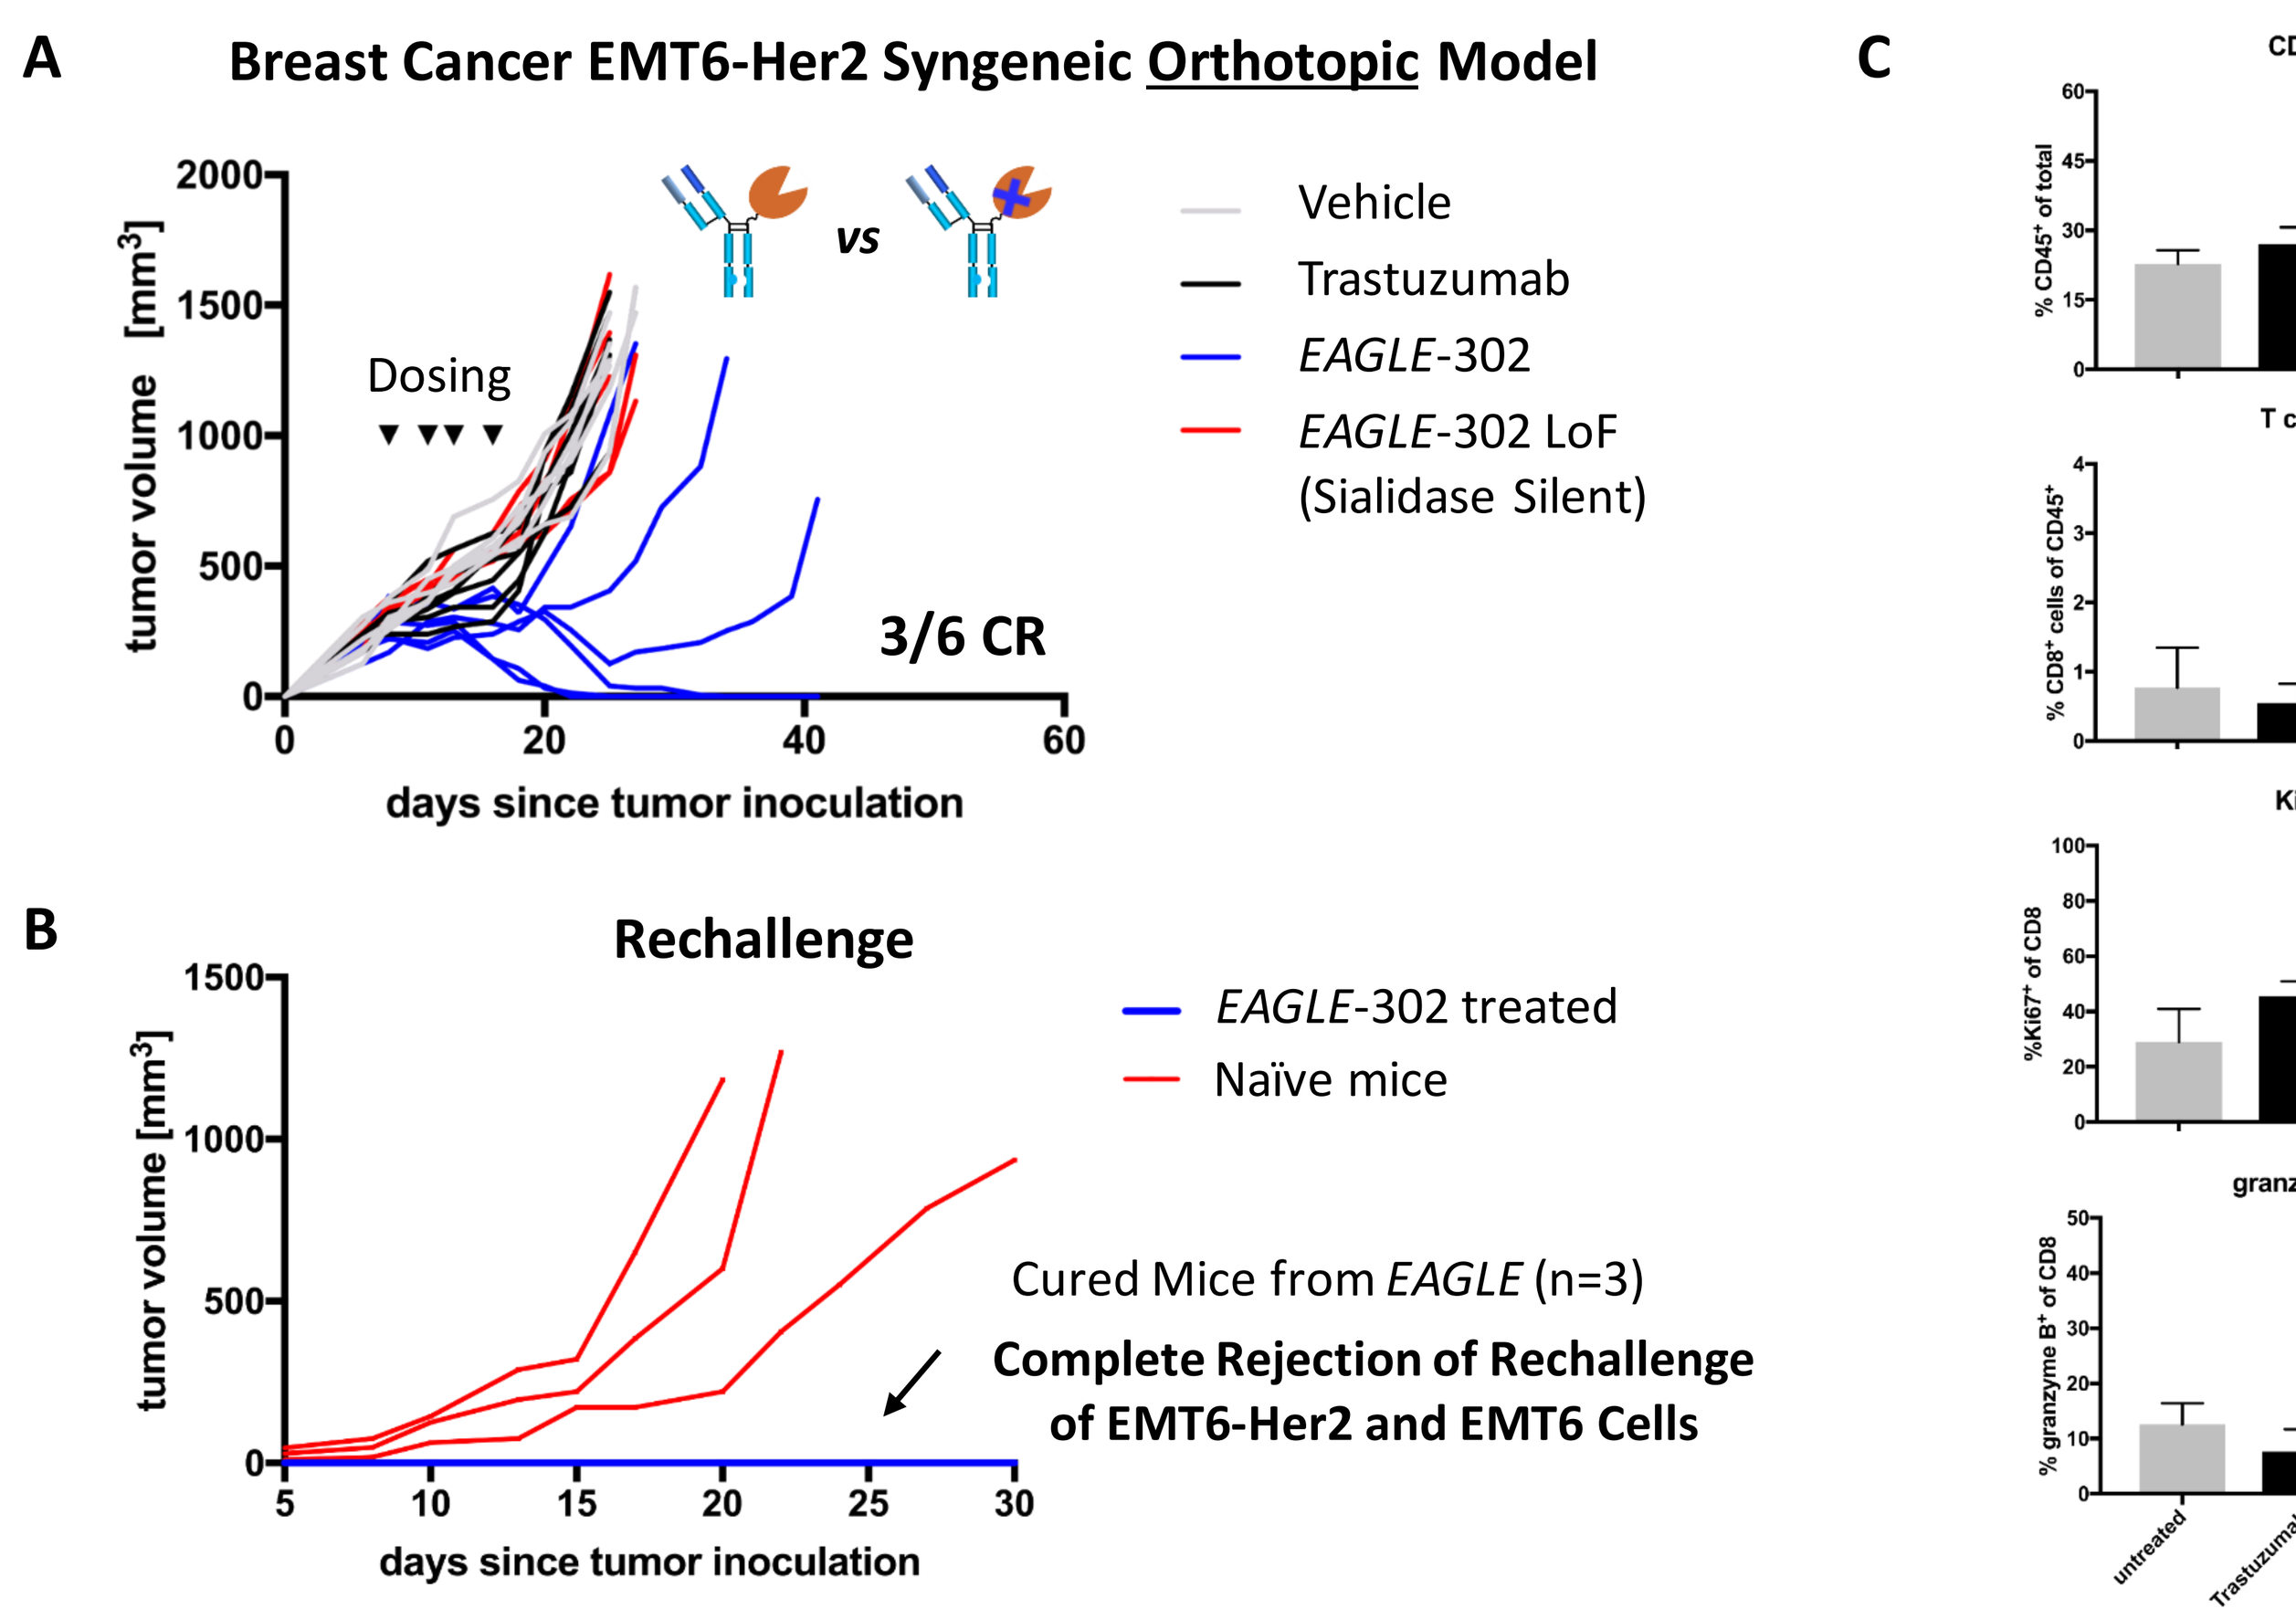
A
Breast Cancer EMT6-Her2 Syngeneic Orthotopic Model
vs
Dosing
Vehicle
Trastuzumab
EAGLE-302
EAGLE-302 LoF (Sialidase Silent)
3/6 CR
tumor volume [mm³]
days since tumor inoculation
B
Rechallenge
EAGLE-302 treated
Naïve mice
Cured Mice from EAGLE (n=3)
Complete Rejection of Rechallenge of EMT6-Her2 and EMT6 Cells
C
CD45
T cells
Ki67
granzyme B
untreated
Trastuzumab
EAGLE-302

Figure 4. Efficacy studies of *EAGLE* in orthotopic syngeneic breast cancer tumor model. (A) *EAGLE* demonstrated sialidase-dependent anti-tumor activity. Wild type BALB/c mice (n = 6 per group) were inoculated with EMT6-Her2 cells into mammary fat and treated with *EAGLE*-302 and its loss-of function control when tumor sizes reached about ~250 mm³. *EAGLEs* and trastuzumab were dosed twice a week for 2 weeks at 10mg/kg. (B) Cured mice from *EAGLE* treatment in experiment (A) rejected re-challenge of EMT6-Her2 and EMT6 cells. (C) FACS analysis of tumor infiltrating lymphocytes after *EAGLE* treatment. *EAGLE* increased T cell infiltration and activation.

The Mechanism of Action of *EAGLE* Involves Innate and Adaptive Immunity

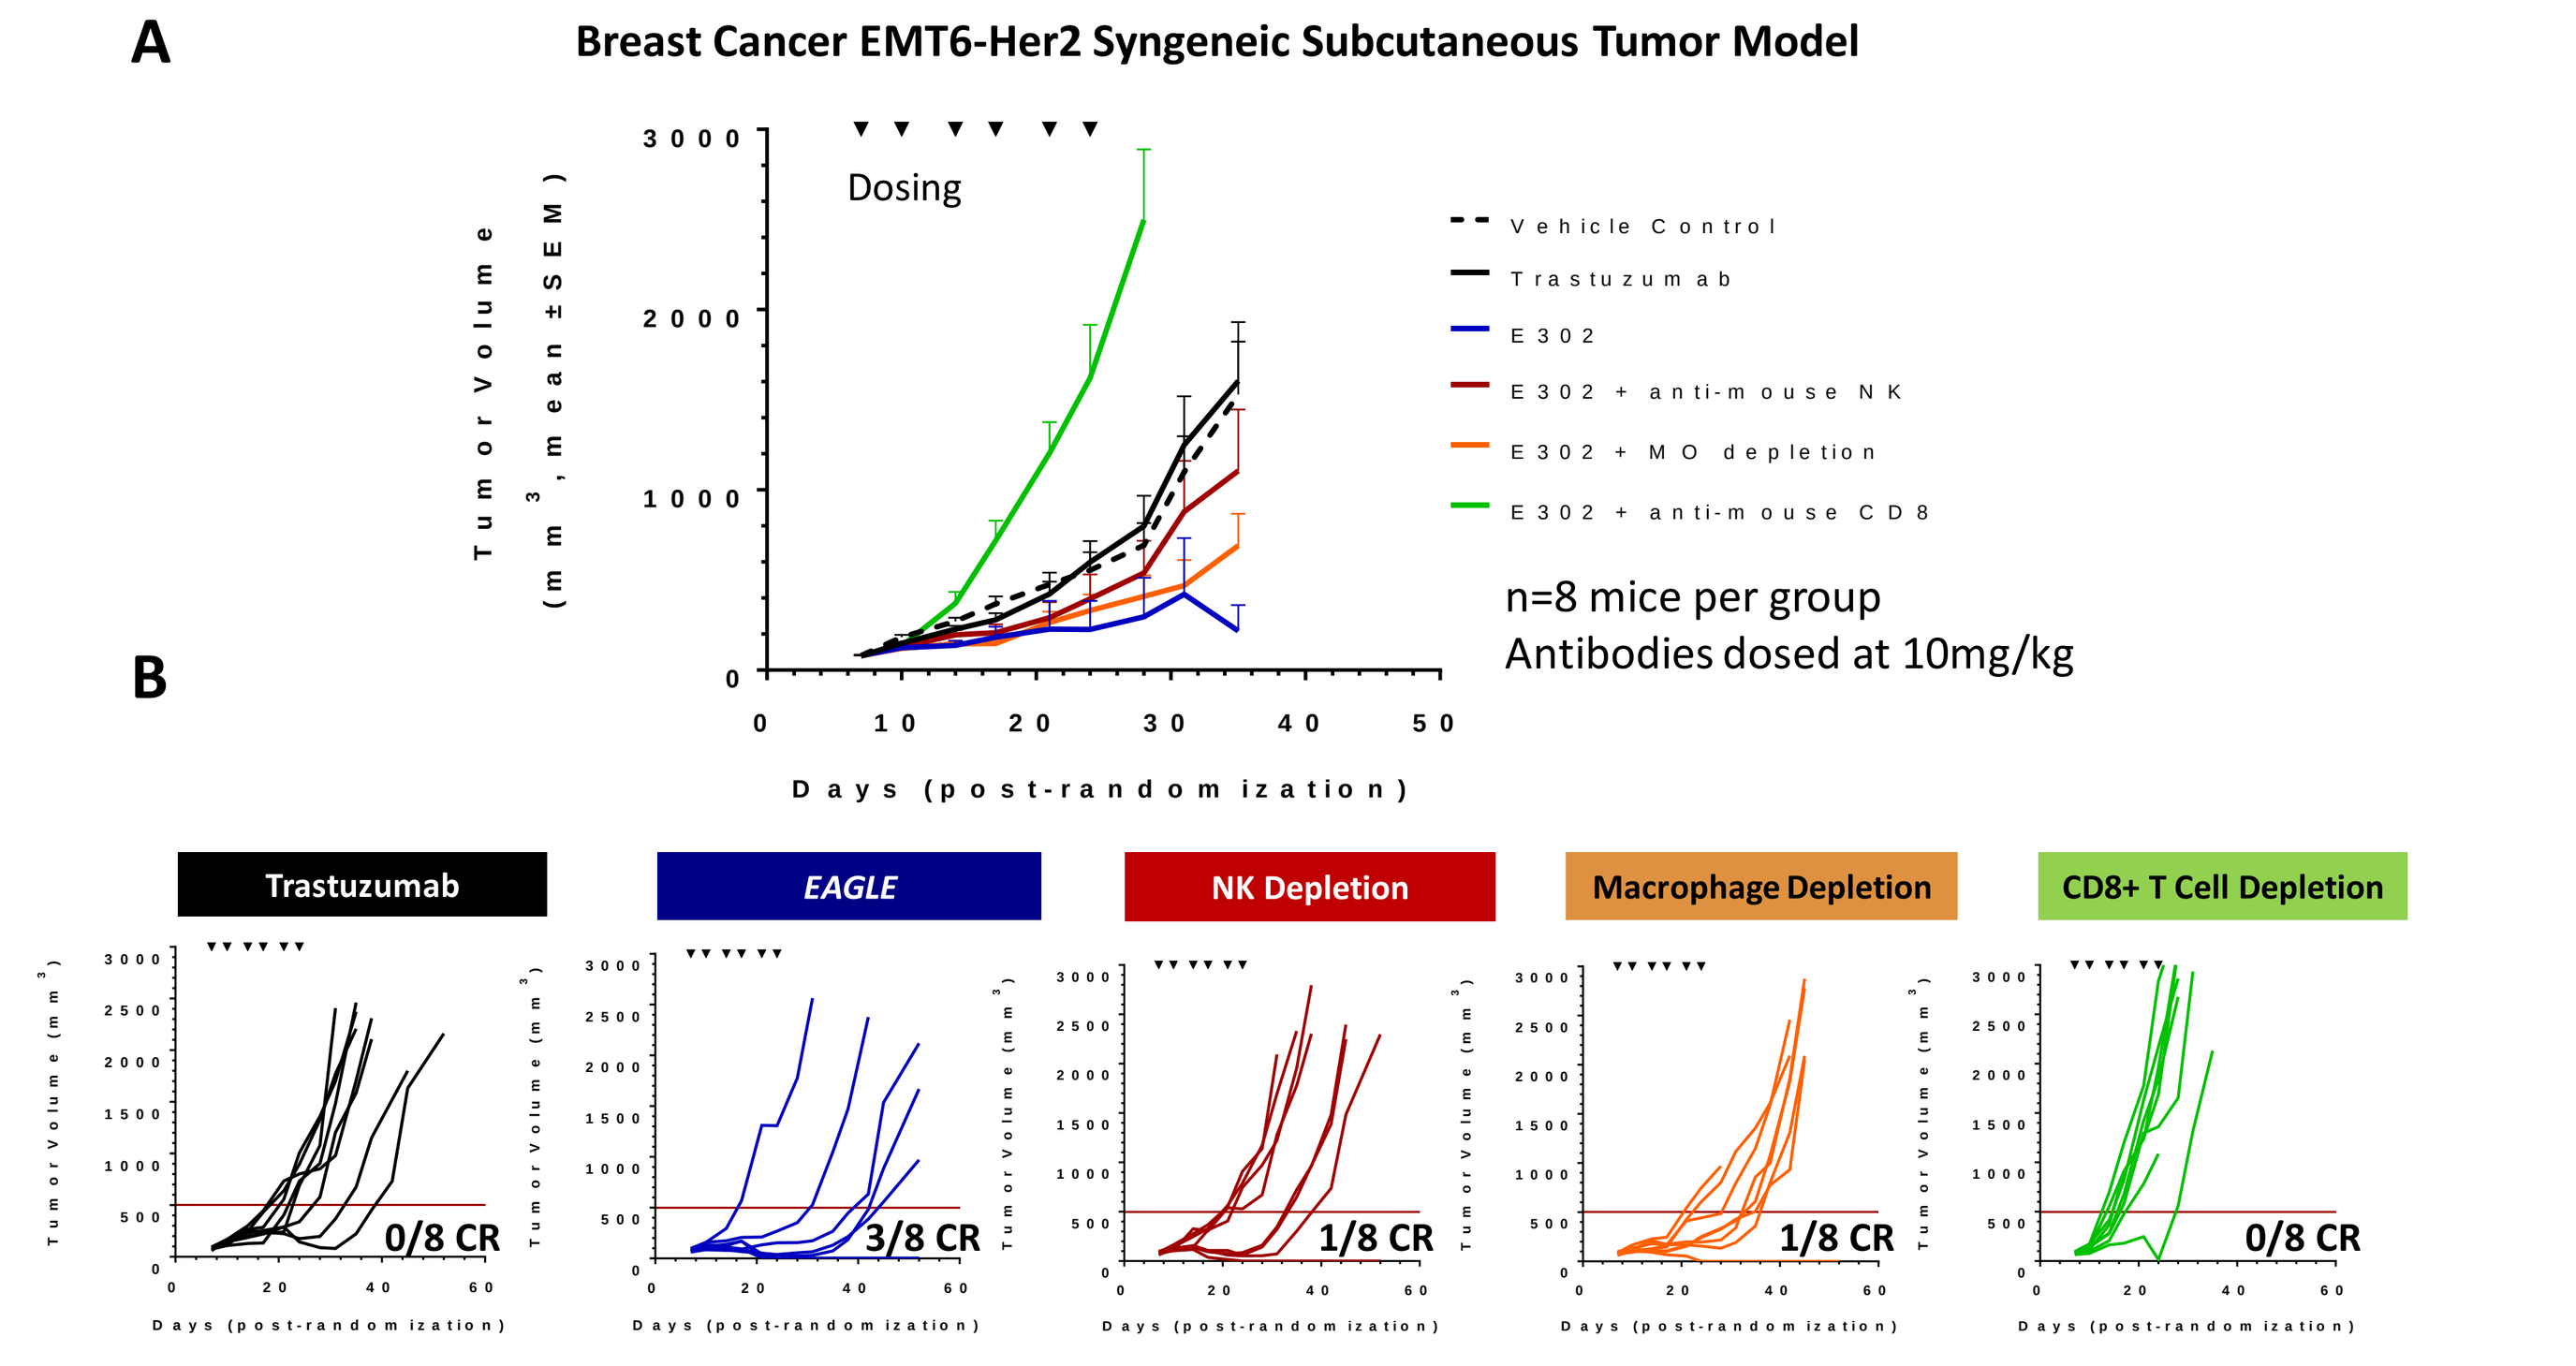
A
Breast Cancer EMT6-Her2 Syngeneic Subcutaneous Tumor Model
Dosing
Vehicle Control
Trastuzumab
E302
E302 + anti-mouse NK
E302 + MO depletion
E302 + anti-mouse CD8
n=8 mice per group
Antibodies dosed at 10mg/kg
Days (post-randomization)
B
Trastuzumab
EAGLE
NK Depletion
Macrophage Depletion
CD8+ T Cell Depletion
0/8 CR
3/8 CR
1/8 CR
1/8 CR
0/8 CR

Figure 5. The MoA of *EAGLE* involves NK cells, macrophages, and CD8+ T-cells. Wild type BALB/c mice (n = 8 per group) were injected s.c. EMT6-Her2 cells. When tumor sizes reached about ~120 mm³, mice were treated on the same days as *EAGLE*-302 with either anti-mouse NK1.1 (10 mg/kg) to deplete natural killer cells, liposomal clodronate (0.5 mg/mouse, three times a week for two weeks) to deplete macrophages, or anti-mouse CD8α (10 mg/kg) to deplete CD8+ T cells. (A) Tumor growth curves of mean tumor sizes of each group. (B) Tumor growth curves of individual mice.

EAGLE has Monotherapy Efficacy Comparable to the Combination of α-PD1 and α-CTLA4 in the "Cold" Tumor B16 Model

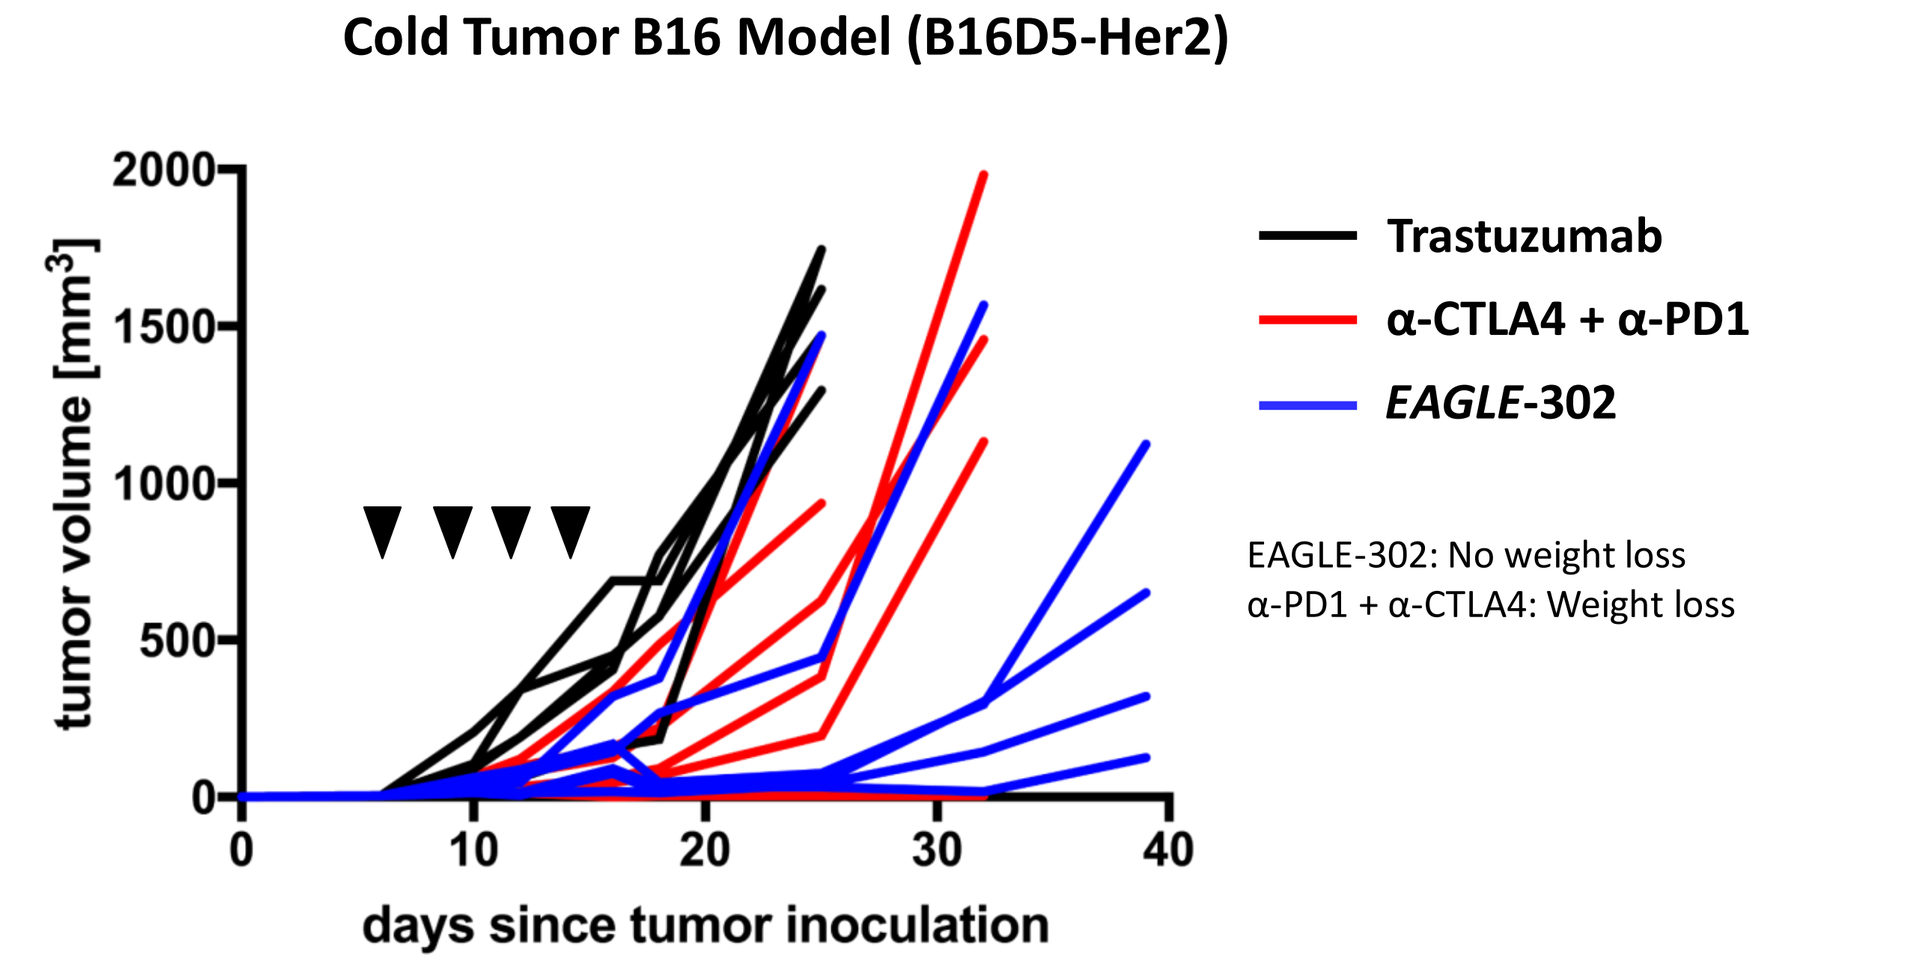
Cold Tumor B16 Model (B16D5-Her2)
Trastuzumab
α-CTLA4 + α-PD1
EAGLE-302
EAGLE-302: No weight loss
α-PD1 + α-CTLA4: Weight loss
tumor volume [mm³]
days since tumor inoculation

Figure 6. *EAGLE* demonstrated single agent anti-tumor activity in B16D5-Her2 syngeneic tumor model. Wild type C57BL/6 mice (n = 6 per group) were inoculated with B16D5-Her2 cells subcutaneously and treated with *EAGLE*-302, the combination of anti-CTLA4 and anti-PD-1, or Trastuzumab when tumor sizes reached about ~100 mm3. All antibodies were dosed twice a week for 2 weeks at 10mg/kg. Each line represented a tumor growth curve of an individual mouse.

EAGLE Has Significant Combination Efficacy with T-Cell Checkpoint Inhibition

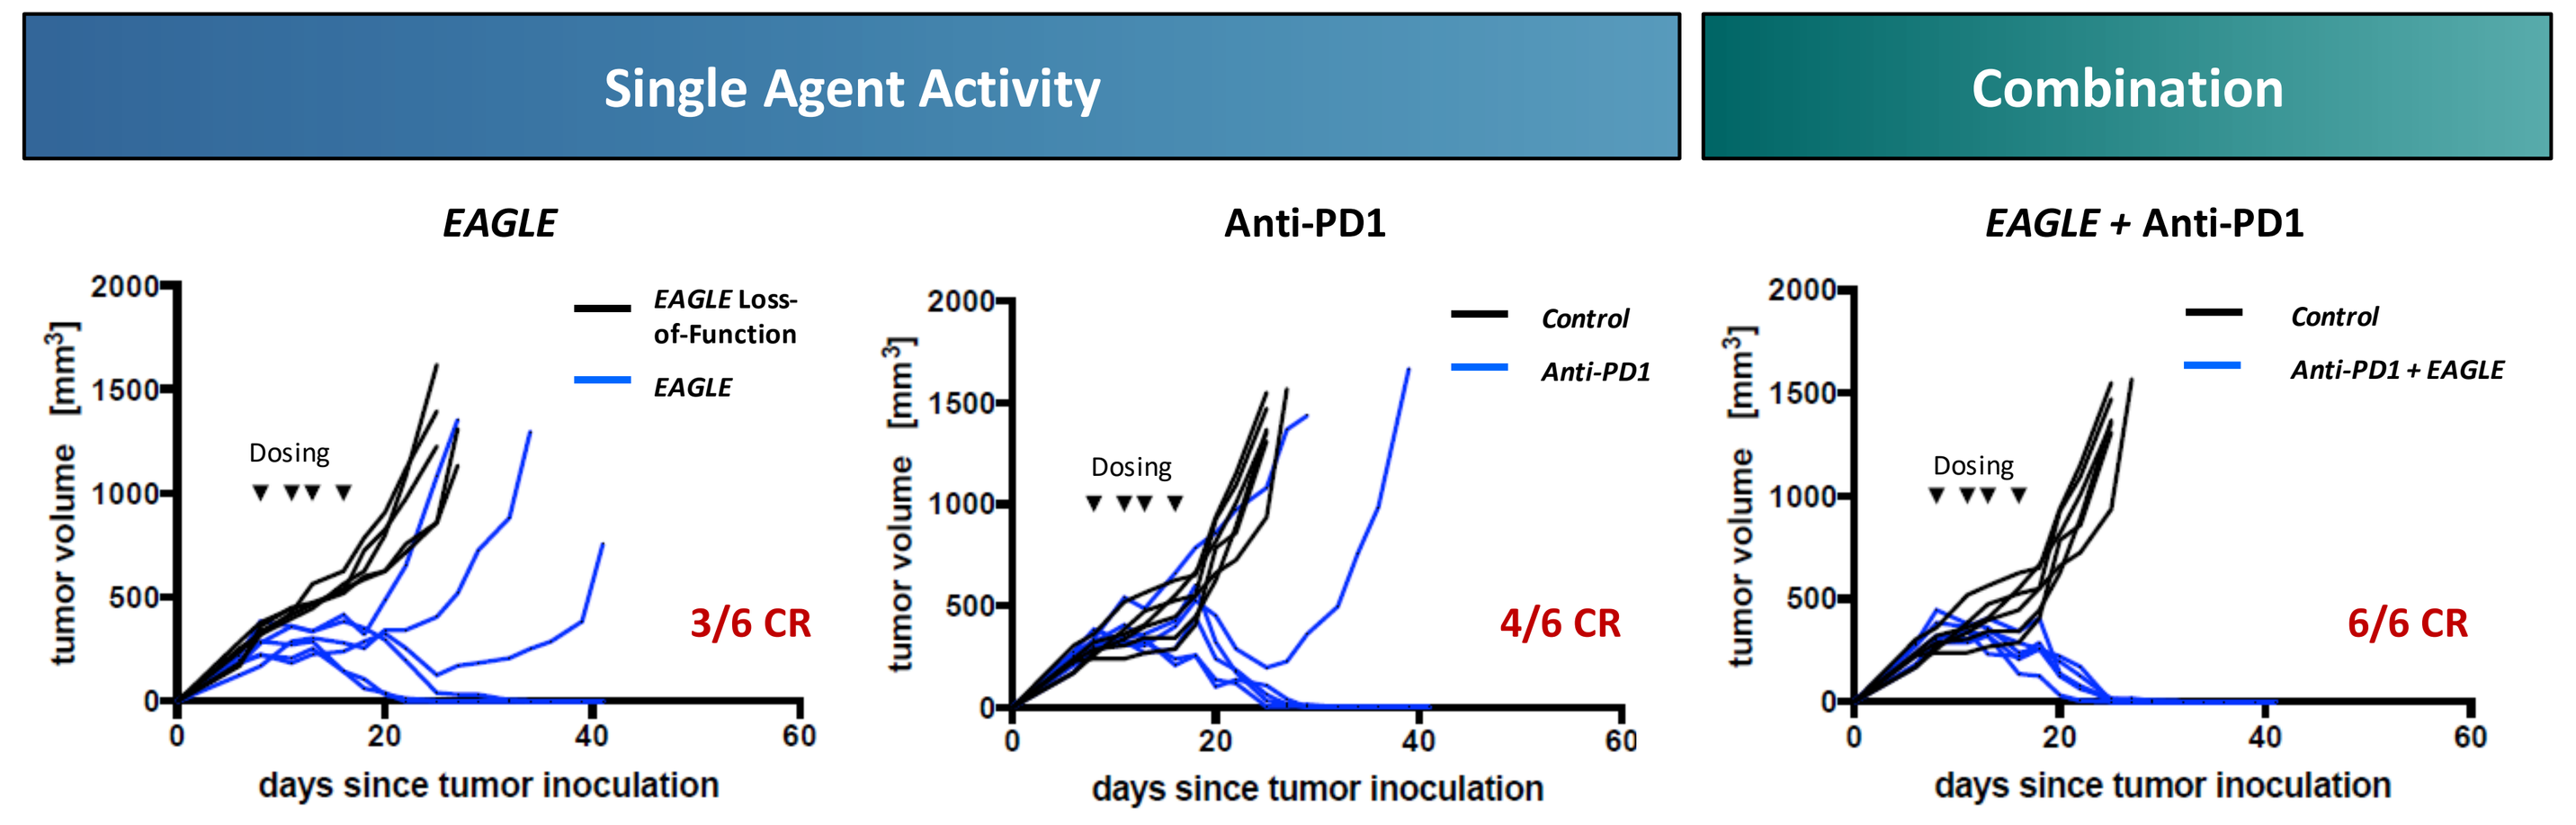
Breast Cancer EMT6-Her2 Syngeneic Orthotopic Model
Single Agent Activity
Combination
EAGLE
EAGLE Loss-of-Function
EAGLE
3/6 CR
Anti-PD1
Control
Anti-PD1
4/6 CR
EAGLE + Anti-PD1
Control
Anti-PD1 + EAGLE
6/6 CR
Dosing
tumor volume [mm³]
days since tumor inoculation

Figure 7. *EAGLE* achieved 100% cures in breast cancer EMT6-Her2 orthotopic model in combination with PD-1/PD-L1 inhibitions. Wild type *BALB/c* mice (n = 6 per group) were inoculated with EMT6-Her2 cells into mammary fat. When tumor sizes reached about ~250 mm3, mice were treated with *EAGLE*-302, anti- PD1 mAb, the combination of *EAGLE*-302 and anti-PD1 mAb, or controls of *EAGLE*-loss-of function and Trastuzumab twice a week for 2 weeks at 10mg/kg.

EAGLEs with Engineered Human Sialidase

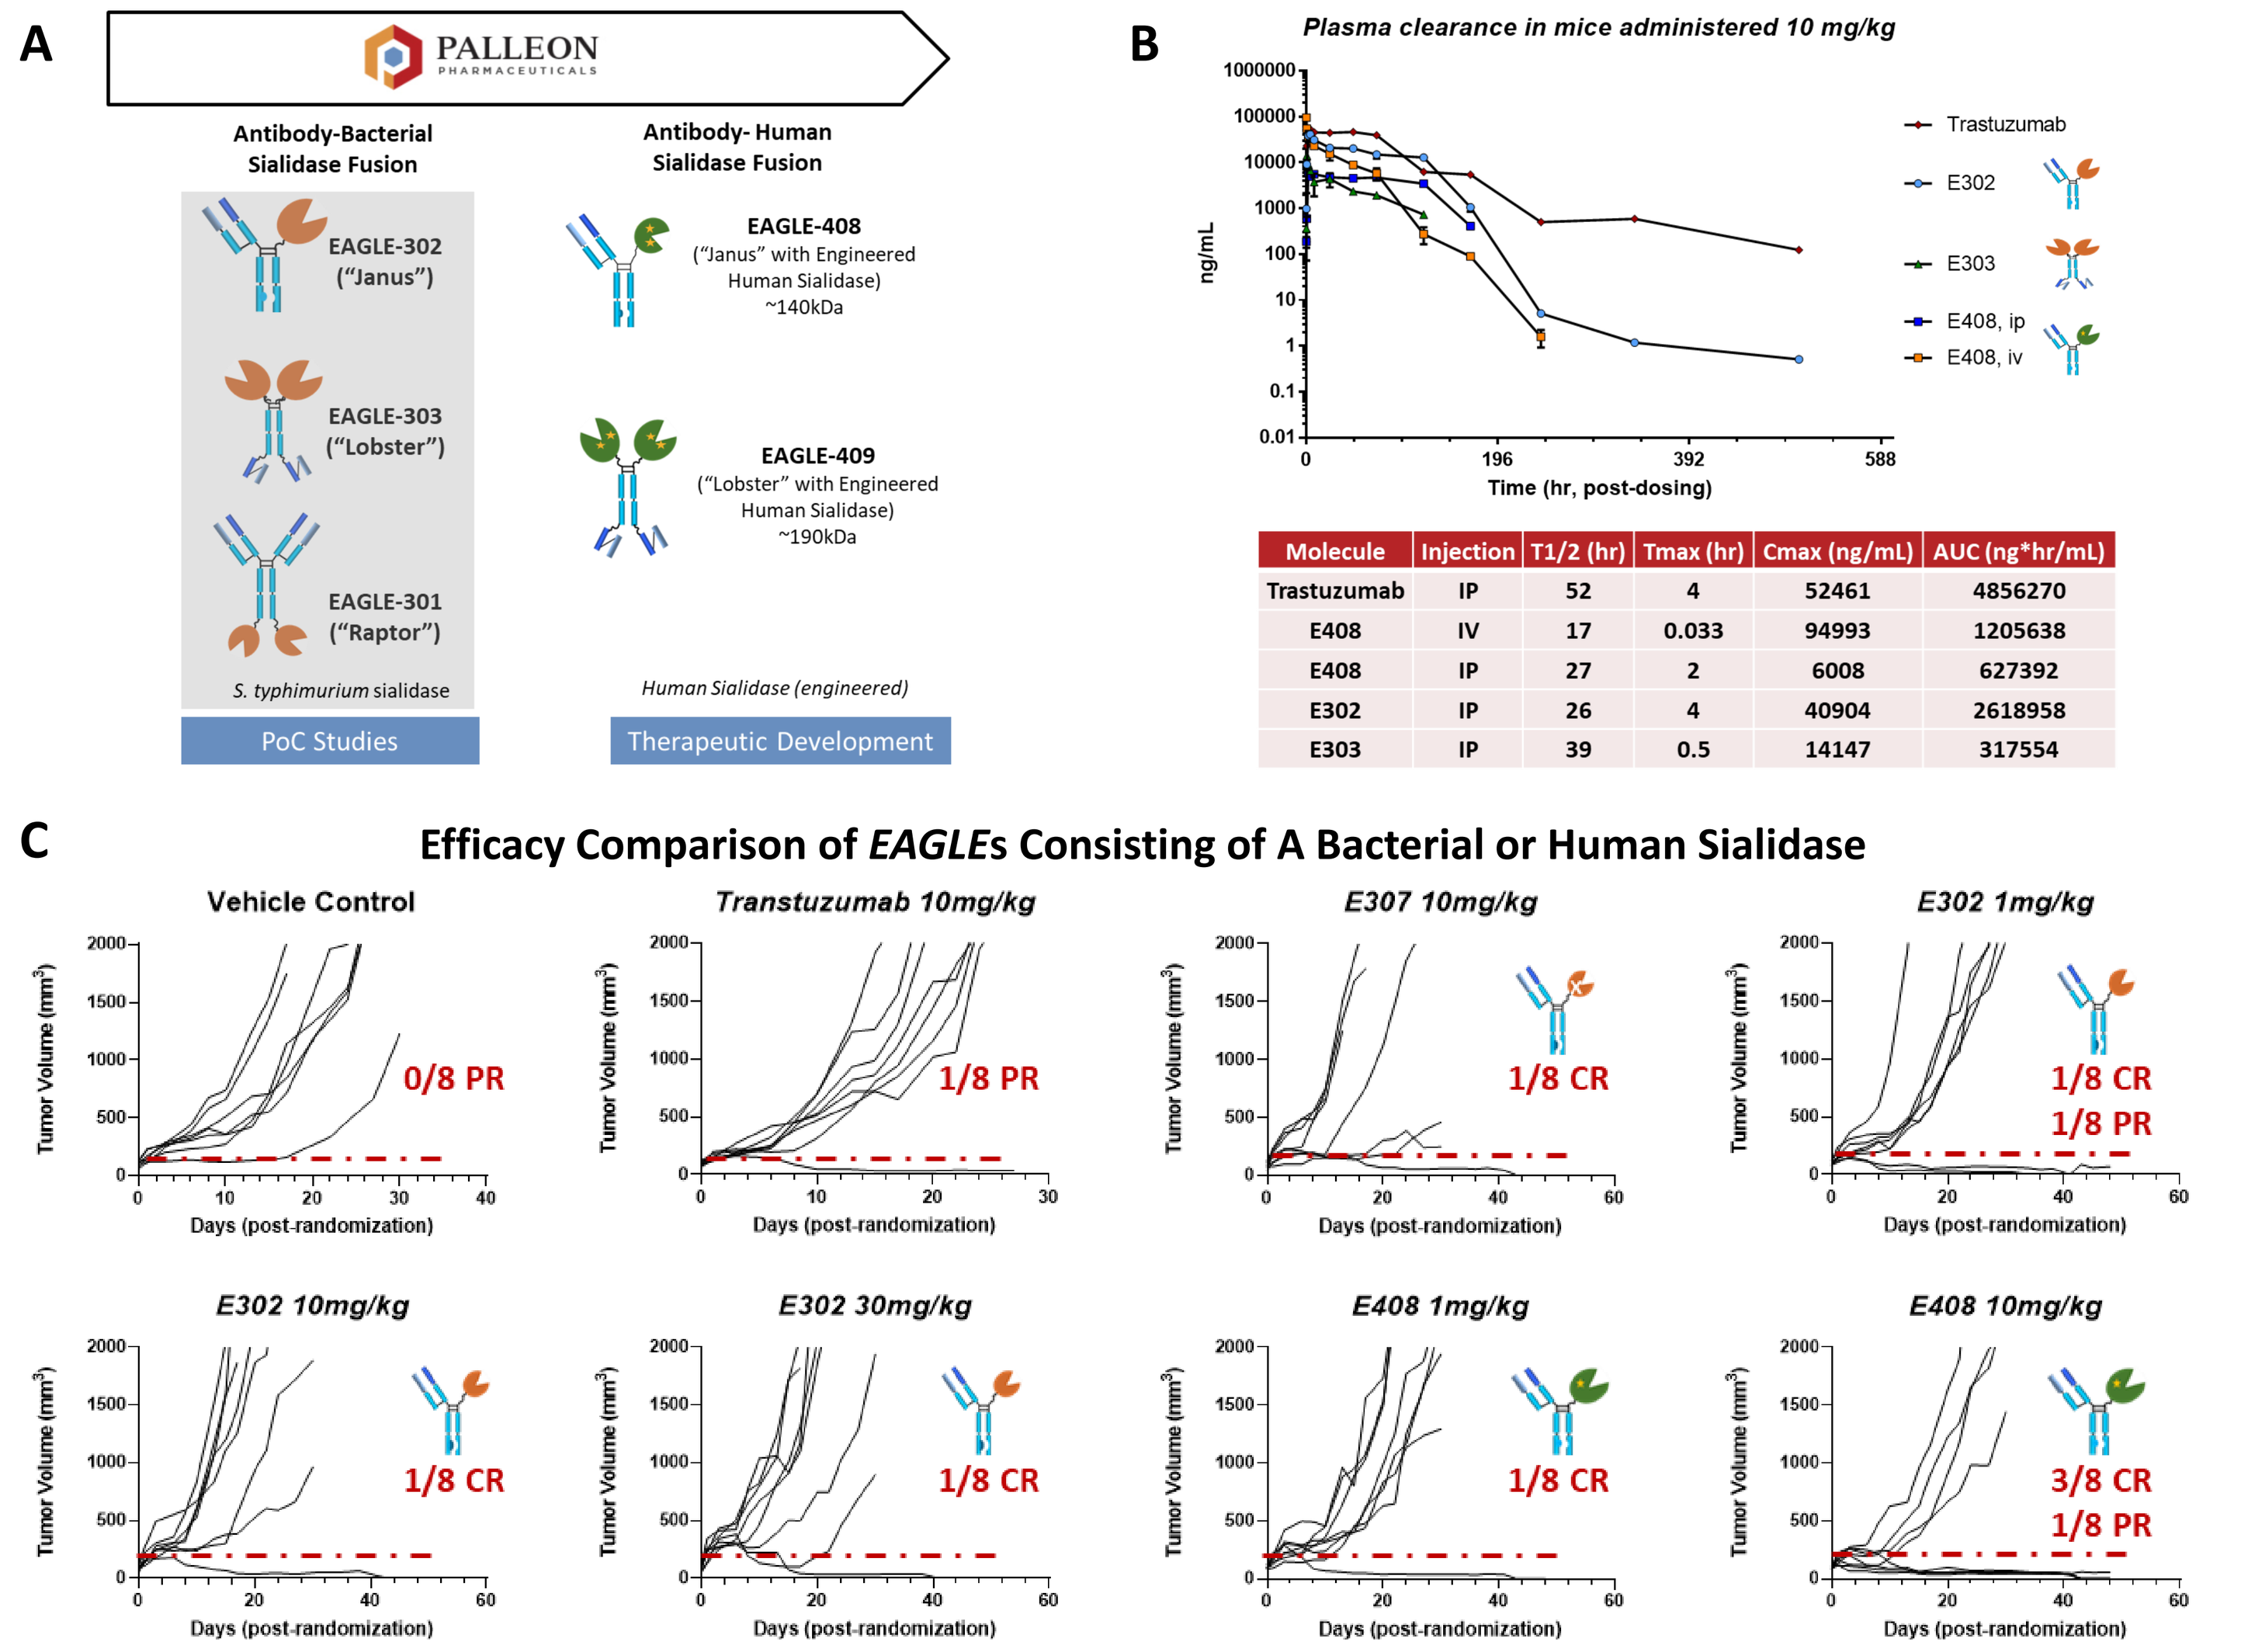
A
PALLEON PHARMACEUTICALS
Antibody-Bacterial Sialidase Fusion
Antibody- Human Sialidase Fusion
EAGLE-302 ("Janus")
EAGLE-303 ("Lobster")
EAGLE-301 ("Raptor")
S. typhimurium sialidase
EAGLE-408 ("Janus" with Engineered Human Sialidase) ~140kDa
EAGLE-409 ("Lobster" with Engineered Human Sialidase) ~190kDa
Human Sialidase (engineered)
PoC Studies
Therapeutic Development
B
Plasma clearance in mice administered 10 mg/kg
Trastuzumab
E302
E303
E408, ip
E408, iv
ng/mL
Time (hr, post-dosing)
C
Efficacy Comparison of EAGLEs Consisting of A Bacterial or Human Sialidase
Vehicle Control
0/8 PR
Transtuzumab 10mg/kg
1/8 PR
E307 10mg/kg
1/8 CR
E302 1mg/kg
1/8 CR
1/8 PR
E302 10mg/kg
1/8 CR
E302 30mg/kg
1/8 CR
E408 1mg/kg
1/8 CR
E408 10mg/kg
3/8 CR
1/8 PR
Tumor Volume (mm³)
Days (post-randomization)

Molecule	Injection	T1/2 (hr)	Tmax (hr)	Cmax (ng/mL)	AUC (ng*hr/mL)
Trastuzumab	IP	52	4	52461	4856270
E408	IV	17	0.033	94993	1205638
E408	IP	27	2	6008	627392
E302	IP	26	4	40904	2618958
E303	IP	39	0.5	14147	317554

Figure 8. Development of human sialidase-based *EAGLEs*. (A) Evolution of *EAGLE* platform. *In vitro* and *in vivo* PoC studies were conducted using the bacterial sialidase. The therapeutic *EAGLEs* were developed using a human sialidase with improved developability and enzymatic activities through protein engineering. (B) PK profiles of multiple *EAGLEs* in mice. *EAGLEs* demonstrated bispecific antibody-like PK profiles. (C) Efficacy comparison of *EAGLEs* consisting of a bacterial or human sialidase in breast cancer EMT6-Her2 syngenetic subcutaneous tumor model. *EAGLEs* with engineered human sialidase (E-408) showed comparable or even superior efficacy than *EAGLEs* of bacterial sialidase (E-302).

Proprietary *Hydra* Biomarker Assays to Detect Tumor Glyco-Codes in Patients

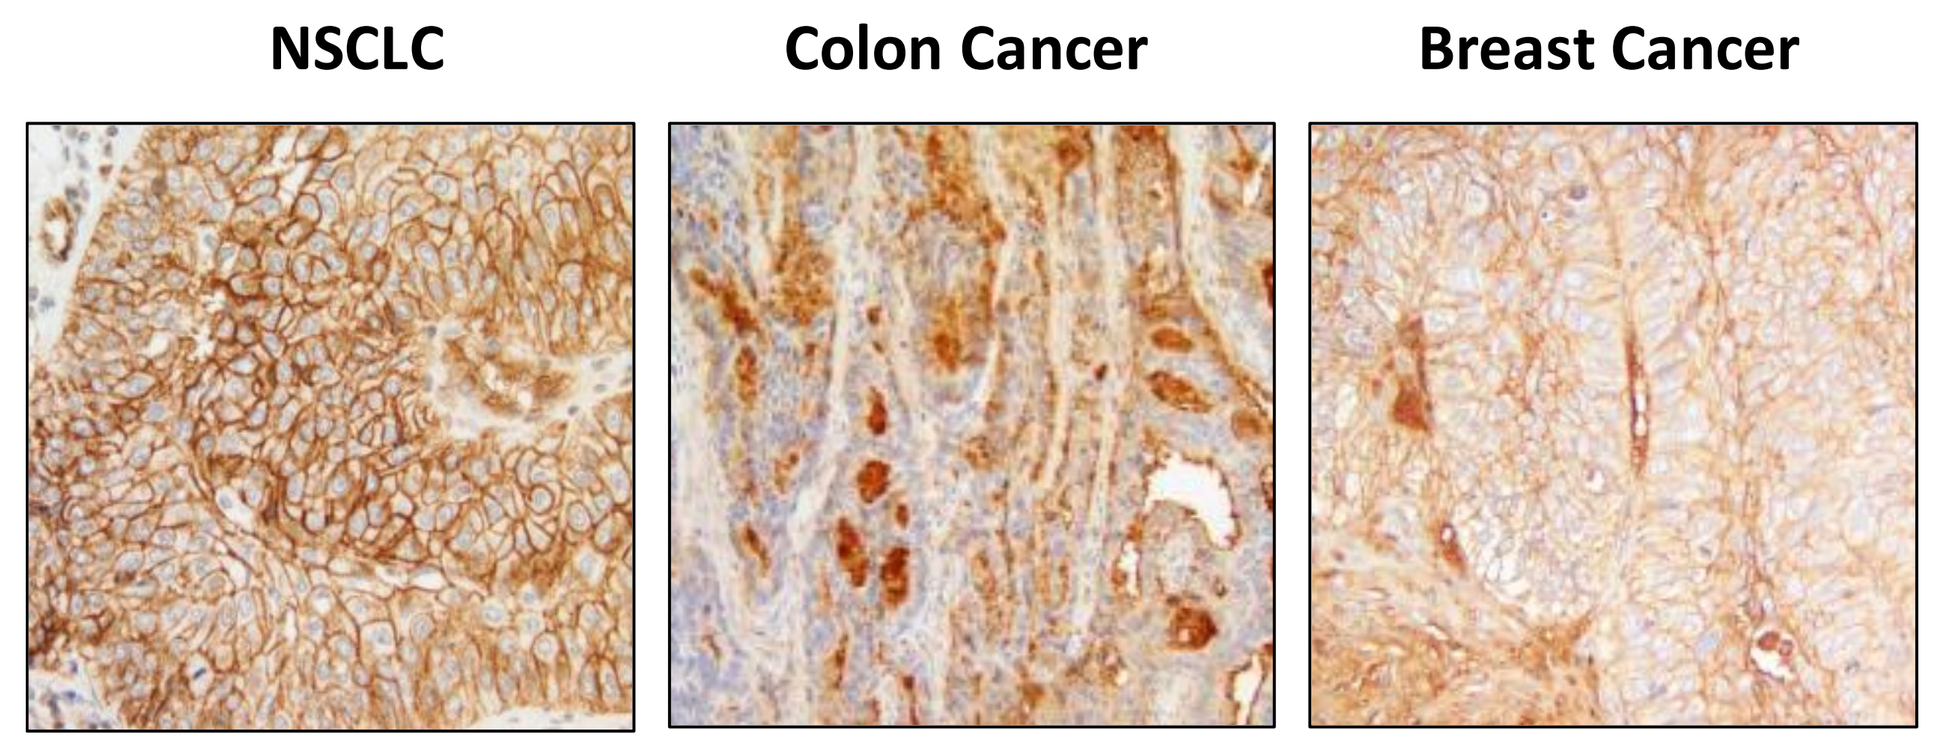
NSCLC
Colon Cancer
Breast Cancer

Figure 9. Representative *Hydra* staining of tumor sialoglycan levels in NSCLC, colon cancer, and breast cancer for patient selection.

Conclusions

- **Glyco-immune checkpoints play critical roles in cancer immune evasion**
 - Innate immune response
 - Adaptive immune response
- ***EAGLE* showed compelling monotherapy efficacy in syngeneic tumor models**
 - Single agent complete regressions with immunological memory
 - Efficacious in cold tumor model
 - Striking activity in combination with PD-1/PD-L1
- ***EAGLE* offers new opportunities to treat cancer targeting glyco-immune checkpoints**
 - Overcomes the heterogeneity challenges of tumor sialoglycans
 - Disables immunosuppressive glycan functions within tumor microenvironment
 - Transform existing tumor targeting mAbs into immune-modulating agents

References

1. Crocker PR, Varki A. Siglecs in the immune system. Immunology. 2001;103:137-45.
2. Adams O.J., et al, Targeting sialic acid-Siglec interactions to reverse immune suppression in cancer, Glycobiology, 2018, 28:640-7
3. Stanczak M., et al., Self-associated molecular patterns mediated cancer immune evasion by engaging Siglecs on T cells, J Clin Invest, 2018, 128: 4912–4923
4. Xiao H., et al, Glycocalyx editing for cancer immunotherapy, PNAS 2016, 113:10304-10309